The CAN04 Antibody Targets IL1RAP and Inhibits Tumor Growth in a PDX Model for NSCLC

David Liberg [1]; Wondossen Sime [2]; Matteo Riva [3]; Ramin Massoumi [2]; Göran Forsberg [1]; Karin von Wachenfeldt [1,3]

[1] Cantargia AB, Medicon Village, SE-22381 Lund, Sweden; [2] Division of Translational Cancer Research, Department of Laboratory Medicine, Lund University, Medicon Village, SE-22381 Lund, Sweden; [3] Truly Labs AB, Medicon Village, SE-223 81 Lund, Sweden

ABSTRACT

Interleukin-1 receptor associated protein (IL1RAP) is a co-receptor of the IL-1 receptor (IL1R1) and the IL-33 receptor (ST2) and is required for signaling through both receptor complexes. We have previously described IL1RAP as a cancer target on leukemic cells and shown that antibodies directed against IL1RAP block leukemic cell proliferation and mediate antibody-dependent cellular cytotoxicity (ADCC). In the present study, we show that IL1RAP is expressed at high levels in various solid tumors, including non-small cell lung cancer (NSCLC) and pancreatic cancer. IL1RAP-expressing solid tumor cell lines respond to IL-1 by NFkB-activation and production of IL-6 and IL-8. This result is in line with the function of IL-1 as a promoter of tumor inflammation and tumor progression. In order to target IL1RAP in both solid tumors and hematological malignancies, we have developed a fully humanized, ADCC-enhanced, IgG1 antibody (CAN04) that binds to IL1RAP with high affinity. The binding mode allows both disruption of IL-1 and IL-33 mediated NFkB activation and targeting of IL1RAP-expressing cells for ADCC. As a consequence, CAN04 reduces inflammatory cytokine production of tumor cell lines and inhibits growth of a patient-derived IL1RAP-expressing NSCLC xenograft in mice. In conclusion, targeting of IL1RAP with the CAN04 antibody can reduce tumor promoting inflammation, target tumor cells for ADCC and inhibit tumor growth in vivo.

CAN04 SUMMARY

Characteristic	CAN04 Properties
Target	Selective for human IL1RAP
Affinity	1.1 x 10^{-10} M (Biacore binding to IL1RAP)
Antibody subclass	Humanized IgG1, low fucose ADCC enhanced (BioWa Potelligent®)
Effects (*in vitro*):	Inhibition of IL-1α induced signaling (EC50 = 4 nM)
	Inhibition of IL-1β induced signaling (EC50 = 2 nM)
	Inhibition* of IL-33 induced signaling (EC50 = 3 nM)
	ADCC induction (EC50 = 0.1 nM)
Effects (*in vivo*):	Highly effective in xenograft models of AML and CML

