145

Nadunolimab inhibits IL-1α/β-induced CXCR1/2 ligand expression and reduces serum levels of CXCL1 and CXCL5 in NSCLC and PDAC patients

Camilla Rydberg Millrud[1]; Elin Jaensson Gyllenbäck[1]; Petter Skoog[1]; Annika Sanfridson[1] and David Liberg[1]

1 Cantargia AB, Ideon Gateway, SE-223 63 Lund, Sweden

Introduction

Interleukin-1 receptor accessory protein (IL1RAP) is expressed on tumor cells and stromal cells in most solid tumors, including pancreatic ductal adenocarcinoma (PDAC) and non-small cell lung cancer (NSCLC). IL1RAP is a co-receptor of the IL-1 receptor (IL1R1), and its dimerization with IL1R1 is required for IL-1 signaling. IL-1 is expressed in the tumor microenvironment (TME) and contributes to chemoresistance and an immune-suppressive TME. Recruitment of immune-suppressive myeloid cells is key to these effects and expression of chemokines can be induced by IL-1 signaling in various cells, including cancer-associated fibroblasts (CAF).

Here we study effects of nadunolimab on ligands for the CXCR1 and CXCR2 receptors that are crucial for migration of immune-suppressive cells into the tumor and the promotion of angiogenesis in the TME.

Nadunolimab (CAN04) is a fully humanized ADCC-enhanced monoclonal IgG1 antibody targeting IL1RAP and disrupting both IL-1α and IL-1β signaling. Nadunolimab is currently evaluated in a phase II clinical trial (NCT03267316) in combination with gemcitabine/nab-paclitaxel (GN) in PDAC and with cisplatin/gemcitabine (CG) in NSCLC. Interim efficacy data are stronger than expected from chemotherapy alone based on historical controls, in a total of 73 PDAC patients, median iPFS of 7.2 months and median OS of 12.7 months is observed. In 30 NSCLC patients, a response rate of 53% is achieved, resulting in median PFS of 6.8 months.

Objective

- To study the effects of nadunolimab on IL-1α- and IL-1β-induced CXCR1/2 ligand expression in different cell types related to the TME and in patients on nadunolimab treatment.

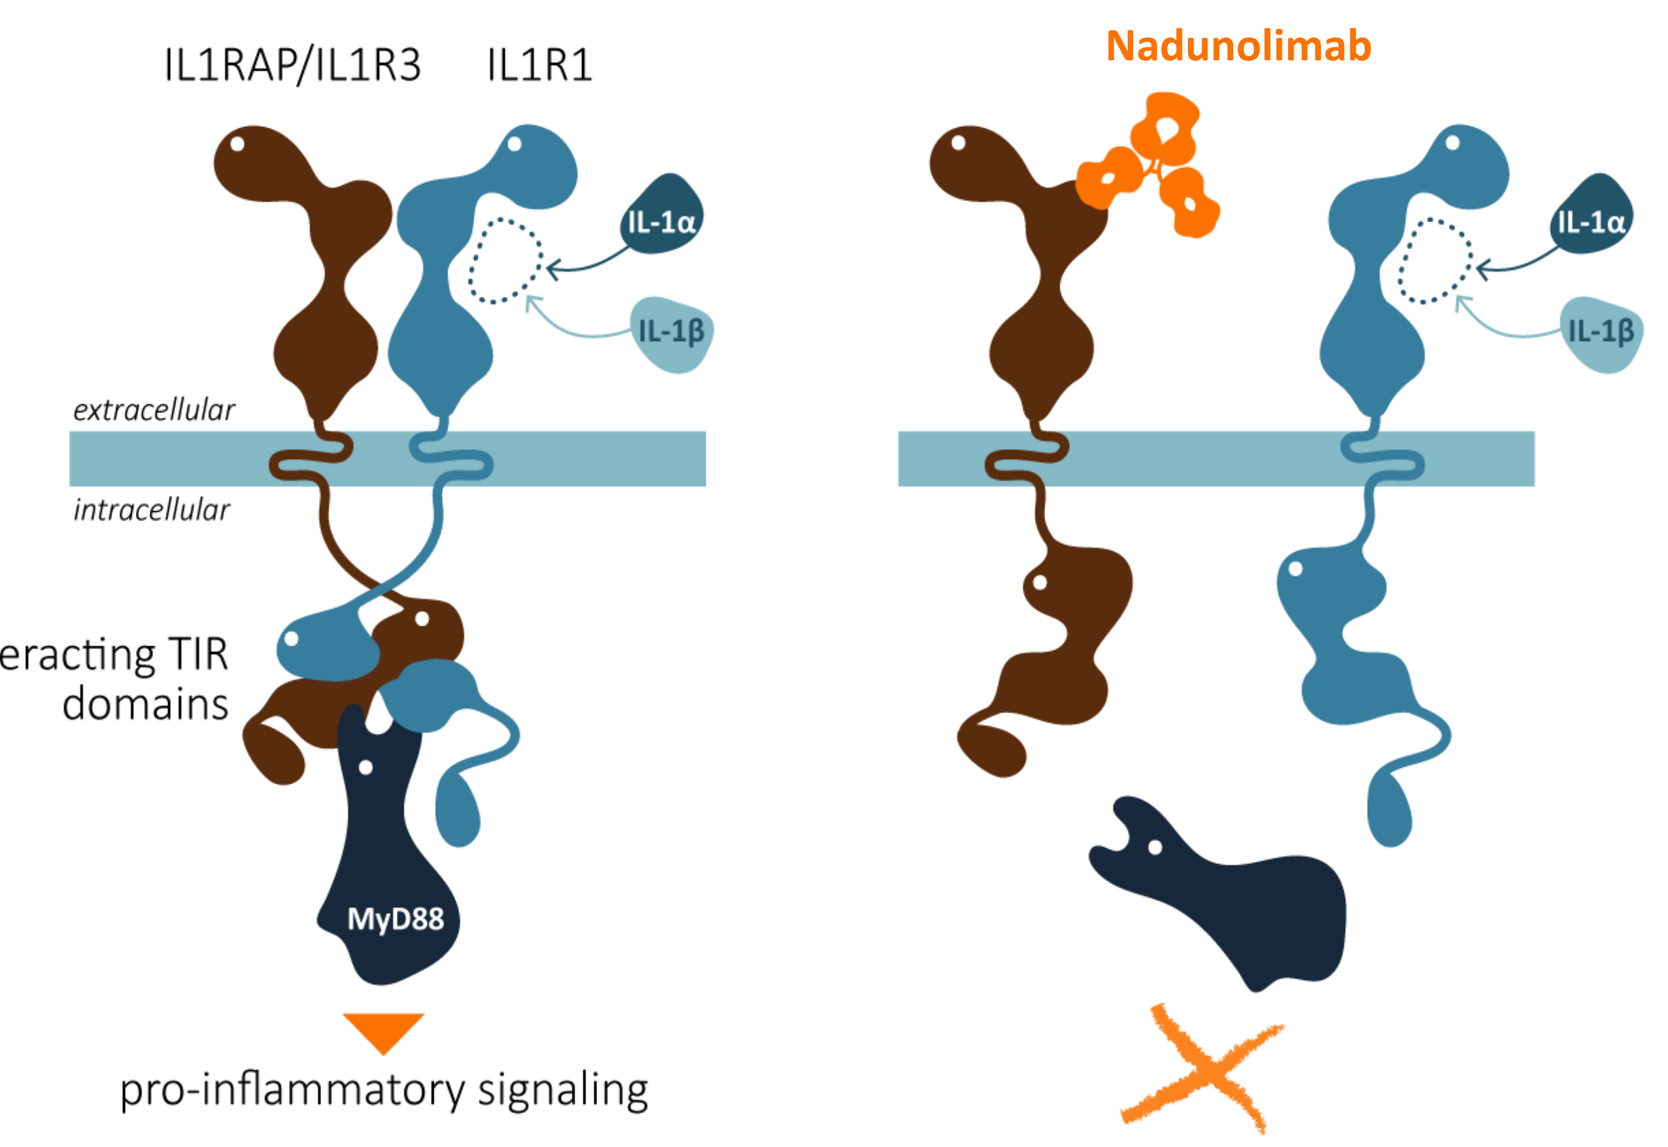
IL1RAP/IL1R3
IL1R1
IL-1α
IL-1β
extracellular
intracellular
interacting TIR domains
MyD88
pro-inflammatory signaling
Nadunolimab

