Phase Ib safety and efficacy of nadunolimab/gemcitabine/carboplatin (NadGC) in metastatic triple negative breast cancer (mTNBC)

Sara López-Tarruella[1,2,3]; Agostina Stradella[4,3]; Pablo Tolosa[5,3]; Silvia Antolin[6,3]; Marta Santisteban[7,8,3]; Javier García-Corbacho[9,3]; Angel Luis Guerrero-Zotano[10,3]; Manuel Ruiz-Borrego[11,3]; María M Caffarel[12,13,3]; Nedjad Losic[14]; Elin Jaensson Gyllenbäck[14]; Petter Skoog[14]; Helén Johansen Blanco[14]; Juan José Soto-Castillo[4]; Manuel Alva[5]; Cristina Reboredo[6]; Maribel Casas[3]; Nuria Caparroso[3]; Susana Bezares[3]; Ignacio Garcia-Ribas[14]

[1]Medical Oncology Service, Hospital General Universitario Gregorio Marañón, Madrid, Spain; [2]Instituto de Investigación Sanitaria Gregorio Marañón (IiSGM),Madrid, Spain; [3]GEICAM Spanish Breast Cancer Group, Madrid, Spain; [4]Institut Catala D'Oncologia-Duran y Reynals (ICO L'Hospitalet), Barcelona, Spain; [5]Hospital Universitario 12 de Octubre, Madrid, Spain; [6]Complejo Hospitalario Universitario A Coruña, A Coruña, Spain; [7]Clínica Universidad de Navarra, Pamplona, Spain; [8]Instituto de Investigación Sanitaria de Navarra (IdiSNA), Pamplona, Spain; [9]UGCI Oncología Médica, Hospitales Universitarios Regional y Virgen de la Victoria, Málaga, Spain; [10]Instituto Valenciano de Oncologia (IVO), Valencia, Spain; [11]Hospital Universitario Virgen del Rocío, Sevilla, Spain; [12] Biodonostia Health Research Institute, San Sebastián, Spain; [13]IKERBASQUE, Basque Foundation for Science, Bilbao, Spain; [14]Cantargia AB, Lund, Sweden