*Partial

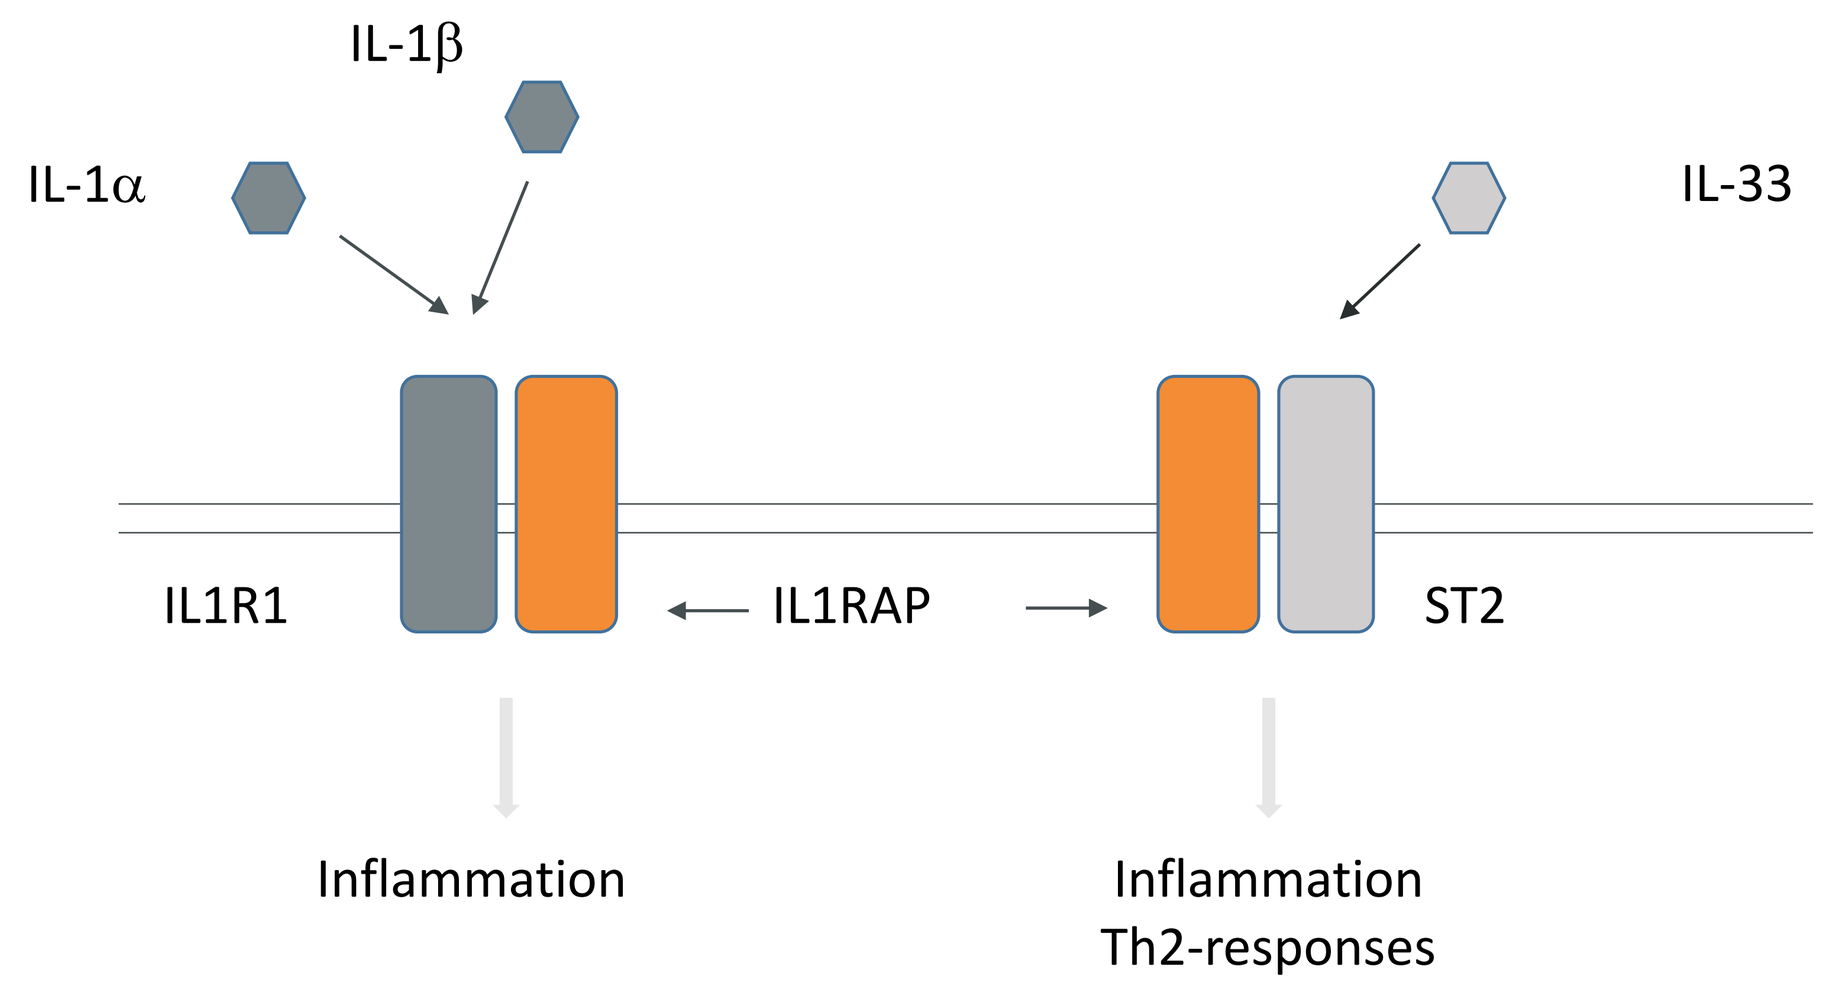
IL-1β
IL-1α
IL-33
IL1R1
IL1RAP
ST2
Inflammation
Inflammation
Th2-responses

IL1RAP binds IL1R1 and ST2 and is involved in signaling from IL1α, IL1β and IL33. Schematic representation of IL1RAP interactions with IL1R1 and ST2.

STUDY OBJECTIVES

- To investigate IL1RAP expression in human solid tumors
- To study CAN04 effects on IL1RAP⁺ solid tumor cells *in vitro* and *in vivo*

