The CAN04 Antibody Targets IL1RAP and Mediates Tumor Growth Inhibition and Increased Cisplatin Sensitivity in a Patient-Derived Xenograft Model for Non-Small Cell Lung Cancer

David Liberg [1], Camilla Rydberg Millrud [1], Hanna Falk Håkansson [2], Janne Persson [2], Liselotte Larsson[1], Karin von Wachenfeldt [1,2], Göran Forsberg [1]

[1] Cantargia AB, Medicon Village, SE-22381 Lund, Sweden; [2] Truly Labs AB, Medicon Village, SE-223 81 Lund, Sweden

ABSTRACT

Interleukin-1 (IL-1) receptor accessory protein (IL1RAP) is a co-receptor of the IL-1 receptor (IL1R1) and is required for IL-1 signaling. IL1RAP is expressed in various solid tumors and leukemias, both on cancer cells and, in solid tumors, on infiltrating immune cells. We have developed a fully humanized IgG1 antibody (CAN04, nidanilimab) that binds to IL1RAP with high affinity (Kd = 1.10 pM). The antibody is engineered to mediate an enhanced antibody-dependent cellular cytotoxicity (ADCC, EC50 < 1 nM) and to disrupt both IL-1α and IL-1β signaling (IC50 = 3.9 and 4.1 nM respectively). CAN04 is undergoing clinical development[1] in non-small cell lung cancer (NSCLC) and pancreatic cancer, two indications with high expression of IL1RAP and where IL-1 signaling has been proven important for tumor development and for chemotherapy resistance to some regimens. IL-1β blockade also counteracted development of lung cancer in a large clinical trial[2].

Platinum-based chemotherapy is a cornerstone in cancer treatment and used commonly in lung cancer. To investigate effects of IL1RAP targeting in combination with cisplatin we selected a NSCLC patient-derived xenograft model shown by RNAseq to express IL1RAP (log2(FPKM) = 5.0), IL1R1 (log2(FPKM) = 4.7), IL-1α (log2(FPKM) = 4.9) and IL-1β (log2(FPKM) = 3.0). *In vitro*, this model responded to IL-1 stimulation by expression of IL-6 which could be blocked by addition of CAN04 (85-100% reduction at 20 µg/ml). *In vivo*, treatment of subcutaneously inoculated tumors with CAN04 led to a reduced tumor growth and an > 2-fold increase in infiltration of NK1.1$^+$ cells into the tumor, in line with an effect also on ADCC. Interestingly, when combining a fixed dose of CAN04 with cisplatin at 2.5 mg/kg or 5 mg/kg an increased effect could be seen at both doses which was more than additive (from 15% inhibition of either therapeutic to 50 % inhibition of the combination). The combination of CAN04 with cisplatin also drastically reduced cisplatin-related toxicity as demonstrated by a reduced weight loss and increased survival.

In summary, IL1RAP is a novel target in lung cancer for antibody-based therapy and targeting of IL1RAP can block IL-1 mediated inflammatory signaling, reduce tumor growth and synergize with platinum-based chemotherapy to increase the anti-tumor effect and reduce toxicity.

CAN04 SUMMARY

Characteristic	CAN04 Properties
Target	Selective for human IL1RAP
Affinity	1.1 x 10^{-10} M (Biacore binding to IL1RAP)
Antibody subclass	Humanized IgG1, low fucose ADCC enhanced (BioWa Potelligent®)