Background

IL1RAP is expressed by tumor, stroma and immune cells in PDAC and NSCLC tumors

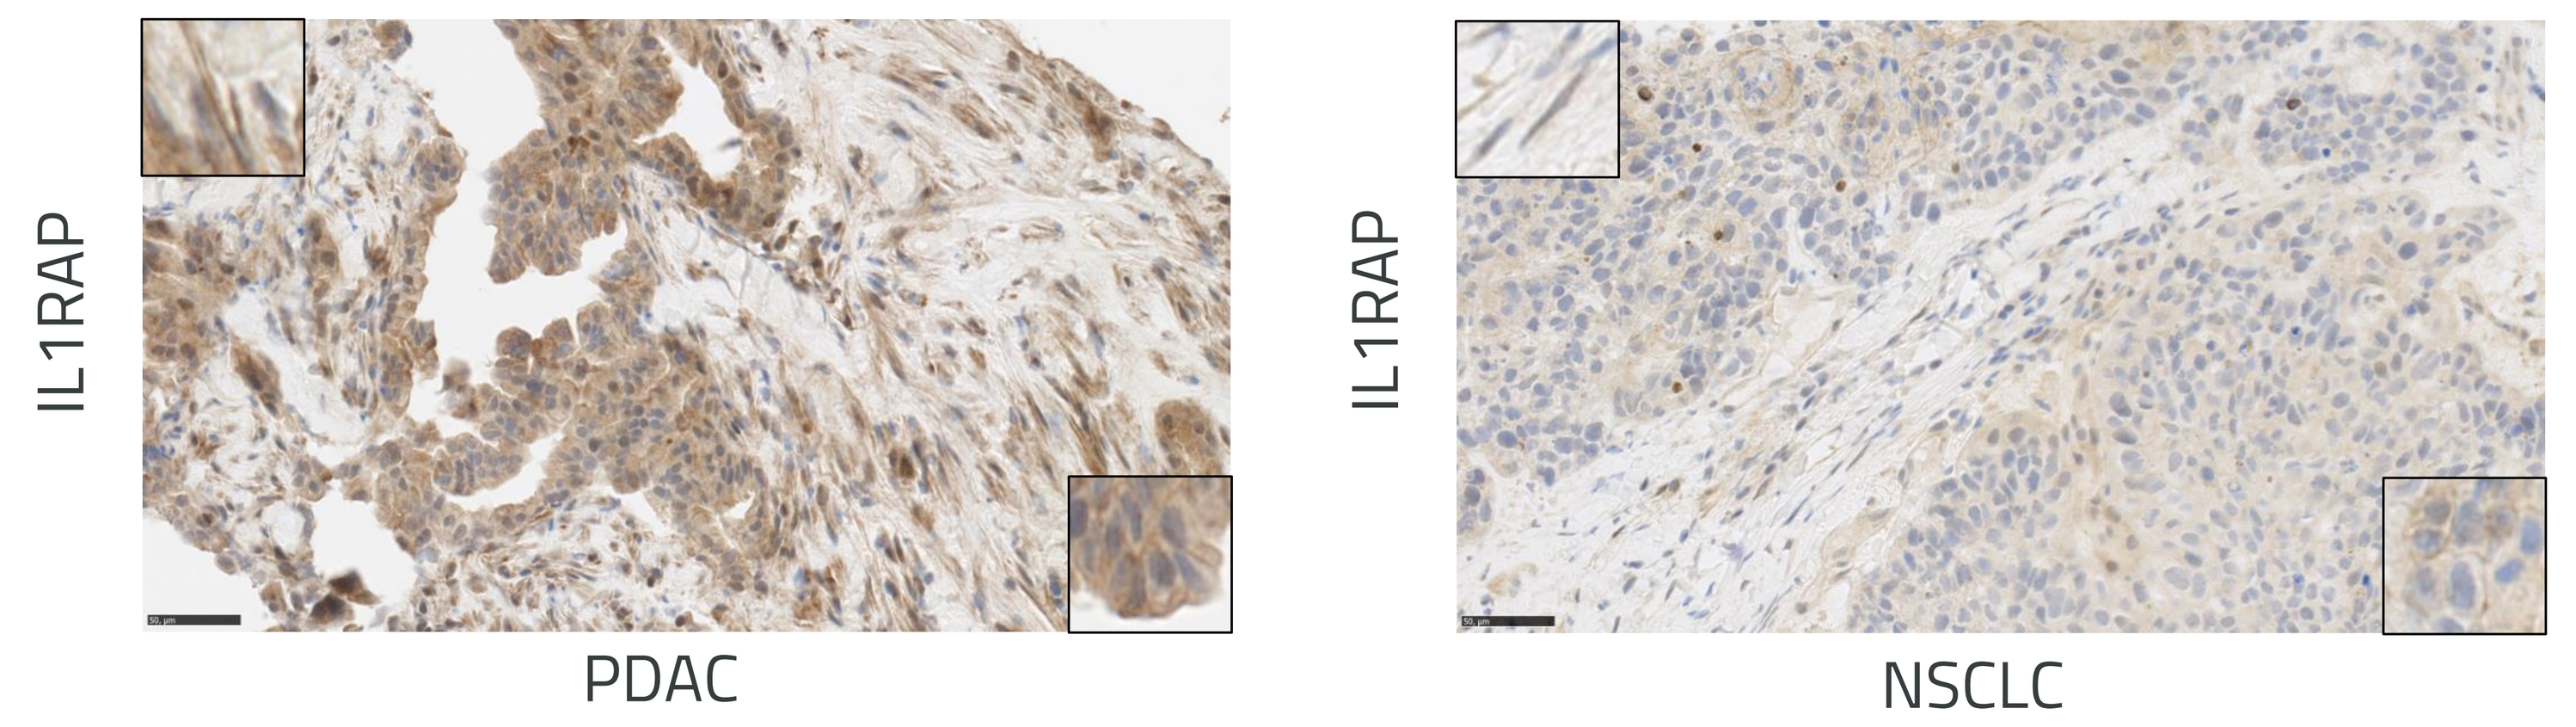
IL1RAP
PDAC
IL1RAP
NSCLC

Figure 1. IL1RAP expression in a pancreatic biopsy from a PDAC patient (left) and a lung biopsy from a squamous NSCLC patient (right). In essentially all biopsies (n=68) analyzed, IL1RAP was detected on CAFs, infiltrating immune cells and on tumor cells. Positive