RESULTS

IL1RAP is expressed in multiple solid tumor types

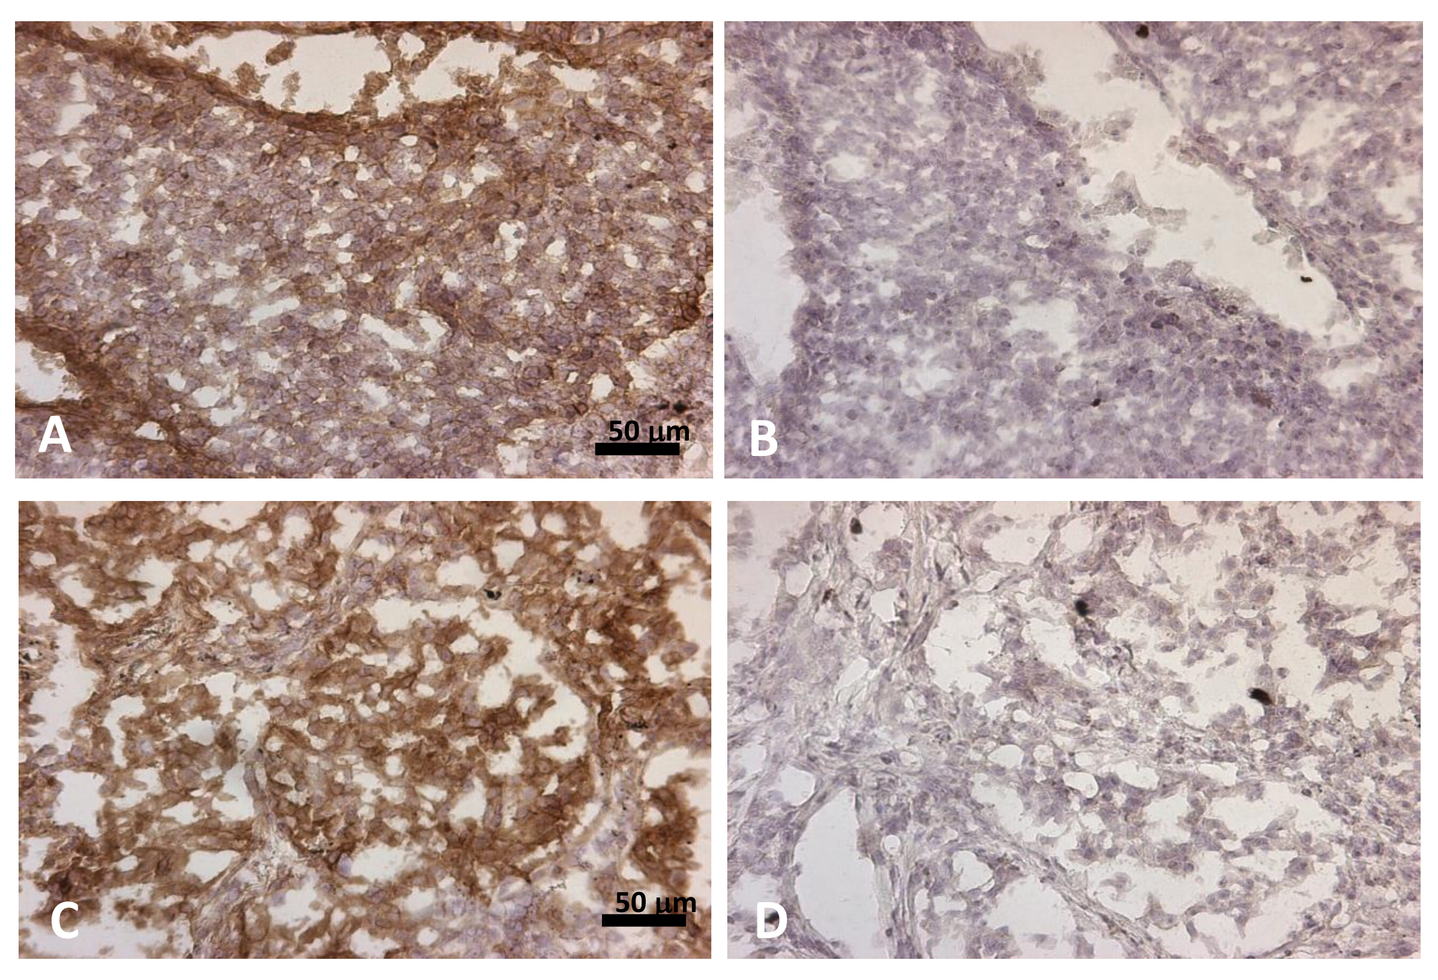
A
B
C
D
50 µm
50 µm

Tumor type	Positive tumor staining	% Positive tumors
Lung (NSCLC)	39/46	85%
Pancreatic	12/14	86%
Melanoma	13/15	87%
Breast	23/44	52%
Colon	10/37	27%

IL1RAP is expressed on a number of different solid tumor types, including NSCLC and Pancreatic tumors. CAN04 has been used to screen a panel of solid tumor specimens as summarized in the table. In lung cancer the percentage of positive tumors exceeds 80%, and include both squamous and adeno carcinomas. All positive tumors were 2+ or 3+. Panel A and B show cryosections from a squamous cell NCSLC whereas panel C and D represent adenocarcinomas. Panels A and C were stained with 1 µg/ml CAN04 and B and D with 1µg/ml isotype control antibody.

CAN04 blocks IL1-induced IL-6 and IL-8 release and induce ADCC of IL1RAP⁺ solid tumor cell lines