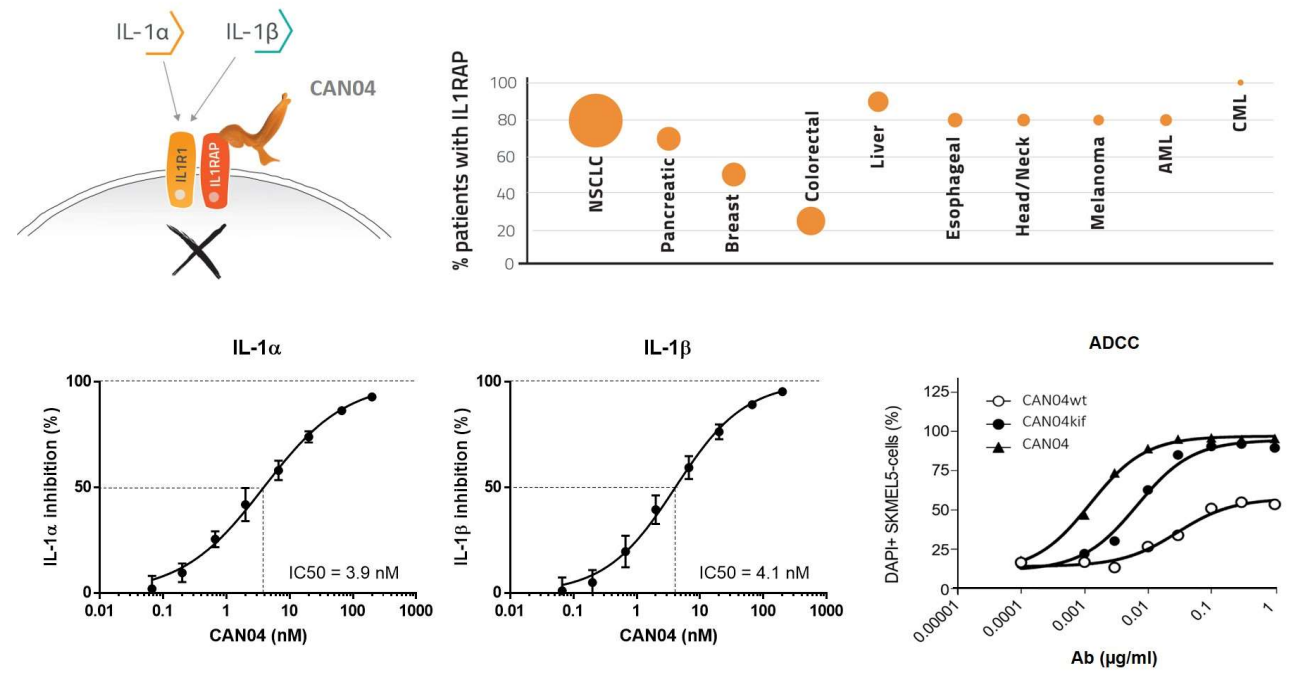
IL-1α
IL-1β
CAN04
IL1R1
IL1RAP
% patients with IL1RAP
NSCLC
Pancreatic
Breast
Colorectal
Liver
Esophageal
Head/Neck
Melanoma
AML
CML
IL-1α
IL-1α inhibition (%)
IC50 = 3.9 nM
CAN04 (nM)
IL-1β
IL-1β inhibition (%)
IC50 = 4.1 nM
CAN04 (nM)
ADCC
DAPI+ SKMEL5-cells (%)
CAN04wt
CAN04kif
CAN04
Ab (µg/ml)

CAN04 targets IL1RAP, blocks signaling from IL1α and IL1β and induces ADCC of IL1RAP-expressing cells. Upper left: IL1RAP associates with IL1R1 to allow IL-1α and IL-1β signaling, CAN04 binds to IL1RAP and blocks IL1RAP function. Upper right: IL1RAP is commonly expressed in solid and hematological cancers. Staining of frozen tissue sections from patient samples of the specified indications (n ranging from 14 to 46) with CAN04. Lower left panels: CAN04 inhibits both IL-1α and IL-1β signaling. Lower right panel: NK cell mediated ADCC is induced by CAN04wt (wt glycosylation), CAN04kif (reduced fucose by expression in presence of kifunensine) or CAN04 (nidanilimab/clinical product, low fucose CAN04 stably produced in Potelligent® CHO cells). Lowering of fucose content increases ADCC > 30-fold.

STUDY OBJECTIVE

- To investigate the potential of combining IL1RAP targeting by CAN04 (nidanilimab) and cisplatin therapy.