staining of stromal cells is shown in enlarged inserts in the top left corners, and positive staining of tumor cells in the lower right corners.

www.cantargia.com

Results

IL-1α and IL-1β induce an IL1RAP-dependent release of CXCR1/2 ligands in human blood

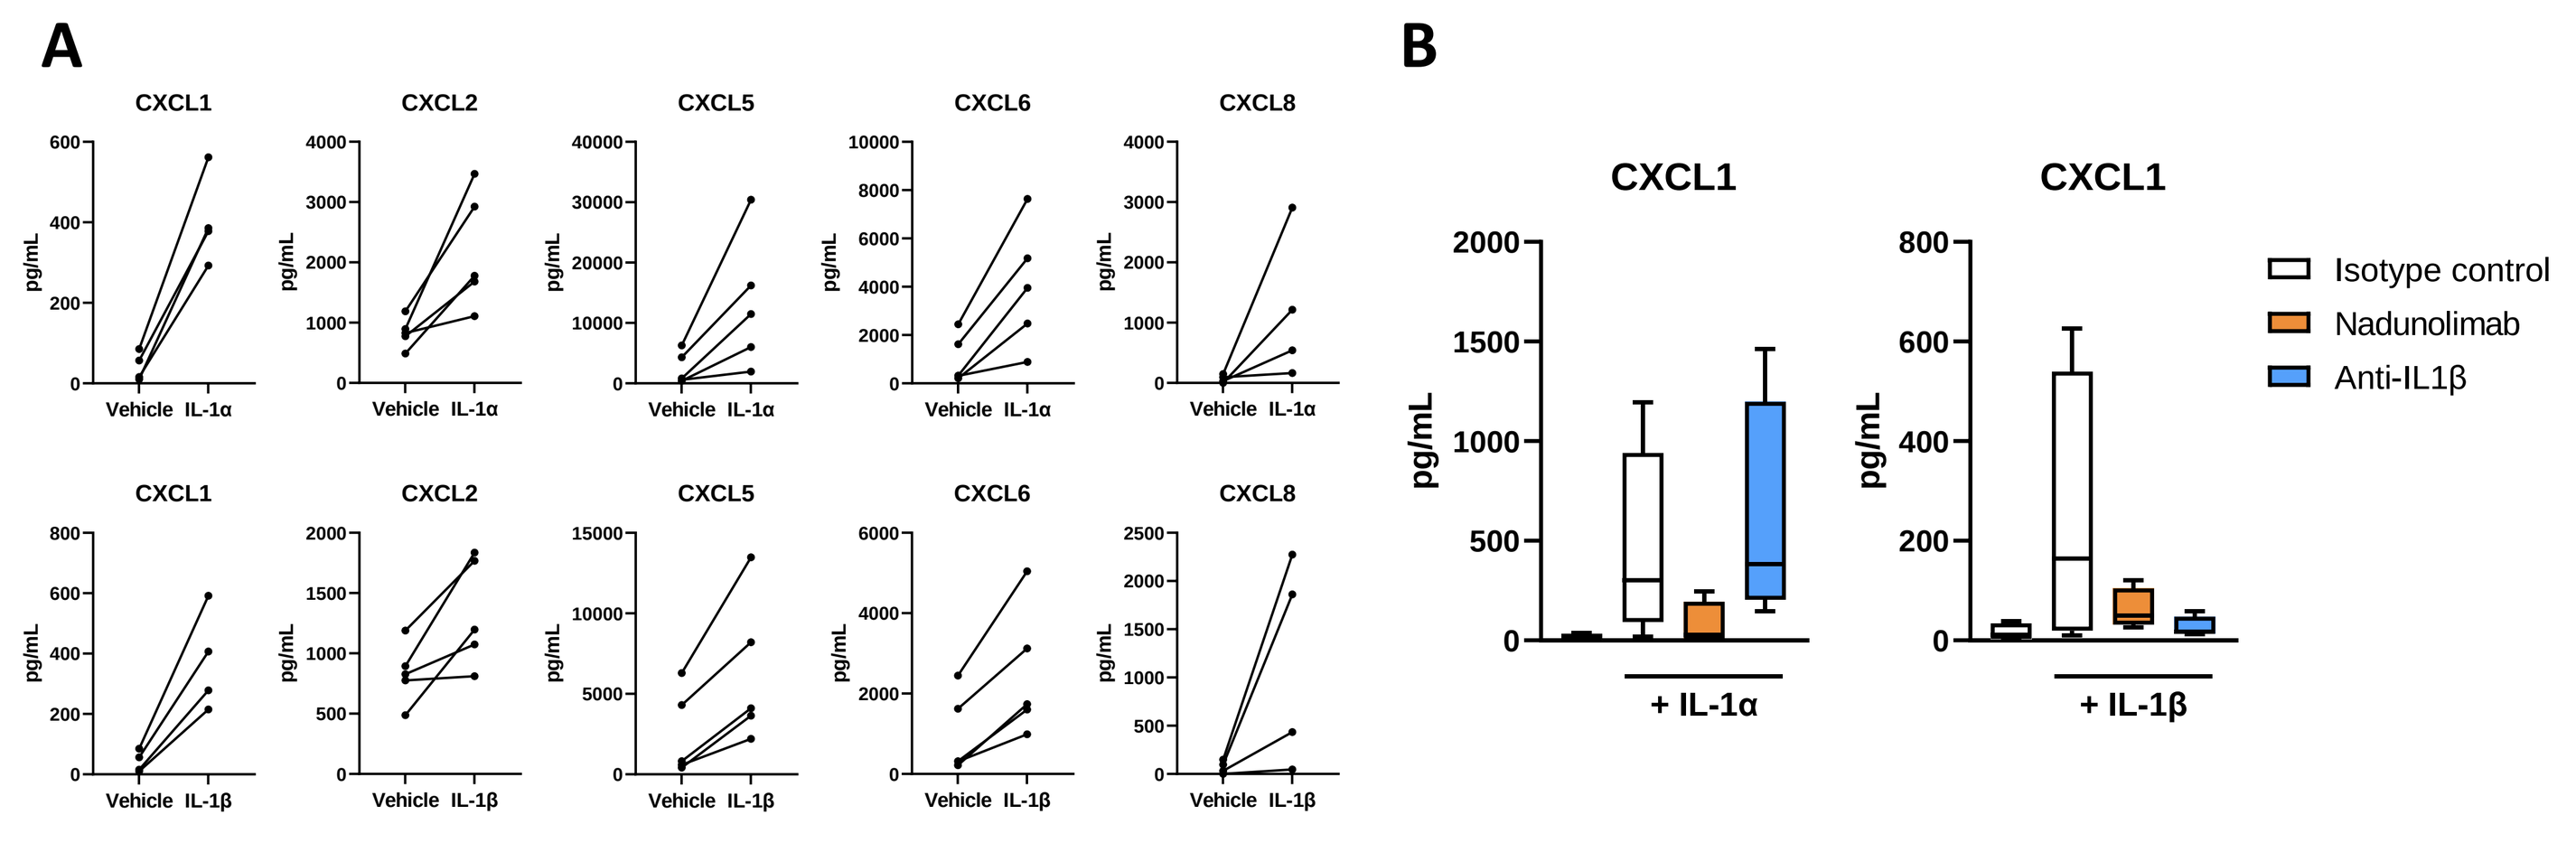
A
CXCL1
CXCL2
CXCL5
CXCL6
CXCL8
pg/mL
Vehicle IL-1α
Vehicle IL-1β
B
CXCL1
+ IL-1α
+ IL-1β
Isotype control
Nadunolimab
Anti-IL1β