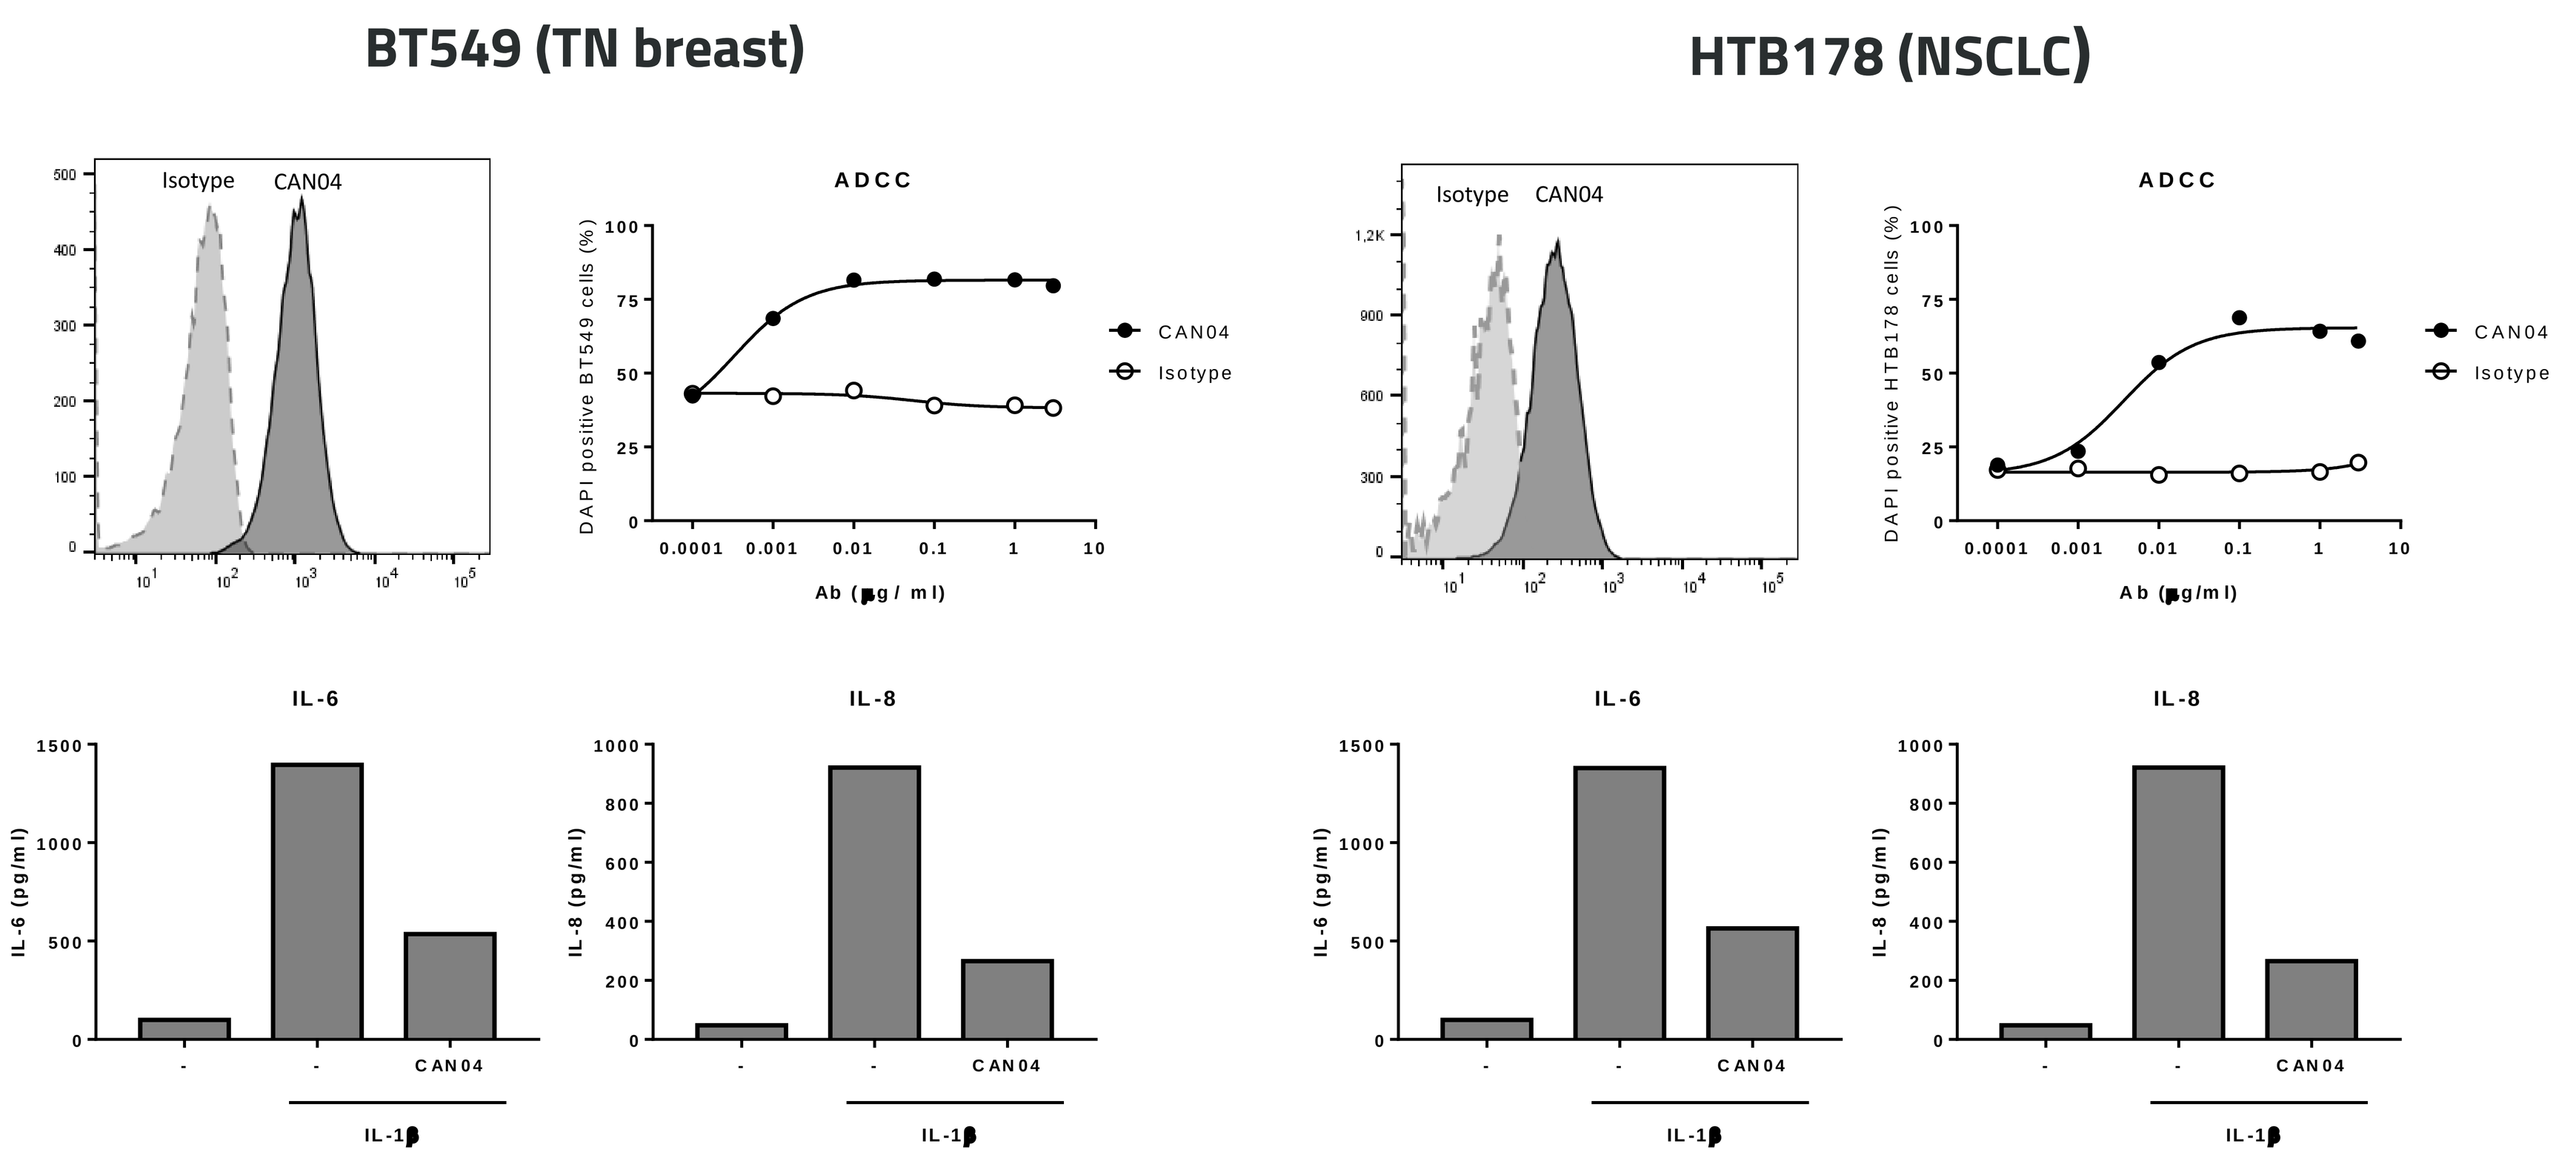
BT549 (TN breast)
HTB178 (NSCLC)
Isotype
CAN04
ADCC
DAPI positive BT549 cells (%)
DAPI positive HTB178 cells (%)
Ab (µg/ml)
CAN04
Isotype
IL-6
IL-8
IL-6 (pg/ml)
IL-8 (pg/ml)
CAN04
IL-1β