RESULTS

IL-1α and IL-1β are present in human lung cancer

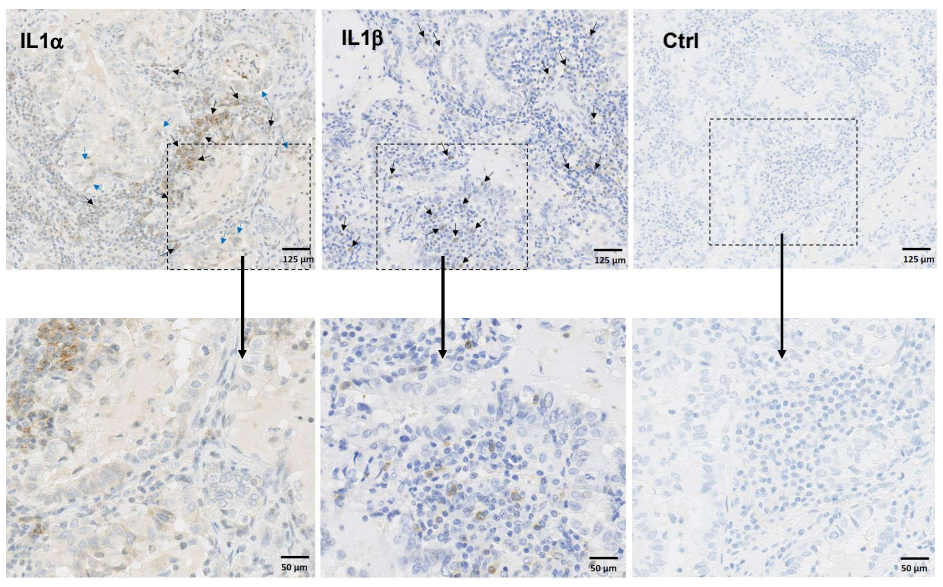
IL1α
IL1β
Ctrl

IL-1α is expressed by infiltrated leukocytes and tumor cells while IL-1β is expressed by infiltrated leukocytes in NSCLC tumors. Eight NSCLC tumors were stained for the expression of IL-1α, IL-1β or antibody diluent as negative control. Left panels: In all tumors, IL-1α-positive leukocytes and weak IL-1α expressing tumor cells were found. Middle panels: The IL-1β staining was foremost found on infiltrated leukocytes. In two out of eight NSCLC tumors, a weak expression of IL-1β was also detected on tumor cells. Right panels: negative control.

CAN04 inhibits IL-1 signaling and reduces tumor growth of an IL1RAP$^+$ NSCLC patient-derived xenograft model (LU2503)

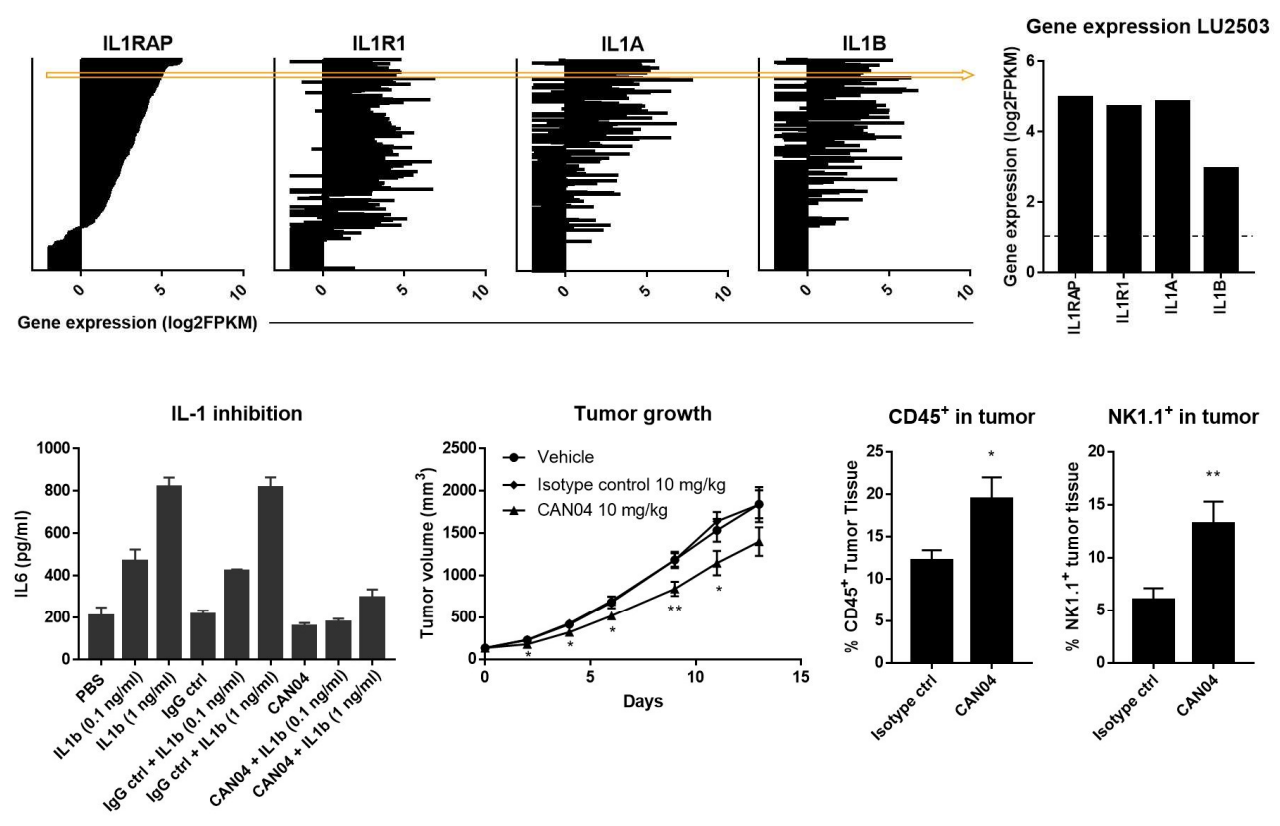
IL1RAP
IL1R1
IL1A
IL1B
Gene expression (log2FPKM)
Gene expression LU2503
Gene expression (log2FPKM)
IL-1 inhibition
IL6 (pg/ml)
PBS
IL1b (0.1 ng/ml)
IL1b (1 ng/ml)
IgG ctrl
IgG ctrl + IL1b (0.1 ng/ml)
IgG ctrl + IL1b (1 ng/ml)
CAN04
CAN04 + IL1b (0.1 ng/ml)
CAN04 + IL1b (1 ng/ml)
Tumor growth
Vehicle
Isotype control 10 mg/kg
CAN04 10 mg/kg
Tumor volume (mm3)
Days
CD45+ in tumor
% CD45+ Tumor Tissue
NK1.1+ in tumor
% NK1.1+ tumor tissue
Isotype ctrl
CAN04