Figure 2. (A) Whole blood was stimulated with IL-1α or IL-1β, supernatants were collected after 18-21 hrs and analyzed by Luminex. A robust induction of CXCL1, CXCL2, CXCL5, CXCL6 and CXCL8 was seen in all donors. (B) CXCL1 was tested for inhibition by nadunolimab. Induction of CXCL1 could be potently blocked by nadunolimab after either IL-1α or IL-1β stimulation while anti-IL-1β antibody only blocked IL-1β-induced CXCL1.

PDAC tumor cells induce secretion of CXCR1/2 ligands from CAFs, which can be blocked by nadunolimab but not anti-IL-1β

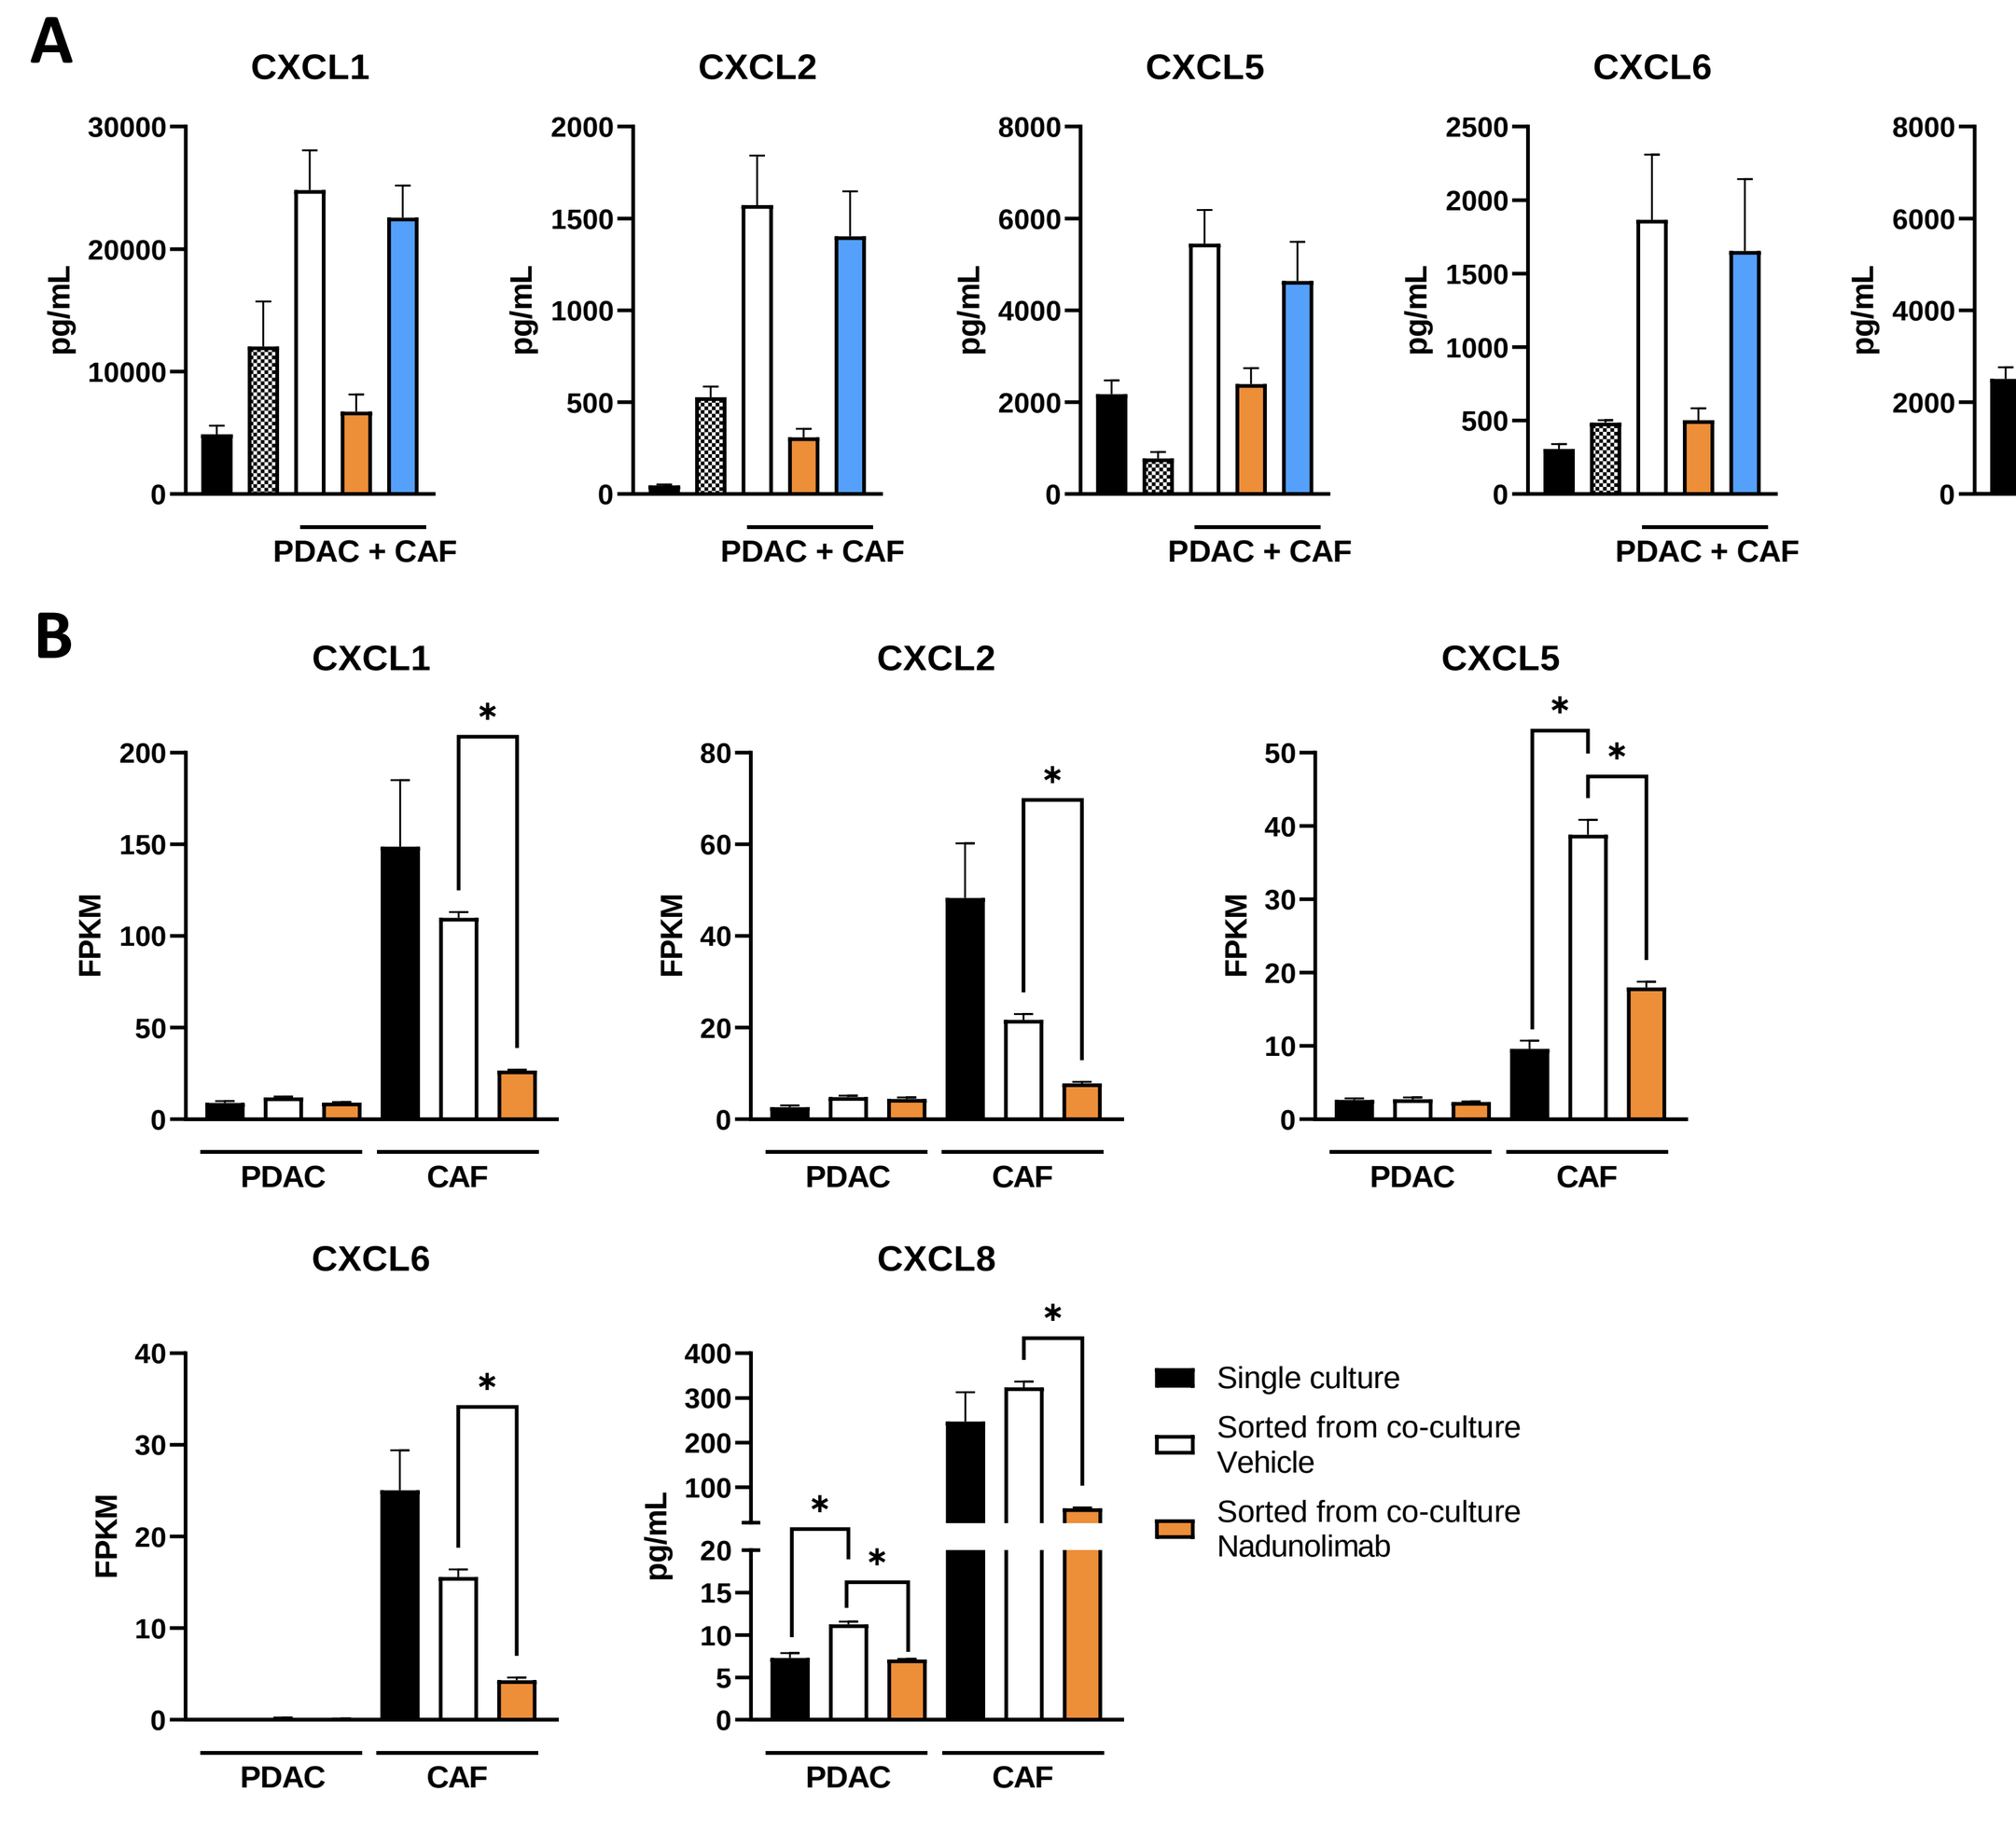
A
CXCL1
CXCL2
CXCL5
CXCL6
CXCL8
pg/mL
PDAC + CAF
PDAC
CAF
Isotype control
Nadunolimab
Anti-IL1β
B
FPKM
Single culture
Sorted from co-culture Vehicle
Sorted from co-culture Nadunolimab