***In vitro* effects of CAN04 on two IL1RAP⁺ solid tumor cell lines, BT549 (triple negative breast cancer) and HTB178 (NSCLC)**. Upper panels: FACS analysis of IL1RAP expression using CAN04-PE and ADCC induced by CAN04 or isotype control. Lower panels: Inhibition of IL-1β induced IL-6 or IL-8 release by CAN04 (20 µg/ml) after stimulation with 0.3 ng/ml IL-1β o/n in the presence or absence of CAN04, IL-6 and IL-8 release was measured by ELISA (R&D DuoSet).

CAN04 inhibits growth of an IL1RAP⁺ NSCLC patient-derived xenograft (PDX)

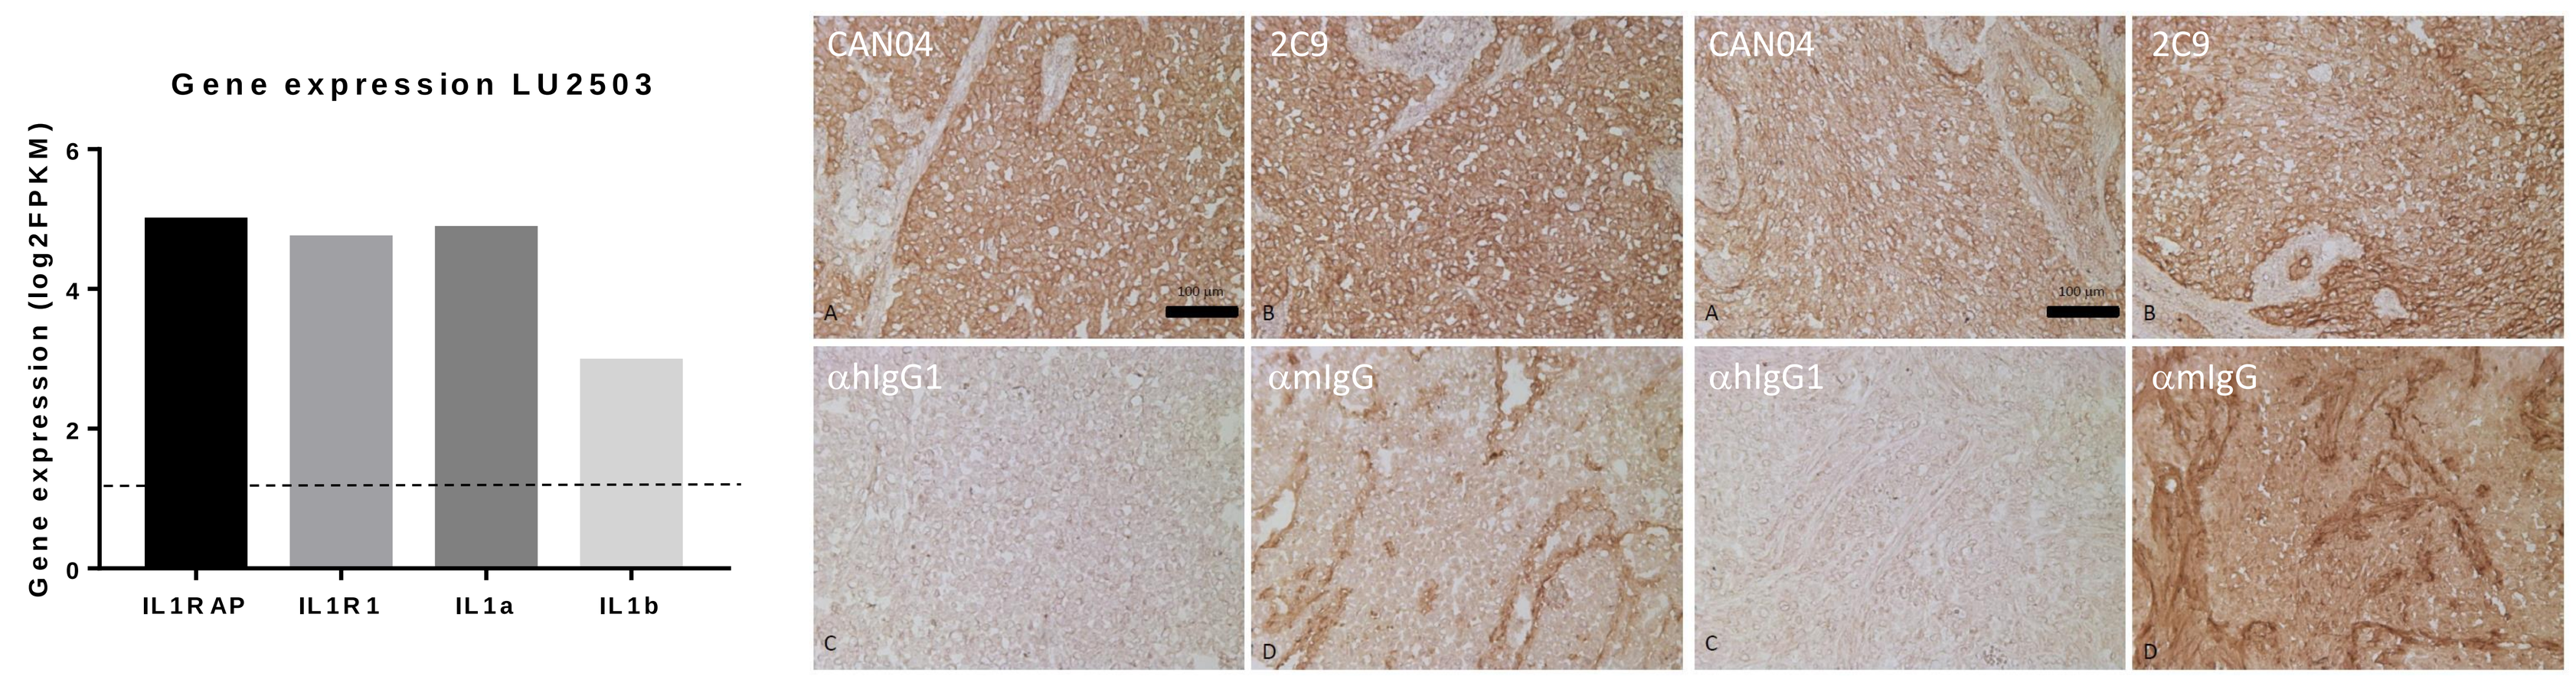
Isotype treated
CAN04 treated
Gene expression LU2503
Gene expression (log2FPKM)
IL1RAP
IL1R1
IL1a
IL1b
CAN04
2C9
αhIgG1
αmIgG