406P

Background

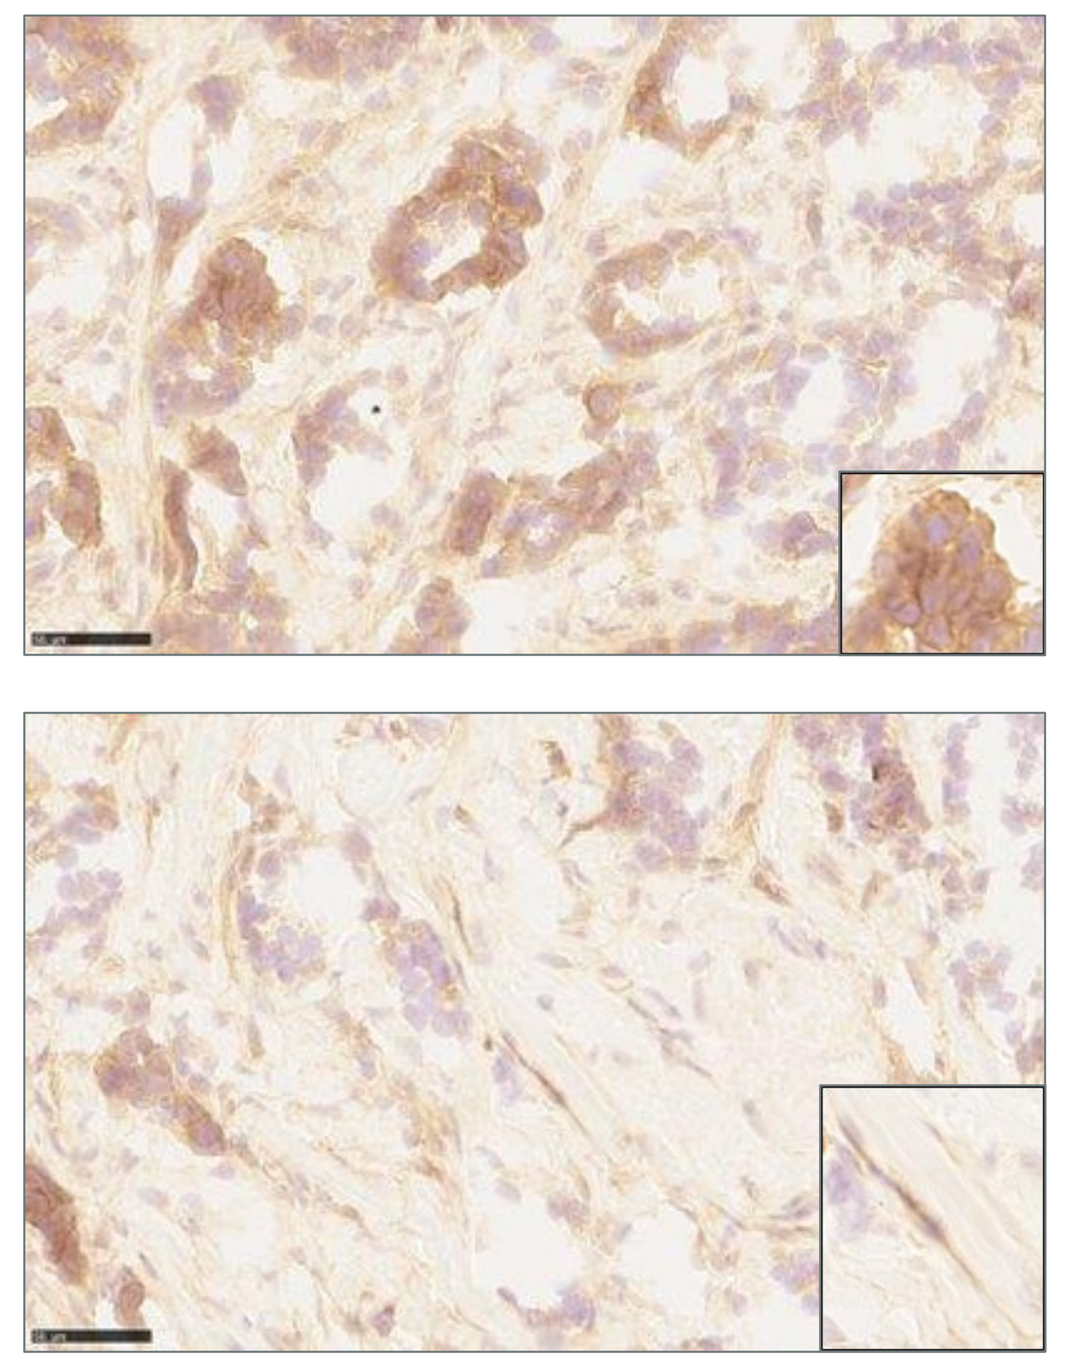

Figure 1: *Immunohistochemistry staining of IL1RAP on tumor cells (top) and fibroblasts (bottom) in TNBC tumor biopsies*

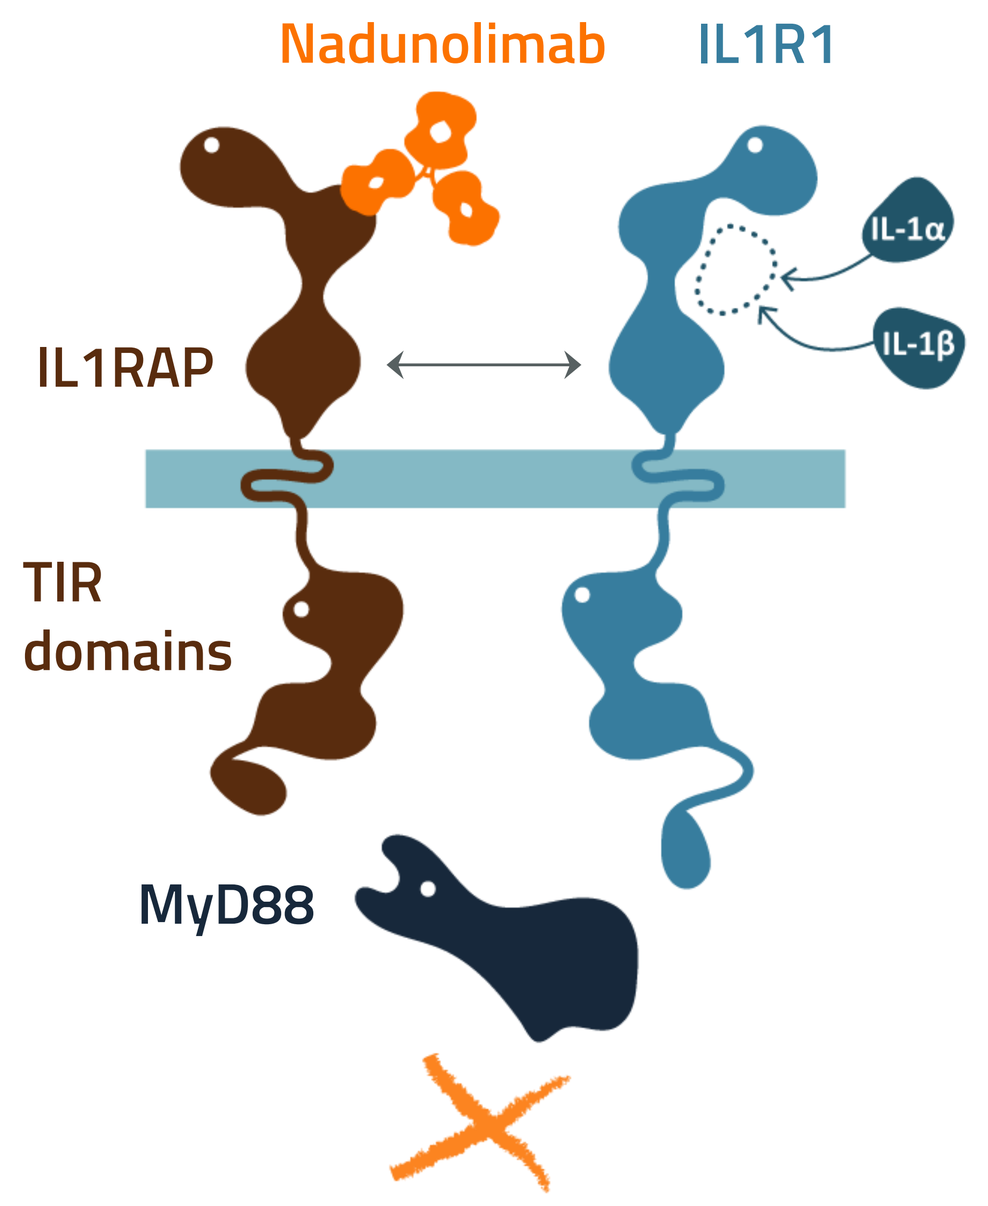
Nadunolimab
IL1R1
IL-1α
IL-1β
IL1RAP
TIR domains
MyD88

Figure 2: *Mode-of-action of nadunolimab*

- Interleukin-1 Receptor Accessory Protein (IL1RAP) is expressed on cancer, stromal and infiltrating immune cells of many solid tumors. Among breast cancer subtypes, triple-negative breast cancer (TNBC) has the highest IL1RAP expression. IL-1α and IL-1β modulate tumor-promoting factors via IL-1 receptor type 1 (IL-1R1), which requires IL1RAP.
- Chemotherapy leads to upregulation of IL-1α which stimulates IL-1β release by stromal cells[1-3]. IL-1α/IL-1β contribute to tumor growth, chemoresistance and immune suppression[4-5]. Blockade of both IL-1α/IL-1β in combination with chemotherapy thus constitutes an attractive approach for cancer treatment.
- Nadunolimab (CAN04) is a fully humanized mono-clonal IgG1 antibody targeting IL1RAP. It inhibits tumor-promoting and chemoresistance signals mediated by IL-1α and IL-1β and induces antibody-dependent cellular cytotoxicity of IL1RAP-expressing cells.
- Interim results for nadunolimab combination with chemotherapy in pancreatic cancer and non-small cell lung cancer patients from the phase I/IIa trial CANFOUR (NCT03267316) showed acceptable safety and promising efficacy compared to historical data. In pancreatic cancer, the strongest clinical benefit was observed in patients with high tumor baseline expression of IL1RAP[6].
- TRIFOUR (NCT05181462) is a phase Ib/non-comparative randomized phase II trial to evaluate nadunolimab + gemcitabine + carboplatin (NadGC) combination in patients with metastatic TNBC (mTNBC). **Herein, preliminary safety and efficacy from the phase Ib dose escalation are presented.**