Figure 3. (A) The human PDAC cell line BxPC3 and pancreatic CAFs were cultured in the ratio 1:2 for 72 hrs in the presence of an isotype control, nadunolimab or anti-IL-1β. Cell supernatants were harvested and analyzed by Luminex for the expression of CXCL1, CXCL2, CXCL5, CXCL6 and CXCL8. All chemokines tested were induced and could be reduced by nadunolimab but not by anti-IL-1β alone. (B) Co-cultured PDAC and pancreatic CAFs were separated based on the expression of EpCAM using FACS, followed by RNAseq analysis. All ligands tested show that mRNA was produced primarily by the CAFs. A significant upregulation of CXCL5 was observed in co-cultured CAFs.

Nadunolimab inhibits IL-1α and IL-1β induced secretion of CXCR1/2 ligands from pancreatic CAFs

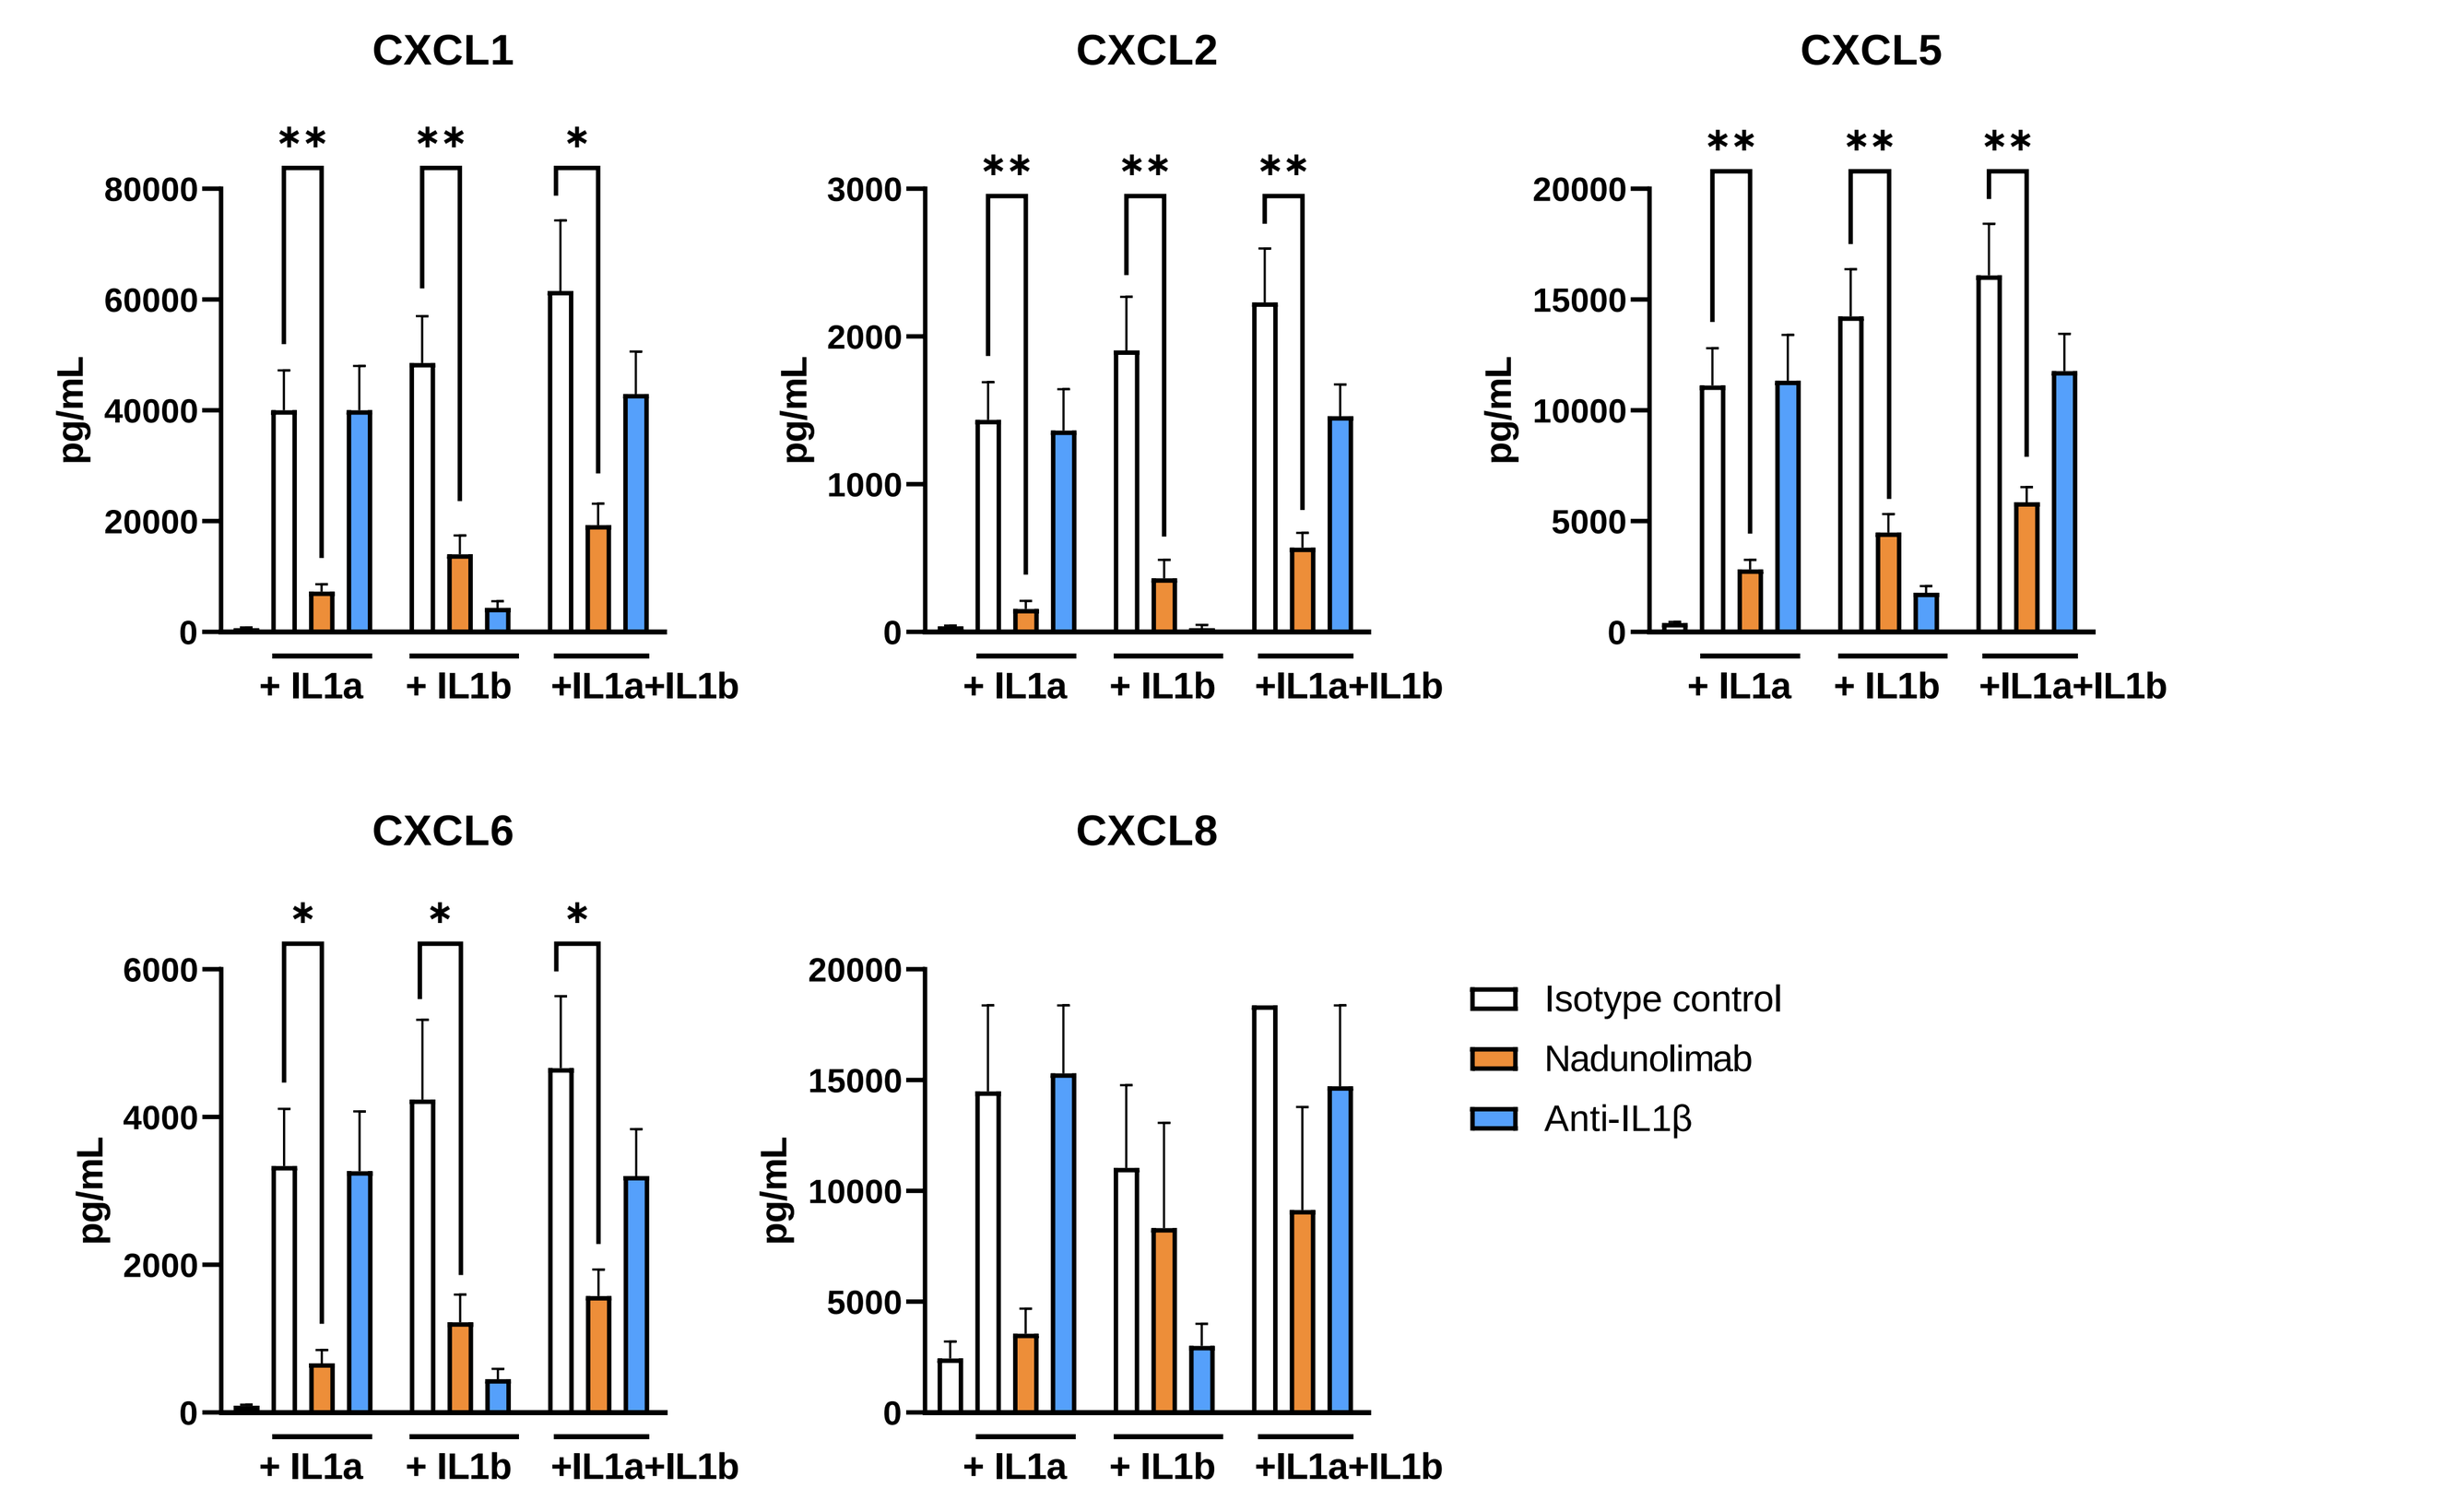
CXCL1
CXCL2
CXCL5
CXCL6
CXCL8
pg/mL
+ IL1a
+ IL1b
+IL1a+IL1b
Isotype control
Nadunolimab
Anti-IL1β

Figure 4. CAFs were treated with isotype control, nadunolimab or anti-IL-1β for 30 min before culture with or without IL-1α, IL-1β or IL-1α+IL-1β for 24 hrs. The supernatants were harvested and analyzed for the secretion of the CXCR1/2 ligands by Luminex. Nadunolimab was able to reduce both IL-1α- and IL-1β-induced ligand secretion from CAFs.

mRNA data indicate an upregulation of IL1RAP, IL1A, IL1B and CXCR1/2 ligands in PDAC tumors (public databases)