CAN04 blocks IL-1 signaling and inhibits tumor growth in the LU2503 NSCLC patient-derived xenograft (PDX) expressing IL1RAP, IL1R1, IL-1α and IL-1β. Upper panels: RNA-seq gene expression data from 300 lung cancer PDX models were obtained from CrownBio huBase®. The PDX models are sorted for IL1RAP expression, the orange arrow and the right panel show IL1RAP, IL1R1, IL-1α and IL-1β expression in the LU2503 NSCLC PDX model. Lower left panels: CAN04 inhibits the IL-1β induced IL-6 production from LU2503 cells *in vitro* and inhibits the growth of LU2503 *in vivo*. Lower right panels: CAN04 treatment leads to an increase in tumor-infiltrating CD45+ and NK1.1+ cells (measured by IHC on tumor cryosections).

CAN04 increases cisplatin sensitivity and reduces toxicity in the LU2503 PDX model

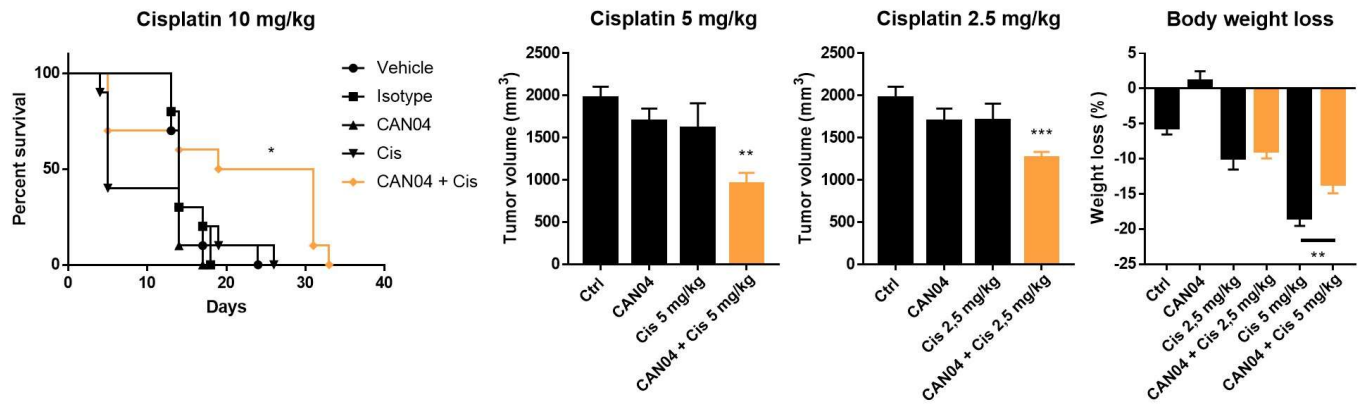
Cisplatin 10 mg/kg
Percent survival
Days
Vehicle
Isotype
CAN04
Cis
CAN04 + Cis
Cisplatin 5 mg/kg
Tumor volume (mm3)
Ctrl
CAN04
Cis 5 mg/kg
CAN04 + Cis 5 mg/kg
Cisplatin 2.5 mg/kg
Cis 2.5 mg/kg
CAN04 + Cis 2.5 mg/kg
Body weight loss
Weight loss (%)

The combination of CAN04 and cisplatin leads to increased efficacy and reduced toxicity. Left panel: Combination of CAN04 and 10 mg/kg cisplatin increases survival compared to cisplatin alone. Middle panels: Combining CAN04 with 2.5 or 5 mg/kg cisplatin leads to reduced tumor growth (tumor volumes at day 12 are shown). Right panel: Reduced toxicity was observed when cisplatin was combined with CAN04, the graph shows maximum body weight loss which occurred at day 10. In general, cisplatin treatment at 10 and 5 mg/kg induced toxicity and mice were terminated according to the left panel in the 10 mg/kg group and at around day 12 in the 5 mg/kg group (50 % of the mice for cisplatin alone compared to 20% of the combination or vehicle).