Study design

TRIFOUR: Phase Ib (dose escalation): RP2D; 3+3 design

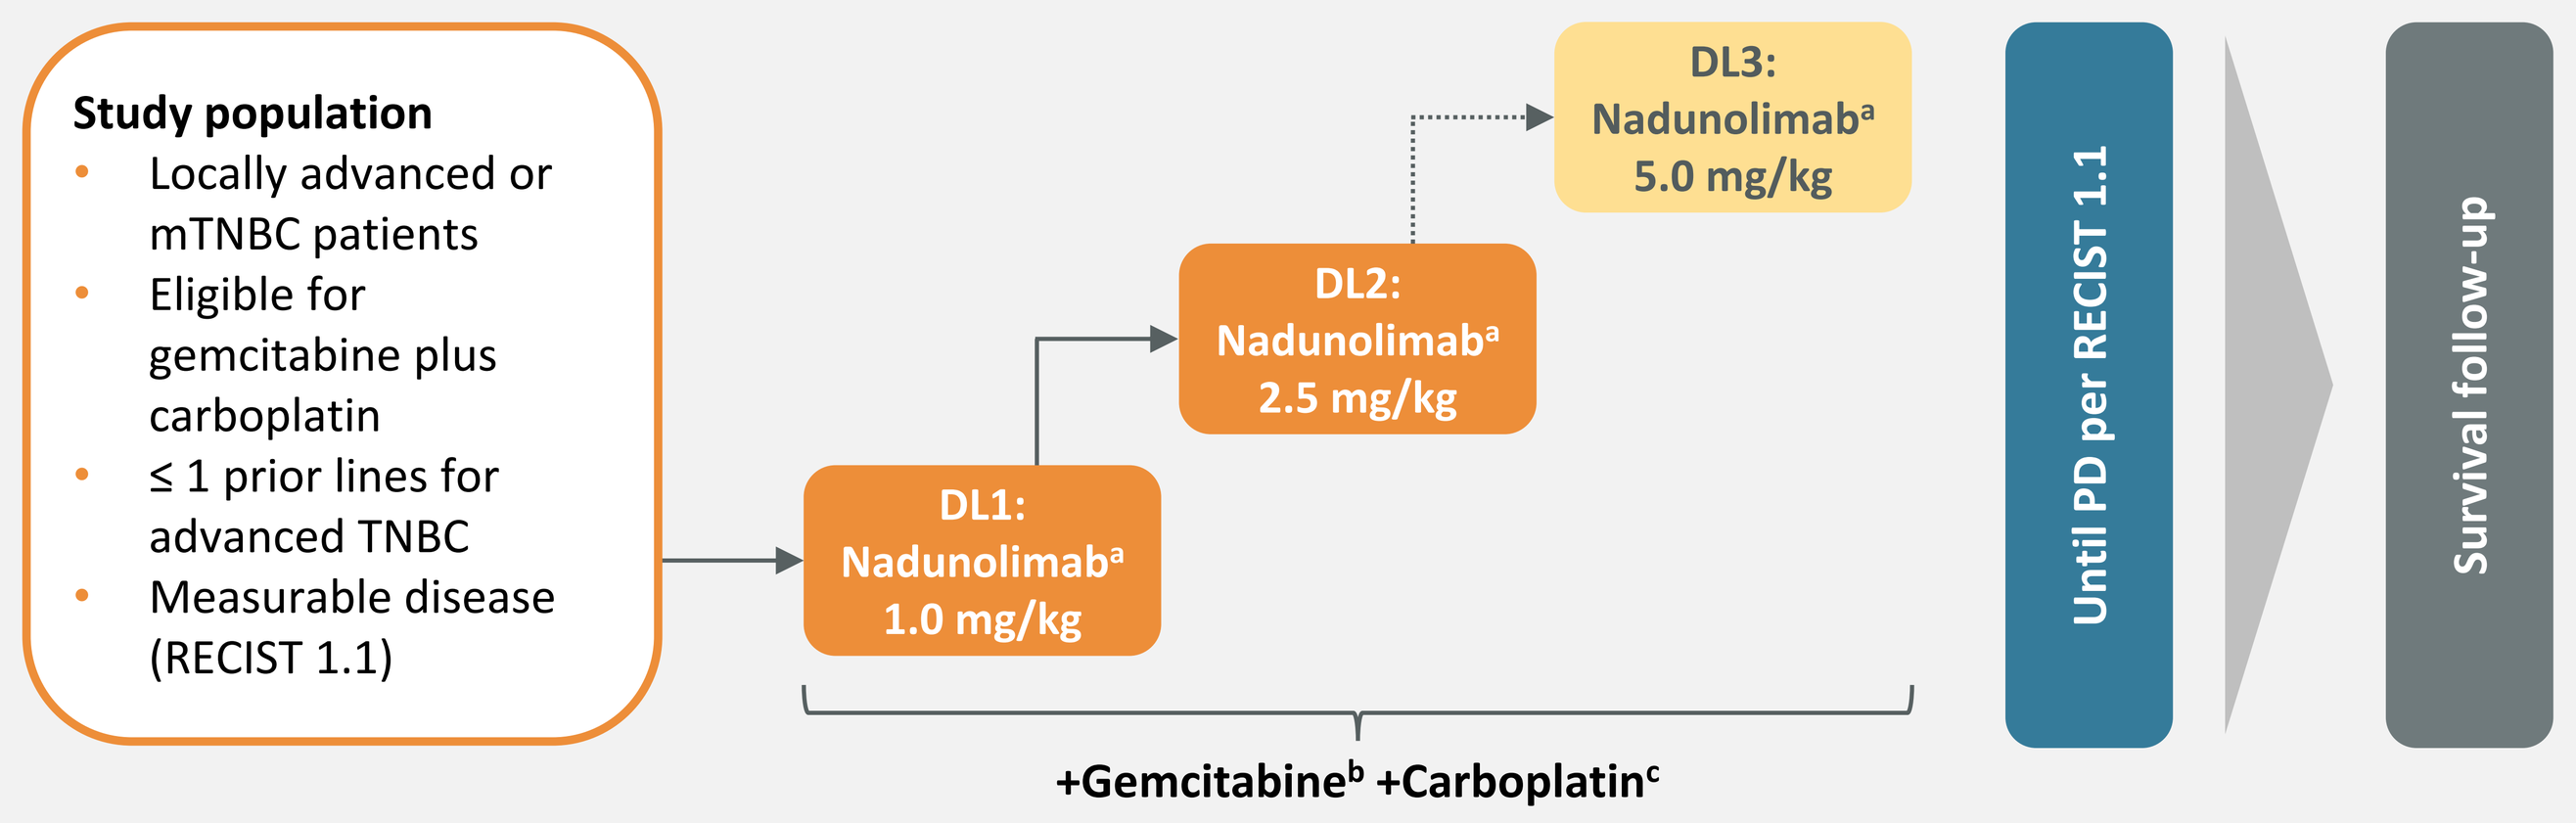
Study population
Locally advanced or mTNBC patients
Eligible for gemcitabine plus carboplatin
≤ 1 prior lines for advanced TNBC
Measurable disease (RECIST 1.1)
DL1: Nadunolimab[a] 1.0 mg/kg
DL2: Nadunolimab[a] 2.5 mg/kg
DL3: Nadunolimab[a] 5.0 mg/kg
+Gemcitabine[b] +Carboplatin[c]
Until PD per RECIST 1.1
Survival follow-up