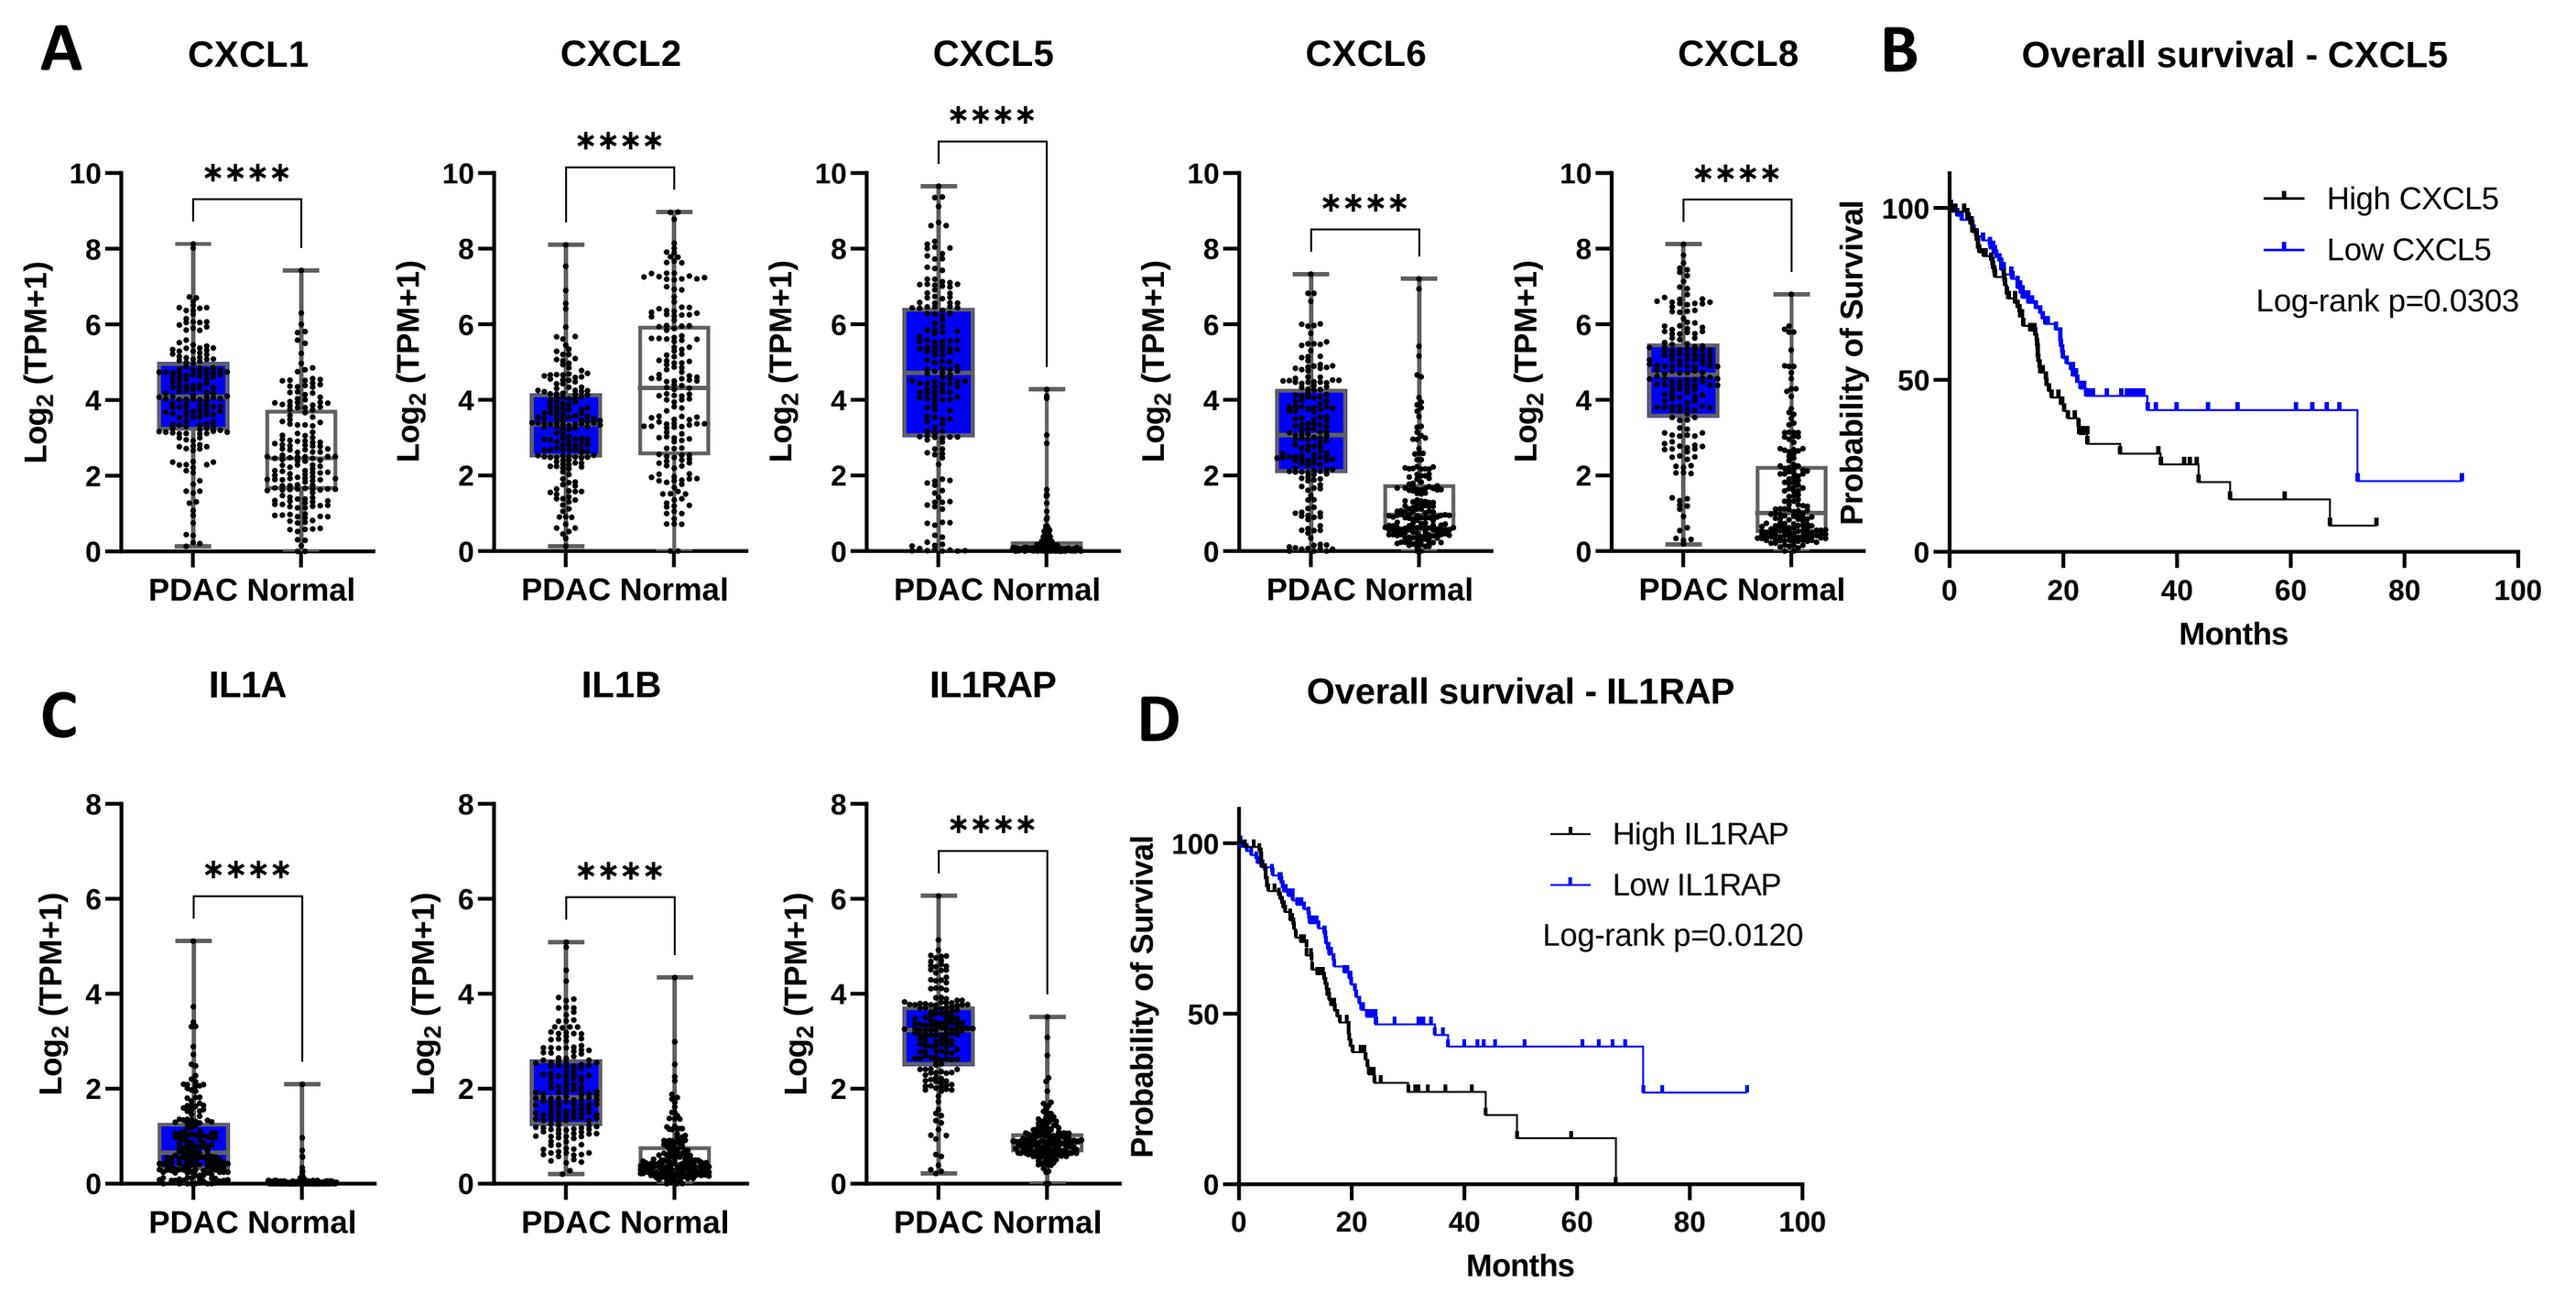
A
CXCL1
CXCL2
CXCL5
CXCL6
CXCL8
Log2 (TPM+1)
PDAC Normal
B
Overall survival - CXCL5
High CXCL5
Low CXCL5
Log-rank p=0.0303
Probability of Survival
Months
C
IL1A
IL1B
IL1RAP
D
Overall survival - IL1RAP
High IL1RAP
Low IL1RAP
Log-rank p=0.0120

Figure 5. (A) RNAseq data from PDAC tumors and normal pancreas from the publicly available databases TCGA and GTEx show an upregulation of the CXCR1/2 ligands CXCL1, CXCL5, CXCL6 and CXCL8 in PDAC. Consistent with the in vitro data (Figure 3 and 4), CXCL5 was the most upregulated gene in PDAC tumors, (B) and a high CXCL5 mRNA expression correlated with shorter overall survival. (C) IL1A, IL1B and IL1RAP mRNA expression was upregulated in PDAC tumors, (D) where high IL1RAP levels correlated with shorter survival.