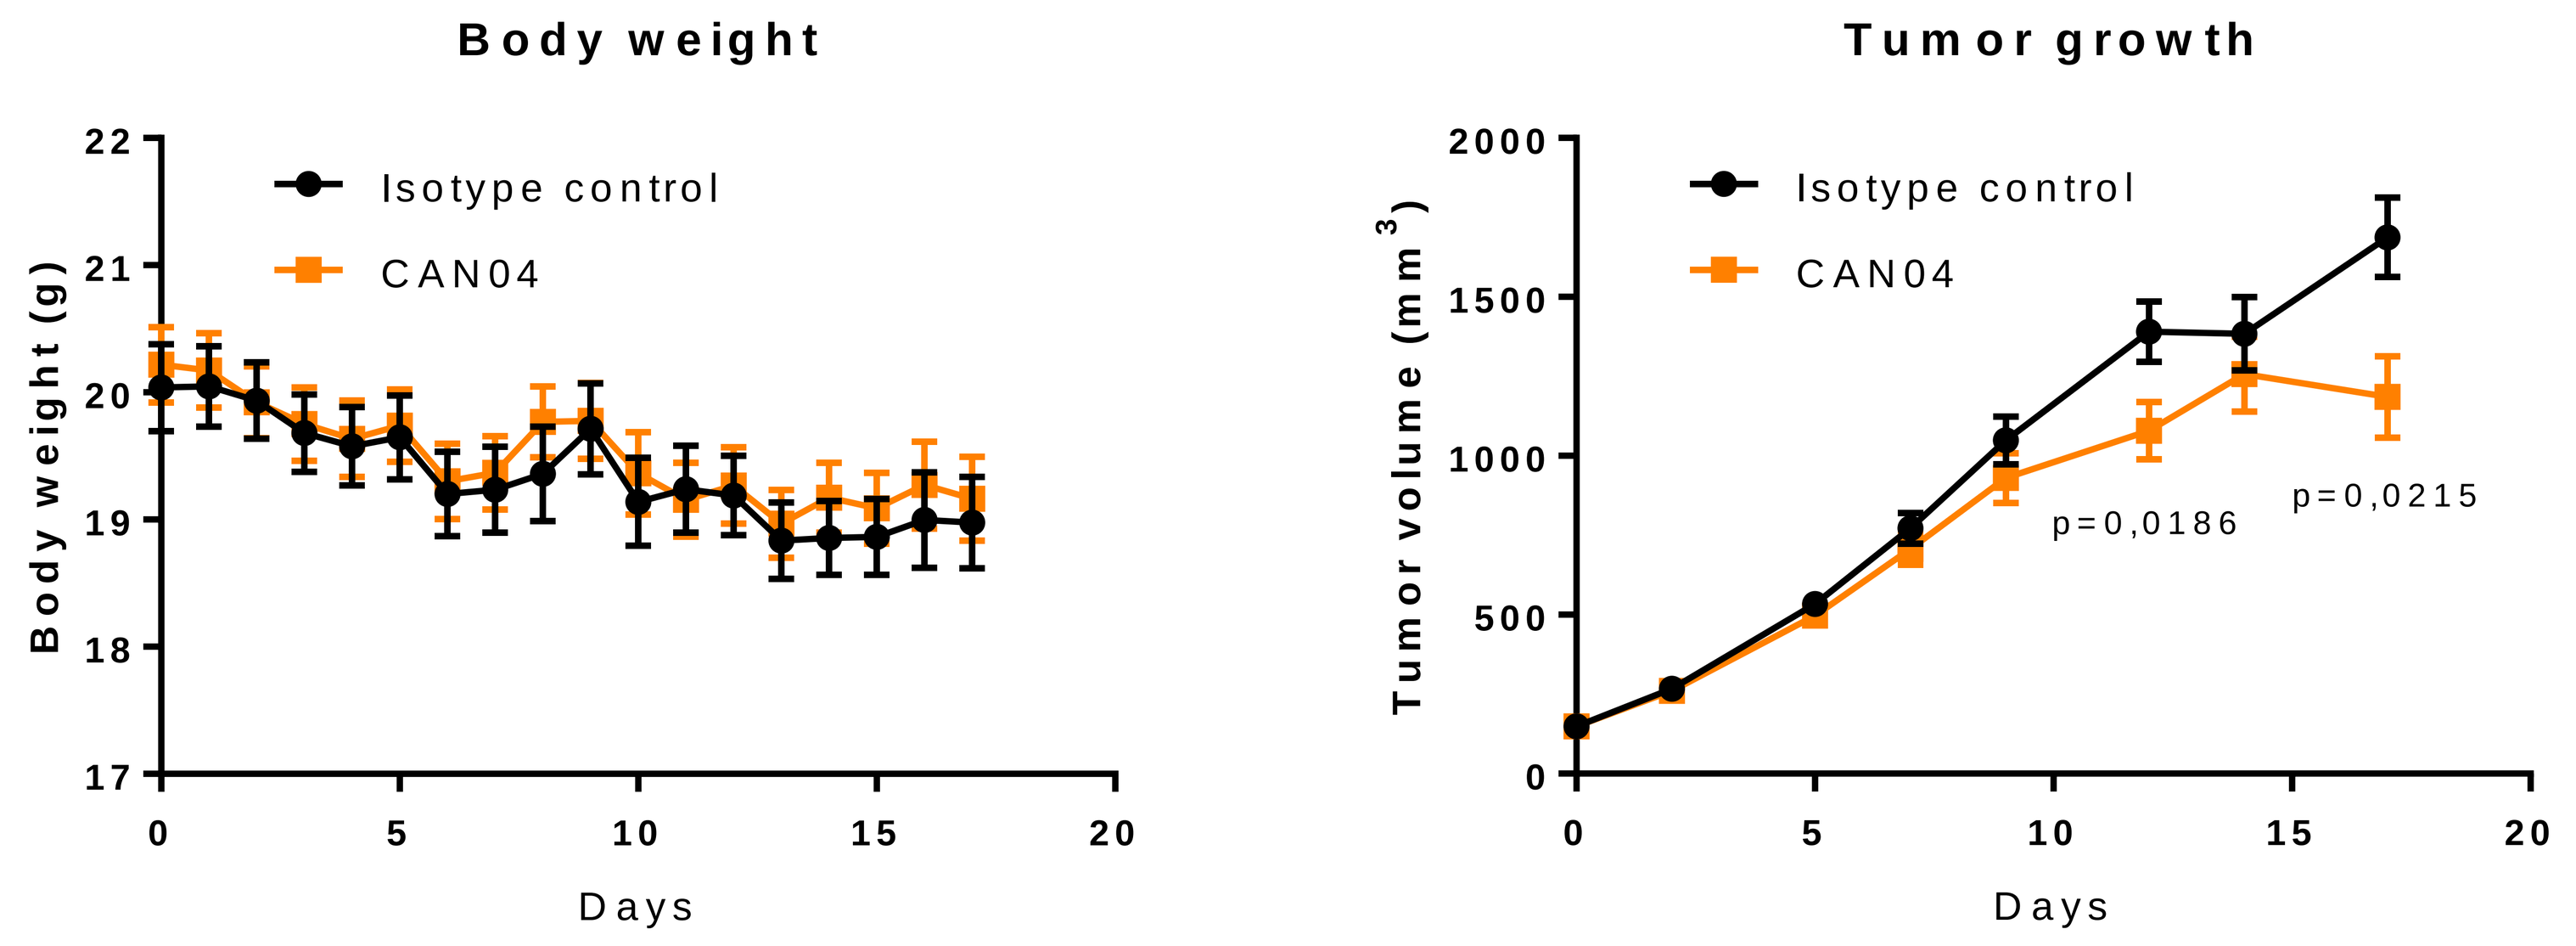
Body weight
Body weight (g)
Isotype control
CAN04
Days
Tumor growth
Tumor volume (mm³)
p=0,0186
p=0,0215