Primary endpoints
Incidence rate of DLT within the first cycle of NadGC

Secondary endpoints
Efficacy: CBR, DCR, DoR, PFS (RECIST); OS
Safety: Adverse events (NCI-CTCAE v. 5.0)

Exploratory endpoints
Effects on biomarkers in serum and tumor biopsies

[a]Nadunolimab dose escalation (DL1: 1.0 mg/kg; DL2: 2.5 mg/kg; DL3: 5.0 mg/kg) i.v. on days 1 and 8/15, in cycles of 3 weeks
[b]Gemcitabine 1000 mg/m^2 i.v. on days 1 and 8/15, in cycles of 3 weeks
[c]Carboplatin Area Under the Curve (AUC) 2 mg/ml/min i.v. on days 1 and 8/15, in cycles of 3 weeks

- A total of 15 patients were included; three dosed at DL1 and 12 at DL2. Patients were allowed to continue on nadunolimab monotherapy at the investigator's discretion.
- To mitigate Grade 3/4 neutropenia and reduce febrile neutropenia, all patients received prophylactic G-CSF, compulsory in Cycle 1 and left to the investigator's discretion from Cycle 2 onwards.

Results

Patient characteristics

Table 1: *Baseline characteristics*

	All patients (n=15)
Age (years); median (range)	50 (32-69)
Body mass index; median (range)	25 (17-32)
Menopausal status; n (%)	
Postmenopausal	10 (67)
Pre-/per-menopausal	5 (33)
ECOG performance status; n (%)	
0	12 (80)
1	3 (20)
Disease-free interval (years); median (range); n=14	1.9 (0.2-13.5)
Triple-negative; n (%)	15 (100)
Ki67 (%); median (range)	68 (10-90)
BRCA; n (%)	
Positive	0
Negative	6 (40)
Not available	9 (60)
Visceral lesions; n (%)	14 (93)
Metastatic locations; n (%)	
≤ 3	10 (67)
> 3	5 (33)
Prior lines of therapy for metastatic disease; n (%)	
None	5 (33)
1	10 (67)
Prior ICI in first line therapy for metastatic disease; n (%)	5 (33)
Prior platinum agent in (neo)adjuvant treatment; n (%)	4 (27)

- Most frequent metastatic lesions were lymph node (67%), lung (60%), liver (47%) and bone (27%).

Safety

Table 2: *Grade 3 or higher treatment-emergent adverse events (TEAEs); n (%)*

	All patients (n=15)	
	Grade ≥3	All grades
Neutropenia	8 (53)	10 (67)
Thrombocytopenia	4 (27)	6 (40)
Anaemia	3 (20)	6 (40)
Febrile neutropenia	2 (13)	2 (13)
COVID-19	1 (7)	1 (7)
Device-related infection	1 (7)	1 (7)
Diarrhoea	1 (7)	4 (27)
Hypocalcaemia	1 (7)	1 (7)
Hypomagnesaemia	1 (7)	2 (13)
Procedural pneumothorax	1 (7)	1 (7)

- Dose escalation was not continued beyond DL2 based on observed safety and results from other trials of nadunolimab with chemotherapy. MTD was not formally achieved; 2.5 mg/kg was the maximal administered dose of nadunolimab.
- TEAEs of Grade ≥3 were reported in 12 (80%) patients, leading to treatment discontinuation in one (7%) patient. Five (33%) patients had serious adverse events (SAEs): febrile neutropenia* (13%), hypocalcaemia* (7%), hypomagnesaemia* (7%), procedural pneumothorax (7%), device-related infection (7%) and COVID-19 (7%). SAEs marked with * were considered related.
- One infusion-related reaction (IRR) of grade 2 was reported at DL2.
- Two DLTs were reported at DL2: Grade 3 neutropenia causing a delay of >7 days in Cycle 2 and Grade 3 febrile neutropenia.
- Mean (range) number of treatment cycles with nadunolimab: 8 (2-17), gemcitabine: 7 (2-14), carboplatin: 7 (2-14). Ten (67%) patients received ≥5 treatment cycles. Fourteen (93%) patients had any dose modifications.
- Ten (67%) patients, three in DL1 and seven in DL2, received subsequent therapy.