Serum levels of CXCL1 and/or CXCL5 are reduced in PDAC and NSCLC patients during treatment with nadunolimab and chemotherapy

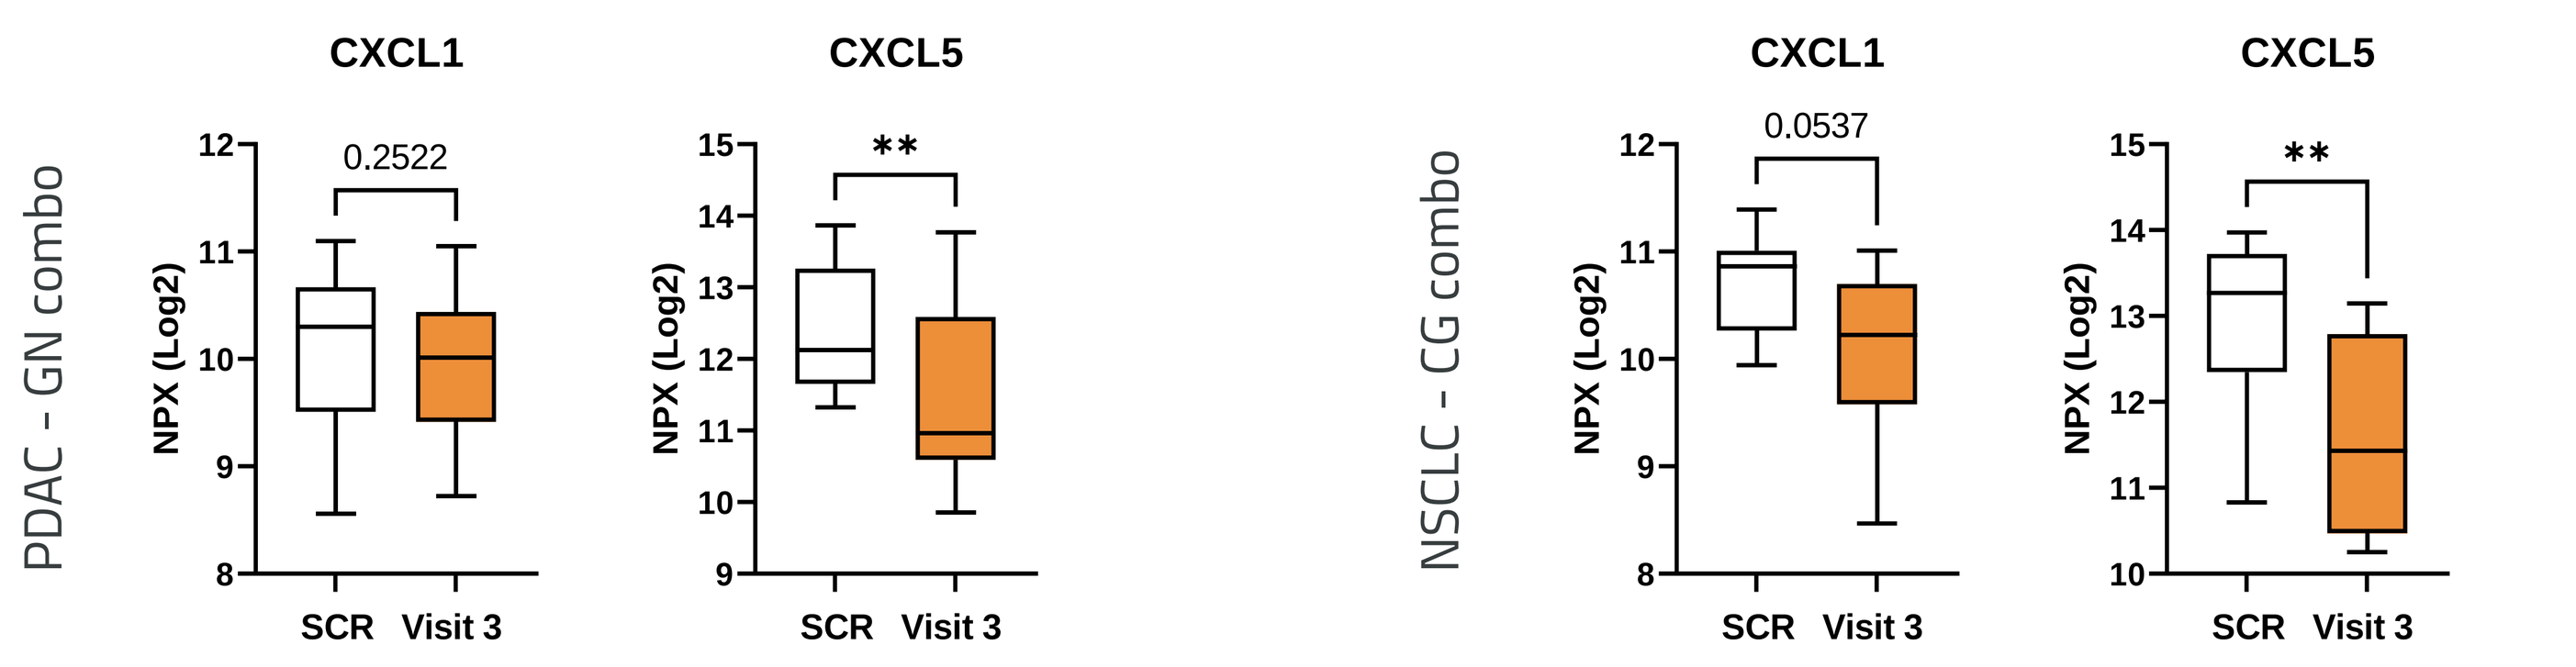
PDAC - GN combo
NSCLC - CG combo
CXCL1
CXCL5
NPX (Log2)
0.2522
0.0537
SCR Visit 3

Figure 6. Serum samples were collected before start of treatment (SCR) and after 2 weeks of treatment (Visit 3) from 16 PDAC patients treated with nadunolimab in combination with GN and from 11 NSCLC patients treated with nadunolimab and GC. Serum levels of CXCL1 and CXCL5 were assessed by PLA (Olink) and data are presented as Normalized Protein expression (NPX) in a Log2 scale. Levels of both chemokines were reduced during treatment with the most pronounced reduction seen for CXCL5.

Conclusions

- IL-1α and IL-1β induced CXCR1/2 ligands in human immune cells and CAFs which was blocked by nadunolimab.
- In an in vitro model of the PDAC TME, PDAC tumor cells direct pancreatic CAFs to secrete CXCR1/2 ligands which was inhibited by nadunolimab.
- Data indicate a strong overexpression of IL1RAP, IL-1α, IL-1β and CXCR1/2 ligands in the PDAC TME.
- CXCL1 and/or CXCL5 were reduced in the serum of both PDAC and NSCLC patients during treatment with nadunolimab and chemotherapy.
- **These data suggest a broad and potent effect of nadunolimab on CXCR1/2 ligands not seen with anti-IL-1β inhibition alone and indicate that nadunolimab treatment may counteract the suppressive TME in part by preventing the influx of myeloid cells.**