Targeting both tumor and endogenous IL1RAP in a syngeneic tumor model strengthens the combination effects

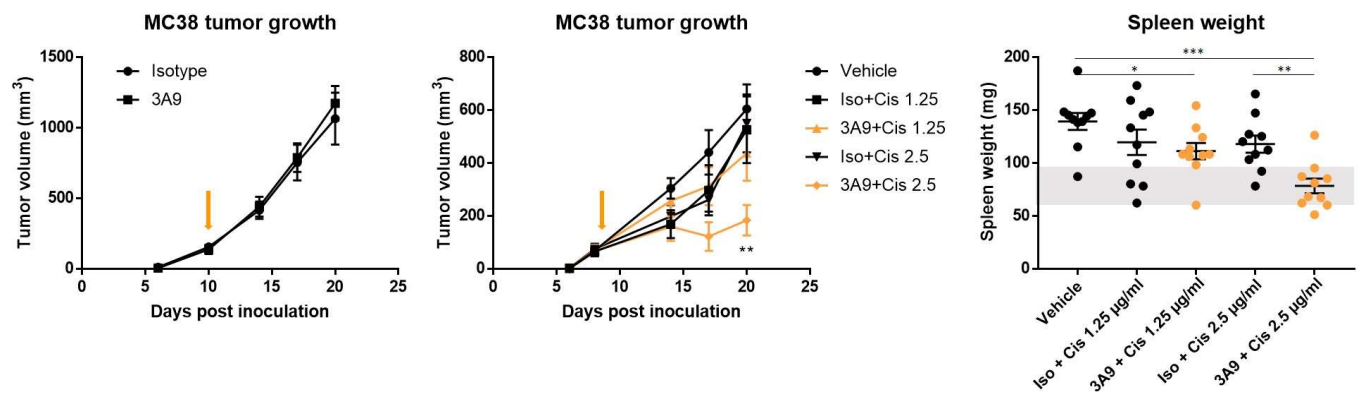
MC38 tumor growth
Isotype
3A9
Tumor volume (mm3)
Days post inoculation
MC38 tumor growth
Vehicle
Iso+Cis 1.25
3A9+Cis 1.25
Iso+Cis 2.5
3A9+Cis 2.5
Spleen weight
Spleen weight (mg)
Vehicle
Iso + Cis 1.25 µg/ml
3A9 + Cis 1.25 µg/ml
Iso + Cis 2.5 µg/ml
3A9 + Cis 2.5 µg/ml

Treatment of syngeneic MC38 tumors with a CAN04 mouse surrogate antibody (3A9) and cisplatin show synergistic effects on tumor growth. Left panel: 3A9 alone show little or no effect on the growth of established MC38 tumors. Arrow=start of treatment. Middle panel: Combining anti-IL1RAP targeting (3A9) with cisplatin provides increased efficacy. Arrow=start of treatment. Right panel: Splenomegaly is frequently observed in tumour models, and a result of pro-inflammatory effects exhibited by the tumor on the immune system. 3A9 in combination with cisplatin normalized the tumor induced increase in spleen size. Grey area indicates normal speen weight in C57Bl/6 animals.

CONCLUSIONS

- CAN04 (nidanilimab) is a humanized, ADCC-enhanced, IgG1 antibody targeting IL1RAP and thereby IL-1 signaling.
- IL1RAP, IL-1α and IL-1β are all present in human lung cancer.
- CAN04 treatment inhibits IL-1β mediated effects and tumor growth in an IL1RAP$^+$ NSCLC PDX model.
- The CAN04/cisplatin combination has improved efficacy to either treatment alone and reduced toxicity compared to cisplatin only.
- Combination effects are even stronger in a syngeneic model, in line with both a tumor cell intrinsic effect of IL1RAP targeting and an effect on the tumor microenvironment.

REFERENCES

1. The CANFOUR study, ClinicalTrials.gov NCT03267316
2. Ridker et al., Lancet 2017

cantargia