Efficacy

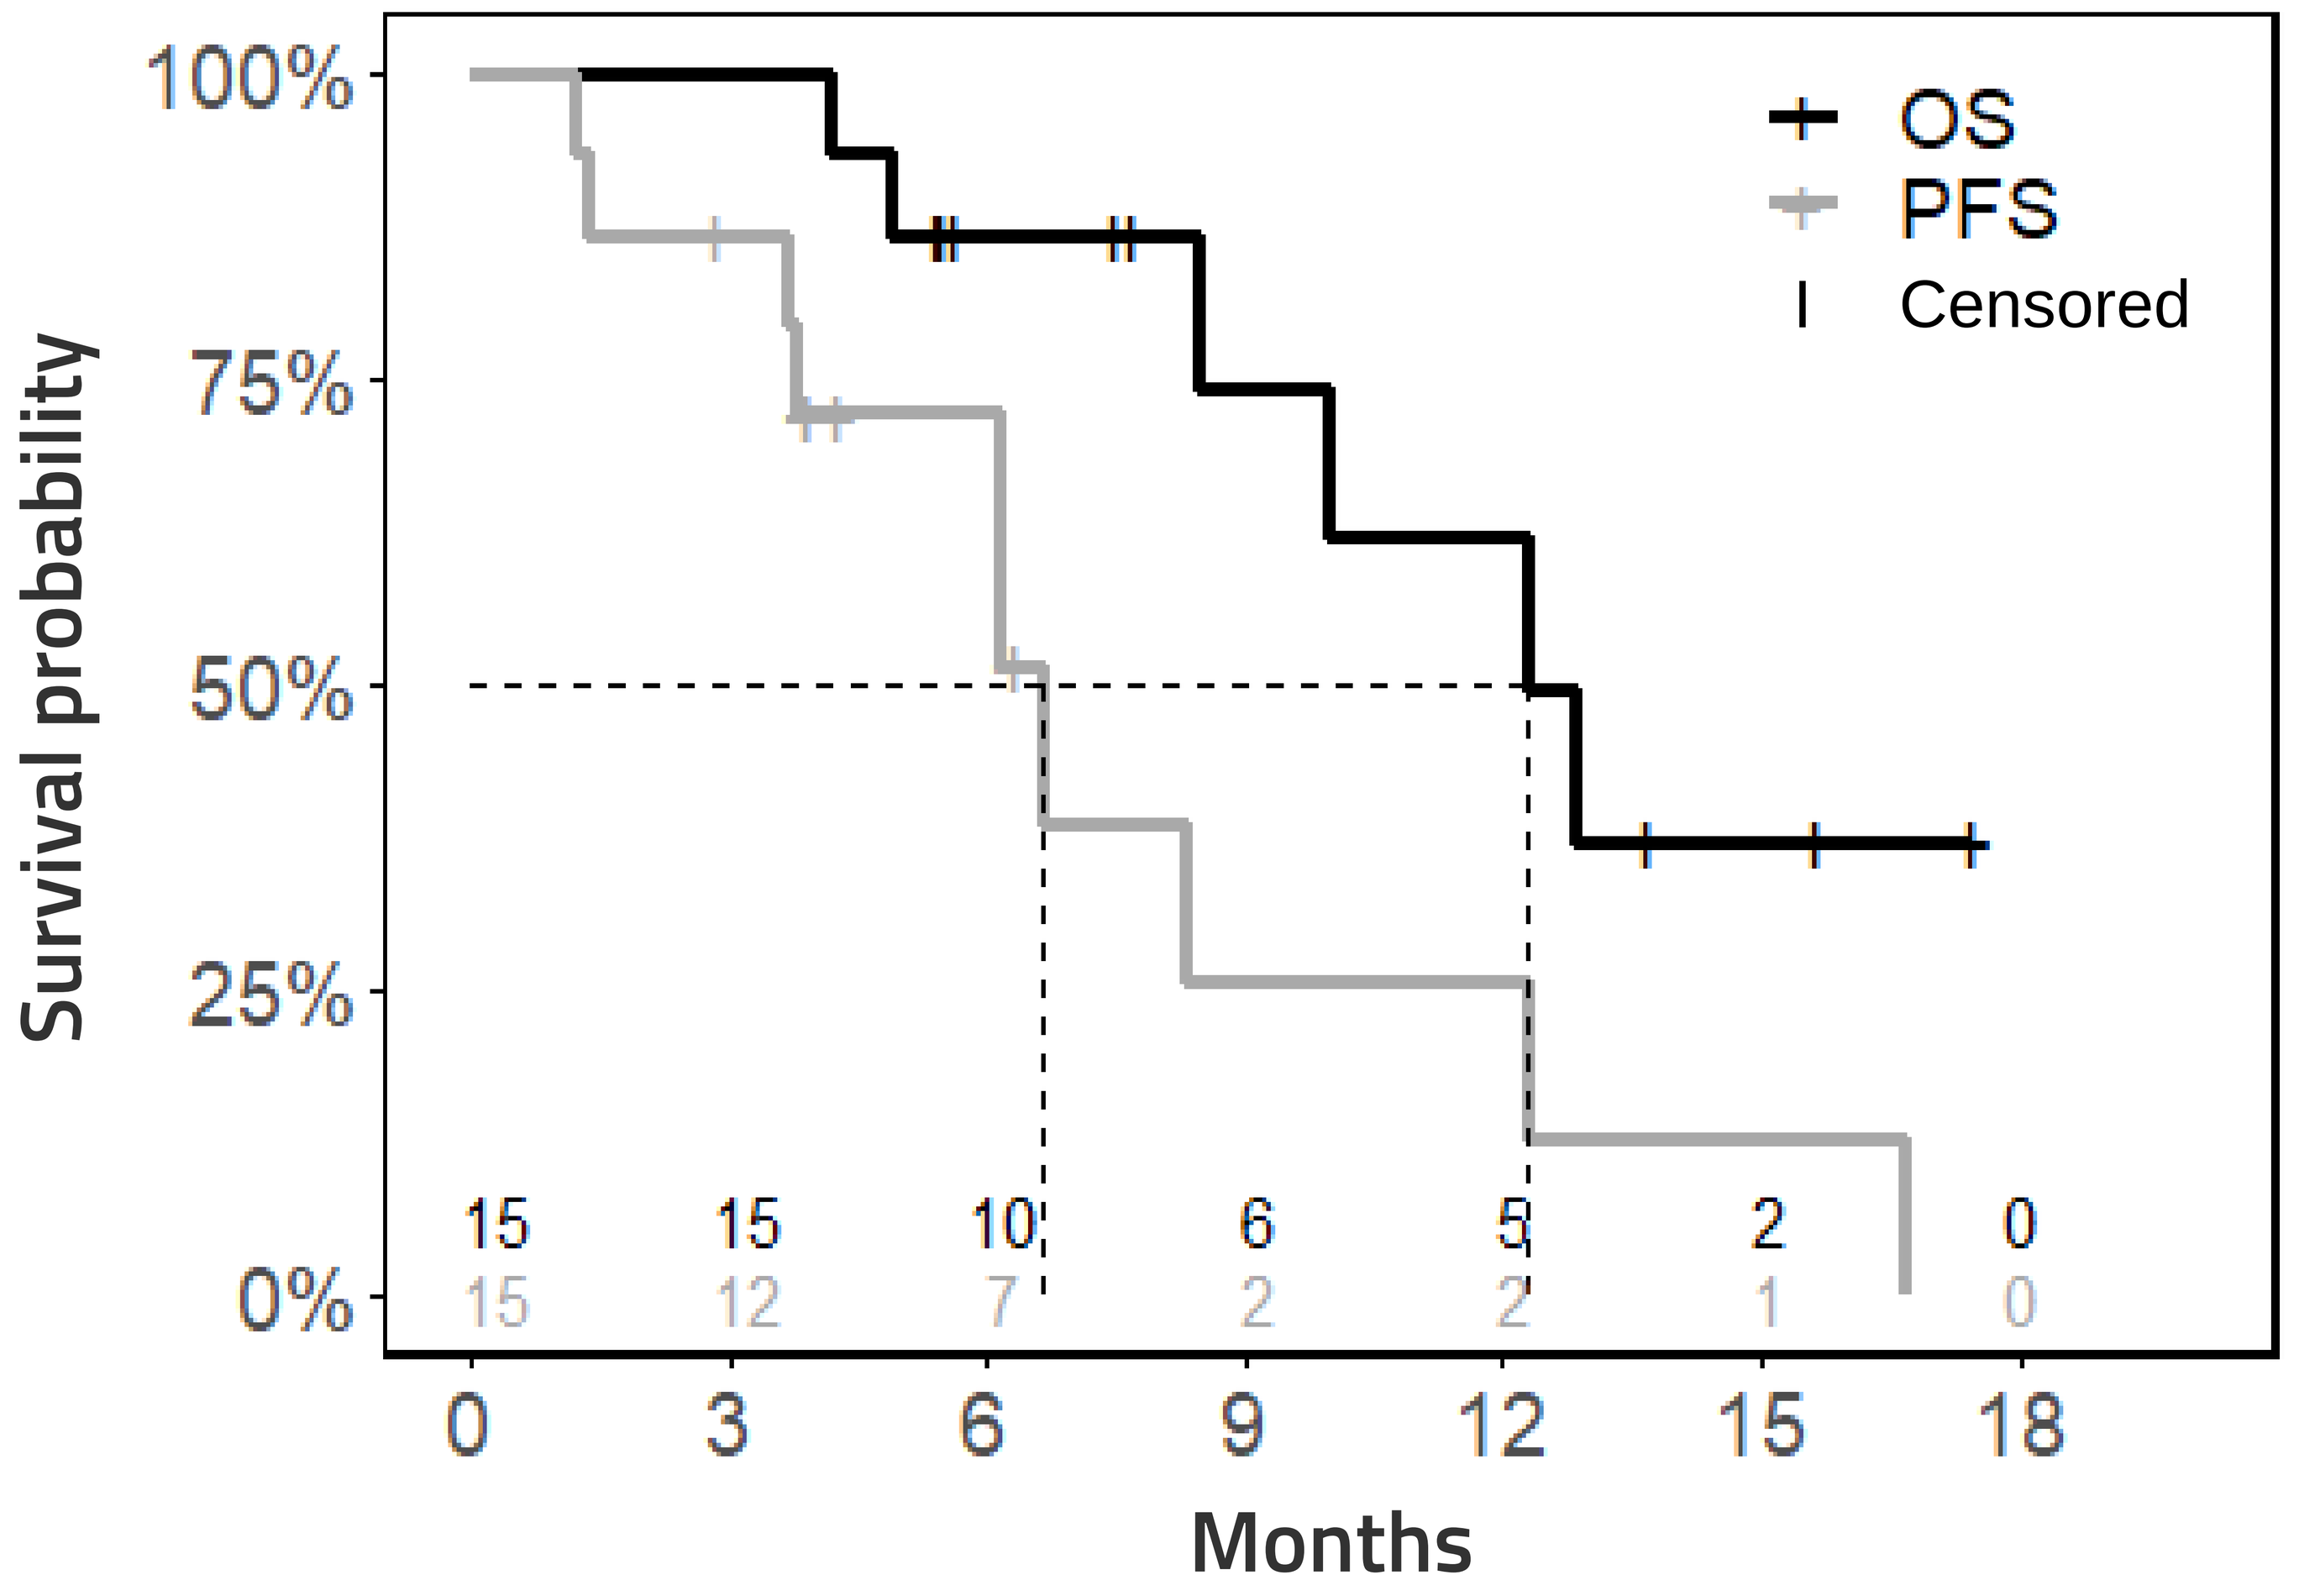
OS
PFS
Censored
100%
75%
50%
25%
0%
Survival probability
15 15 10 6 5 2 0
15 12 7 2 2 1 0
0 3 6 9 12 15 18
Months