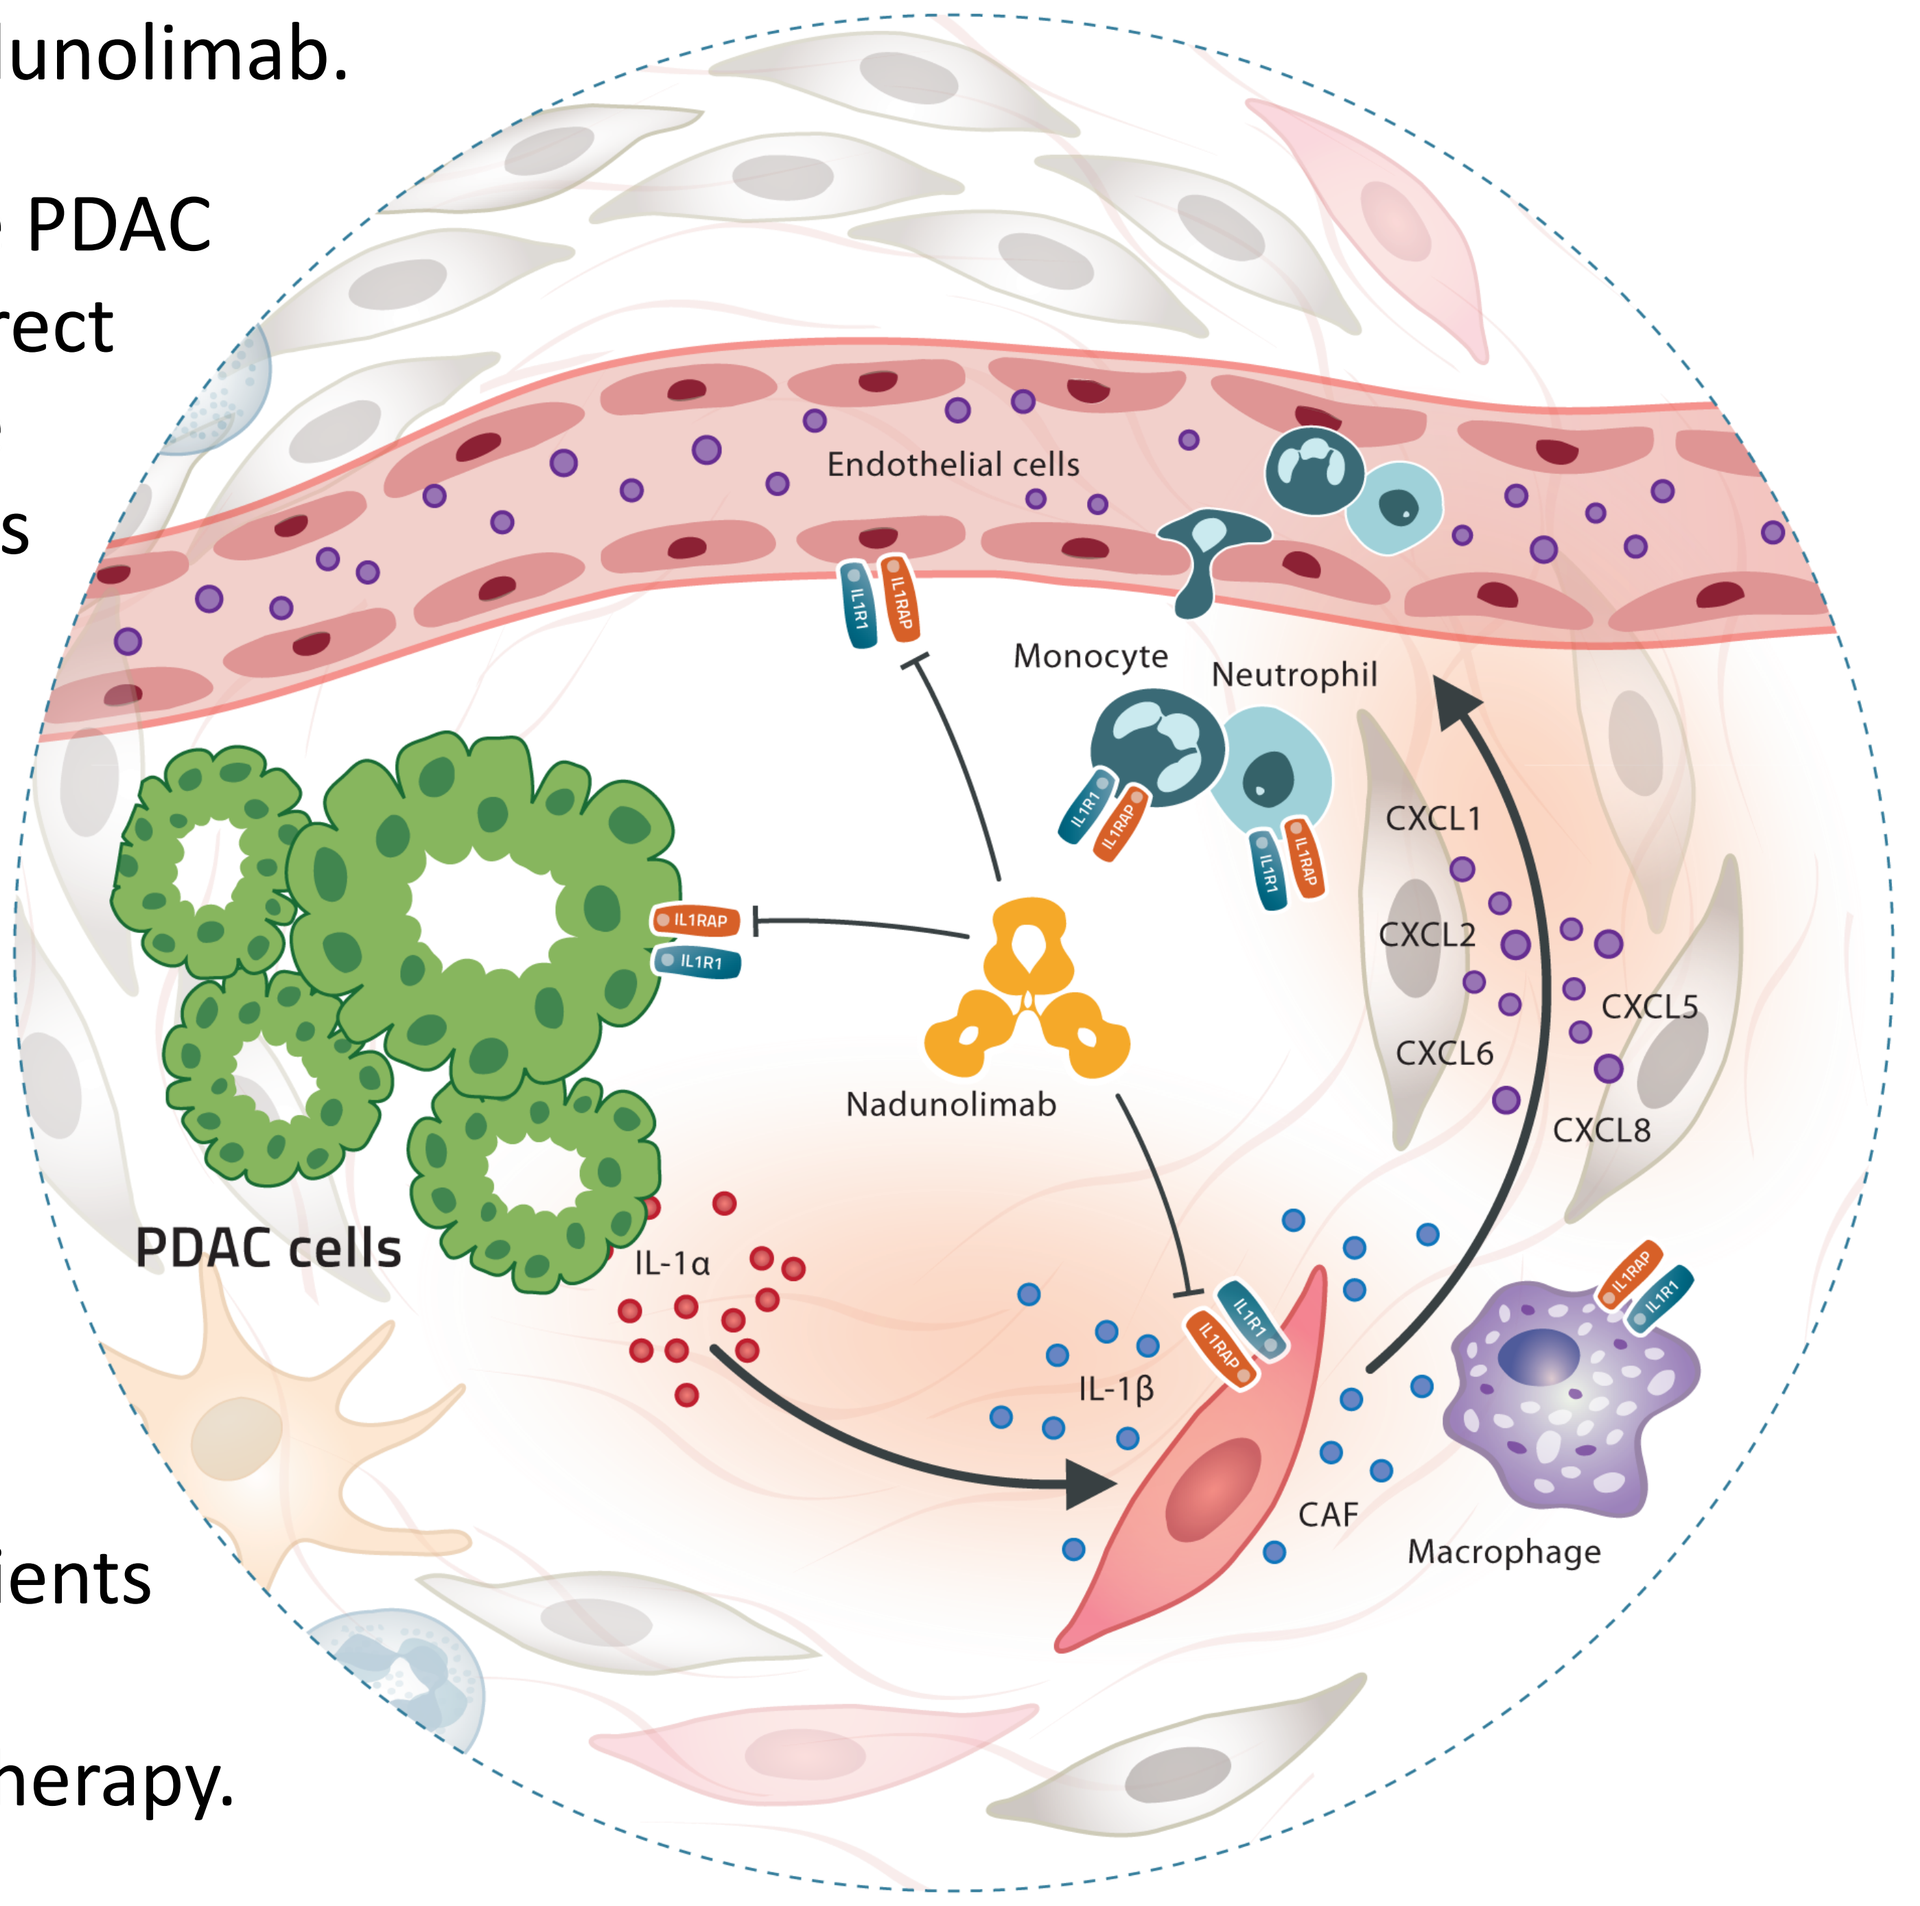
Endothelial cells
Monocyte
Neutrophil
CXCL1
CXCL2
CXCL5
CXCL6
CXCL8
Nadunolimab
PDAC cells
IL-1α
IL-1β
CAF
Macrophage

Acknowledgements

We would like to thank the patients and their families for participating in the study, and all study staff at the clinical sites. This study was sponsored by Cantargia AB.