The LU2503 NSCLC patient-derived xenograft expresses IL1RAP, IL1R1, IL-1α and IL-1β, administration of CAN04 targets the antibody to the tumor and inhibits tumor growth. Upper left panel: RNA-seq gene expression data from CrownBio huBase®. Upper middle and right panels: IHC with the indicated antibodies on tumor cryosections. CAN04 and 2C9 targets non-overlapping IL1RAP epitopes, the CAN04 used for treatment was in mIgG2a format. Lower panels: CAN04 treatment inhibits growth of the LU2503 PDX (CrownBio) in nude mice (n=26 mice/group) without having adverse effects on body weight.

CAN04 treatment increases the stromal compartment of the tumor and induces prominent leukocyte infiltration

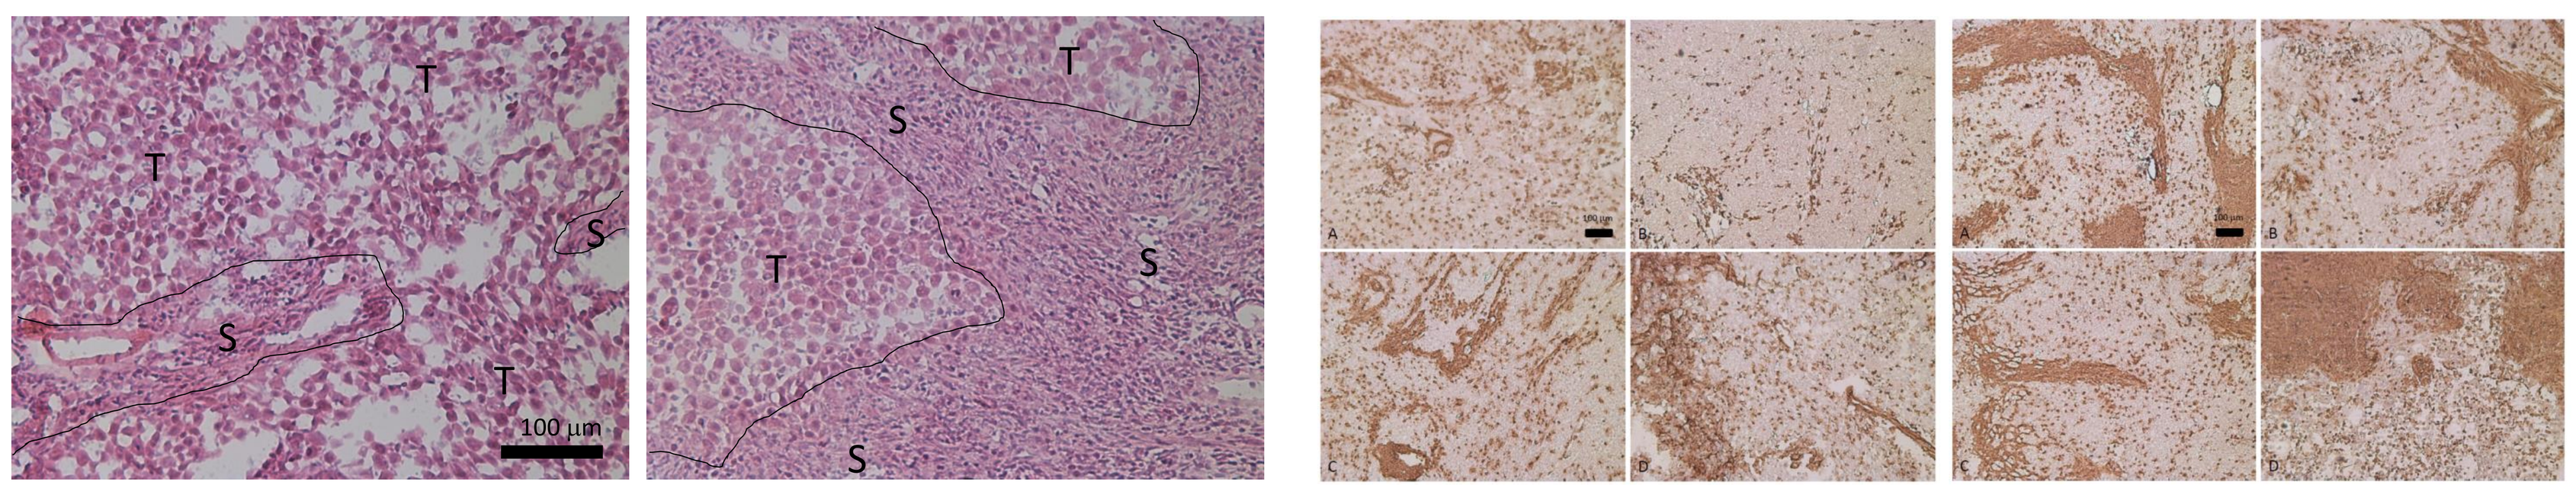
Isotype treated
CAN04 treated
Isotype treated
CAN04 treated
T
S
100 µm

Image analysis:	Isotype	CAN04
CD45⁺	14.9 %	24.3%
Stroma (H/E)	12 %	34%

Treatment of LU2503 with CAN04 targets increases the stromal tumor compartment relative to the tumor cell part and increases leukocyte infiltration into the tumor. Left panels: Hematoxylin/Eosin staining of representative isotype control and CAN04 treated tumors, stroma (S) and tumor cell (T) regions are indicated. Right panels: CD45 staining of four representative isotype treated or CAN04 treated tumors. The bottom table shows the fraction of stromal and CD45⁺ tumor region of isotype or CAN04 treated tumors (n=5 of each).

CONCLUSIONS

- IL1RAP is expressed on several different solid tumors.
- CAN04 inhibits IL-1β induced IL-6 and IL-8 release and induces ADCC of IL1RAP⁺ solid tumor cells.
- CAN04 treatment inhibits NSCLC tumor growth in a PDX model
- CAN04 treated tumors have an increased leukocyte infiltration and a reduced fraction of cancer cells in the tumor

REFERENCES

1. Järås et al., Isolation and killing of candidate chronic myeloid leukemia stem cells by antibody targeting of IL-1 receptor accessory protein, PNAS 2010
2. Ågerstam *et al.*, Antibodies targeting human IL1RAP (IL1R3) show therapeutic effects in xenograft models of acute myeloid leukemia, PNAS 2015
3. Ågerstam *et al.*, IL1RAP antibodies block IL1-induced expansion of candidate CML stem cells and mediate cell killing in xenograft models, Blood 2016

Lund University
cantargia