Figure 3: *Kaplan-Meier curves for survival analysis (OS and PFS; n=15)*

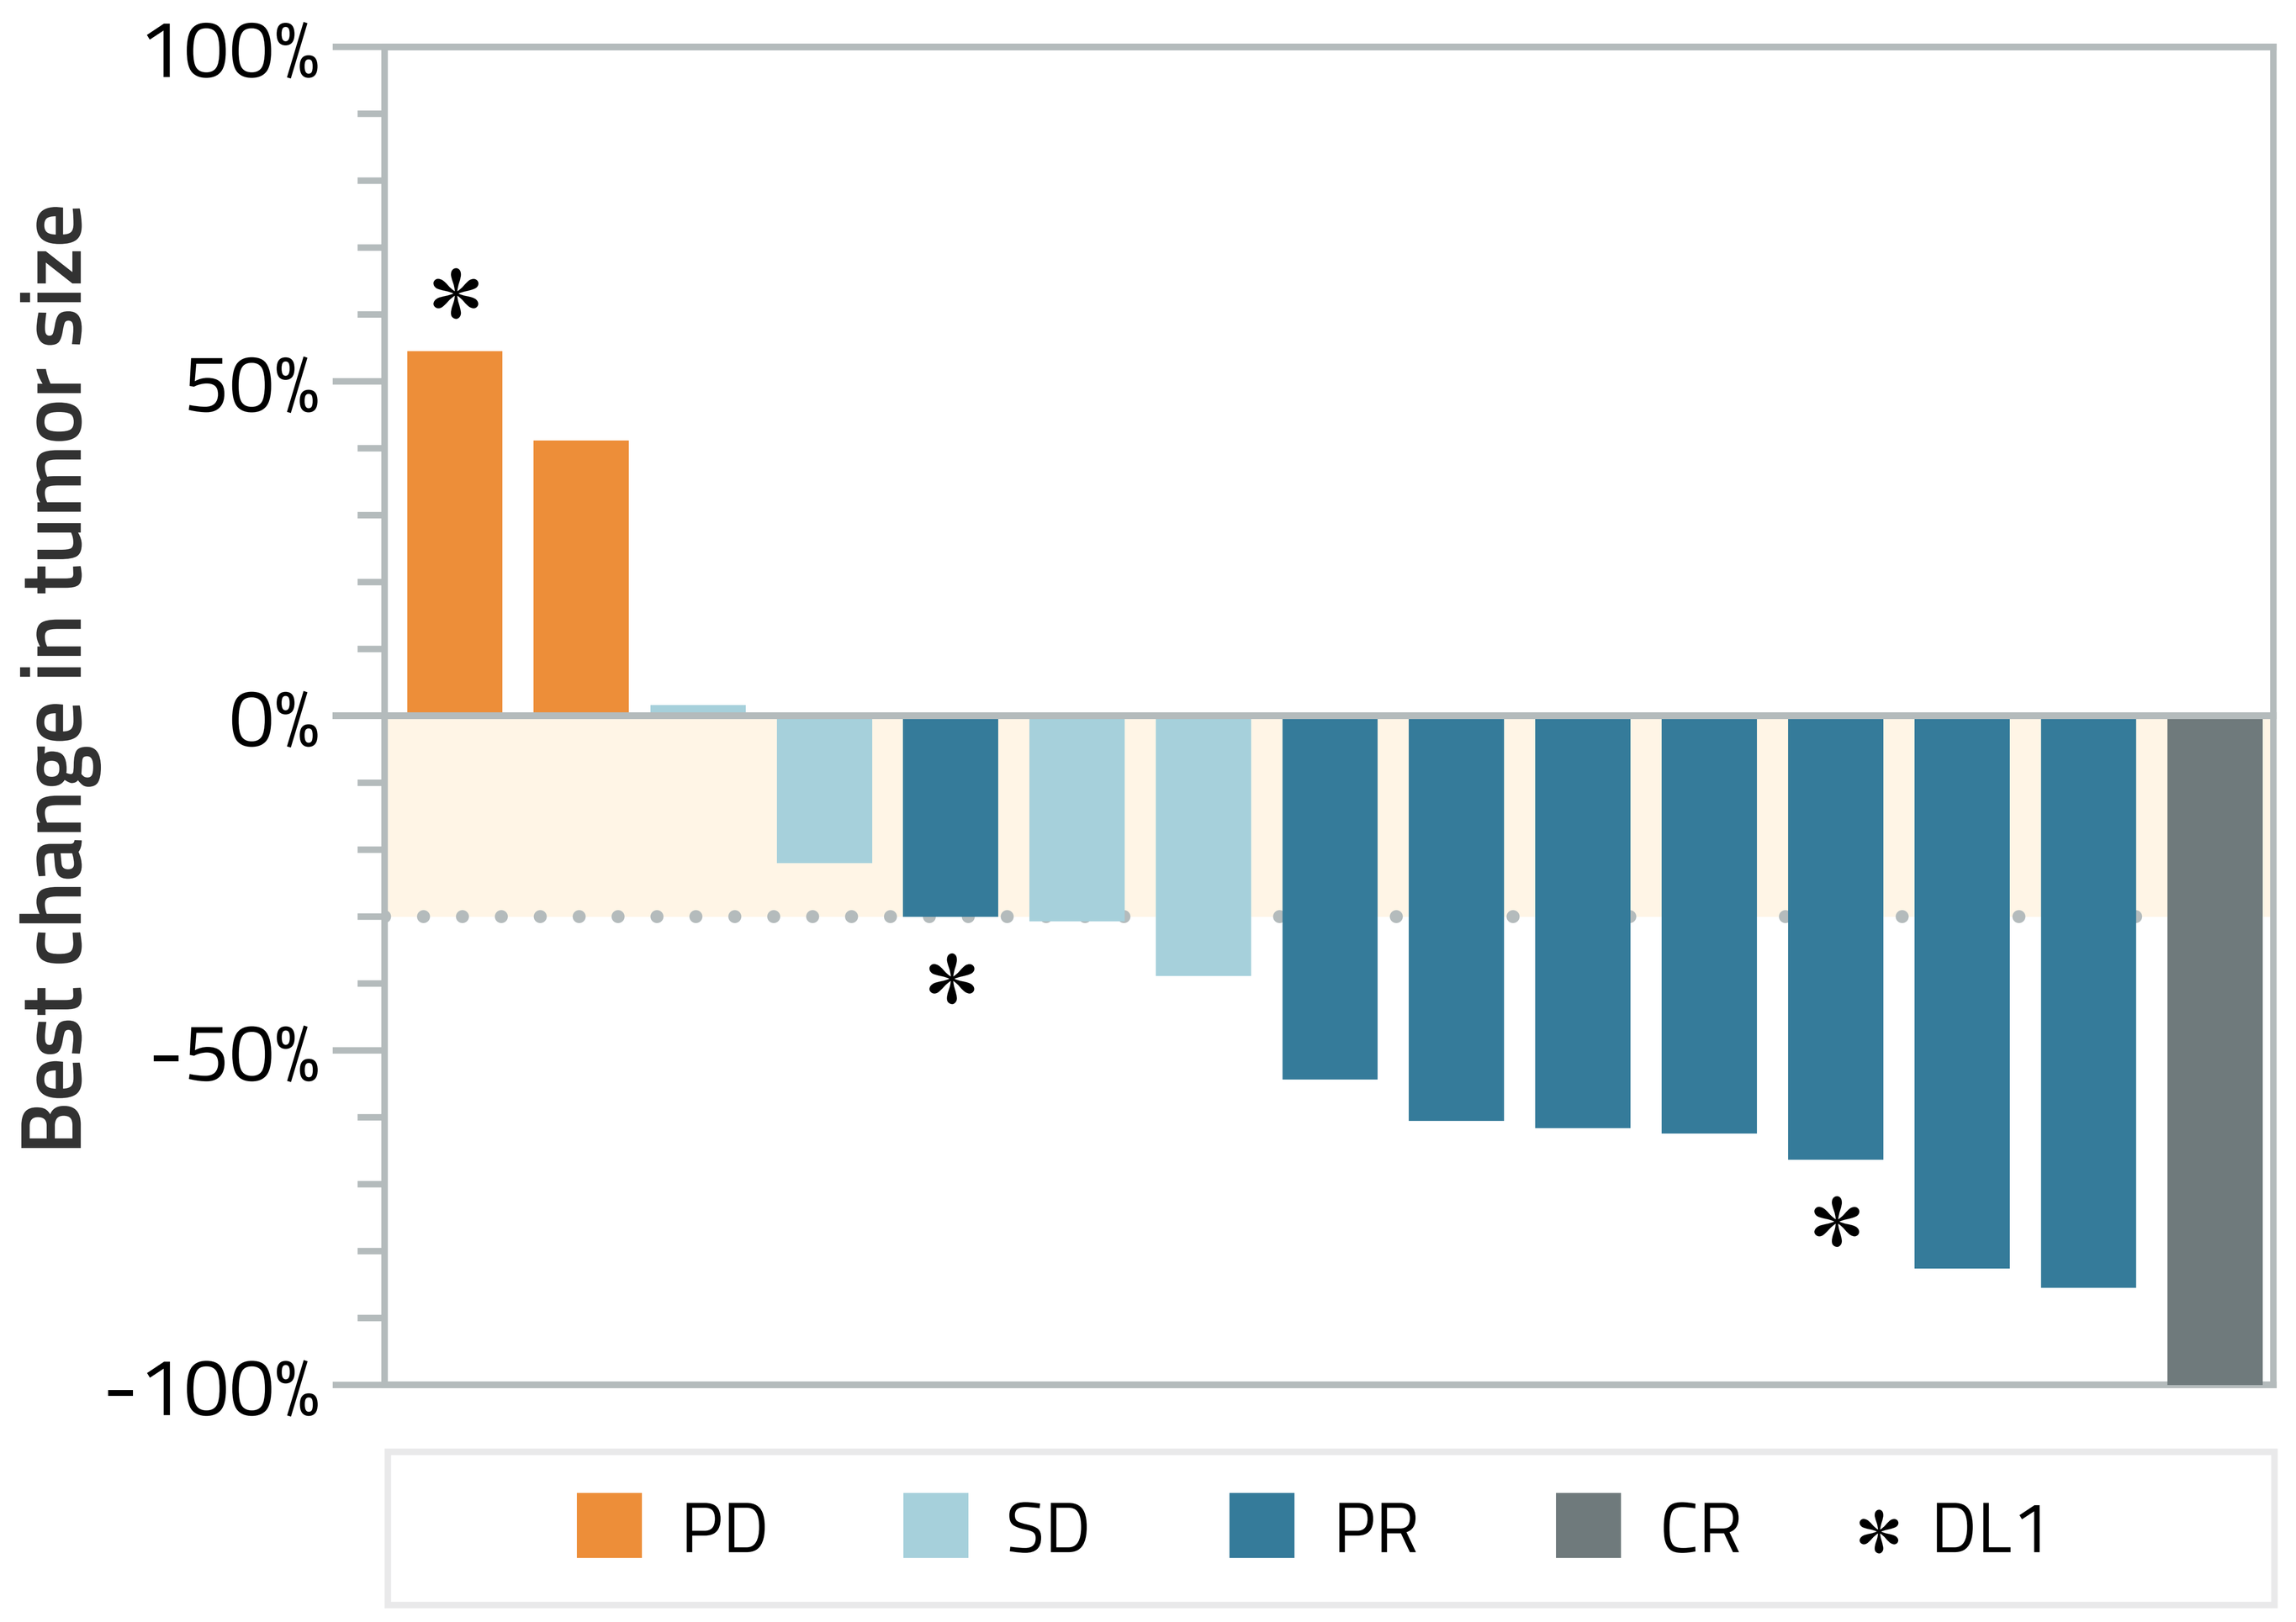
100%
50%
0%
-50%
-100%
Best change in tumor size
PD
SD
PR
CR
DL1

Figure 4: *Waterfall plot of the best percent change in target lesion size (n=15)*

- At data cut-off (17 July 2023), two (13%) patients were on treatment and six (40%) had died. The cause of death was breast cancer.
- Preliminary ORR was 60%, including one confirmed complete response (CR) and eight confirmed partial responses (PR).
- Preliminary median OS was 12.3 months (95% CI: 8.5-NE) and preliminary median PFS was 6.6 months (95% CI: 3.7-12.3).

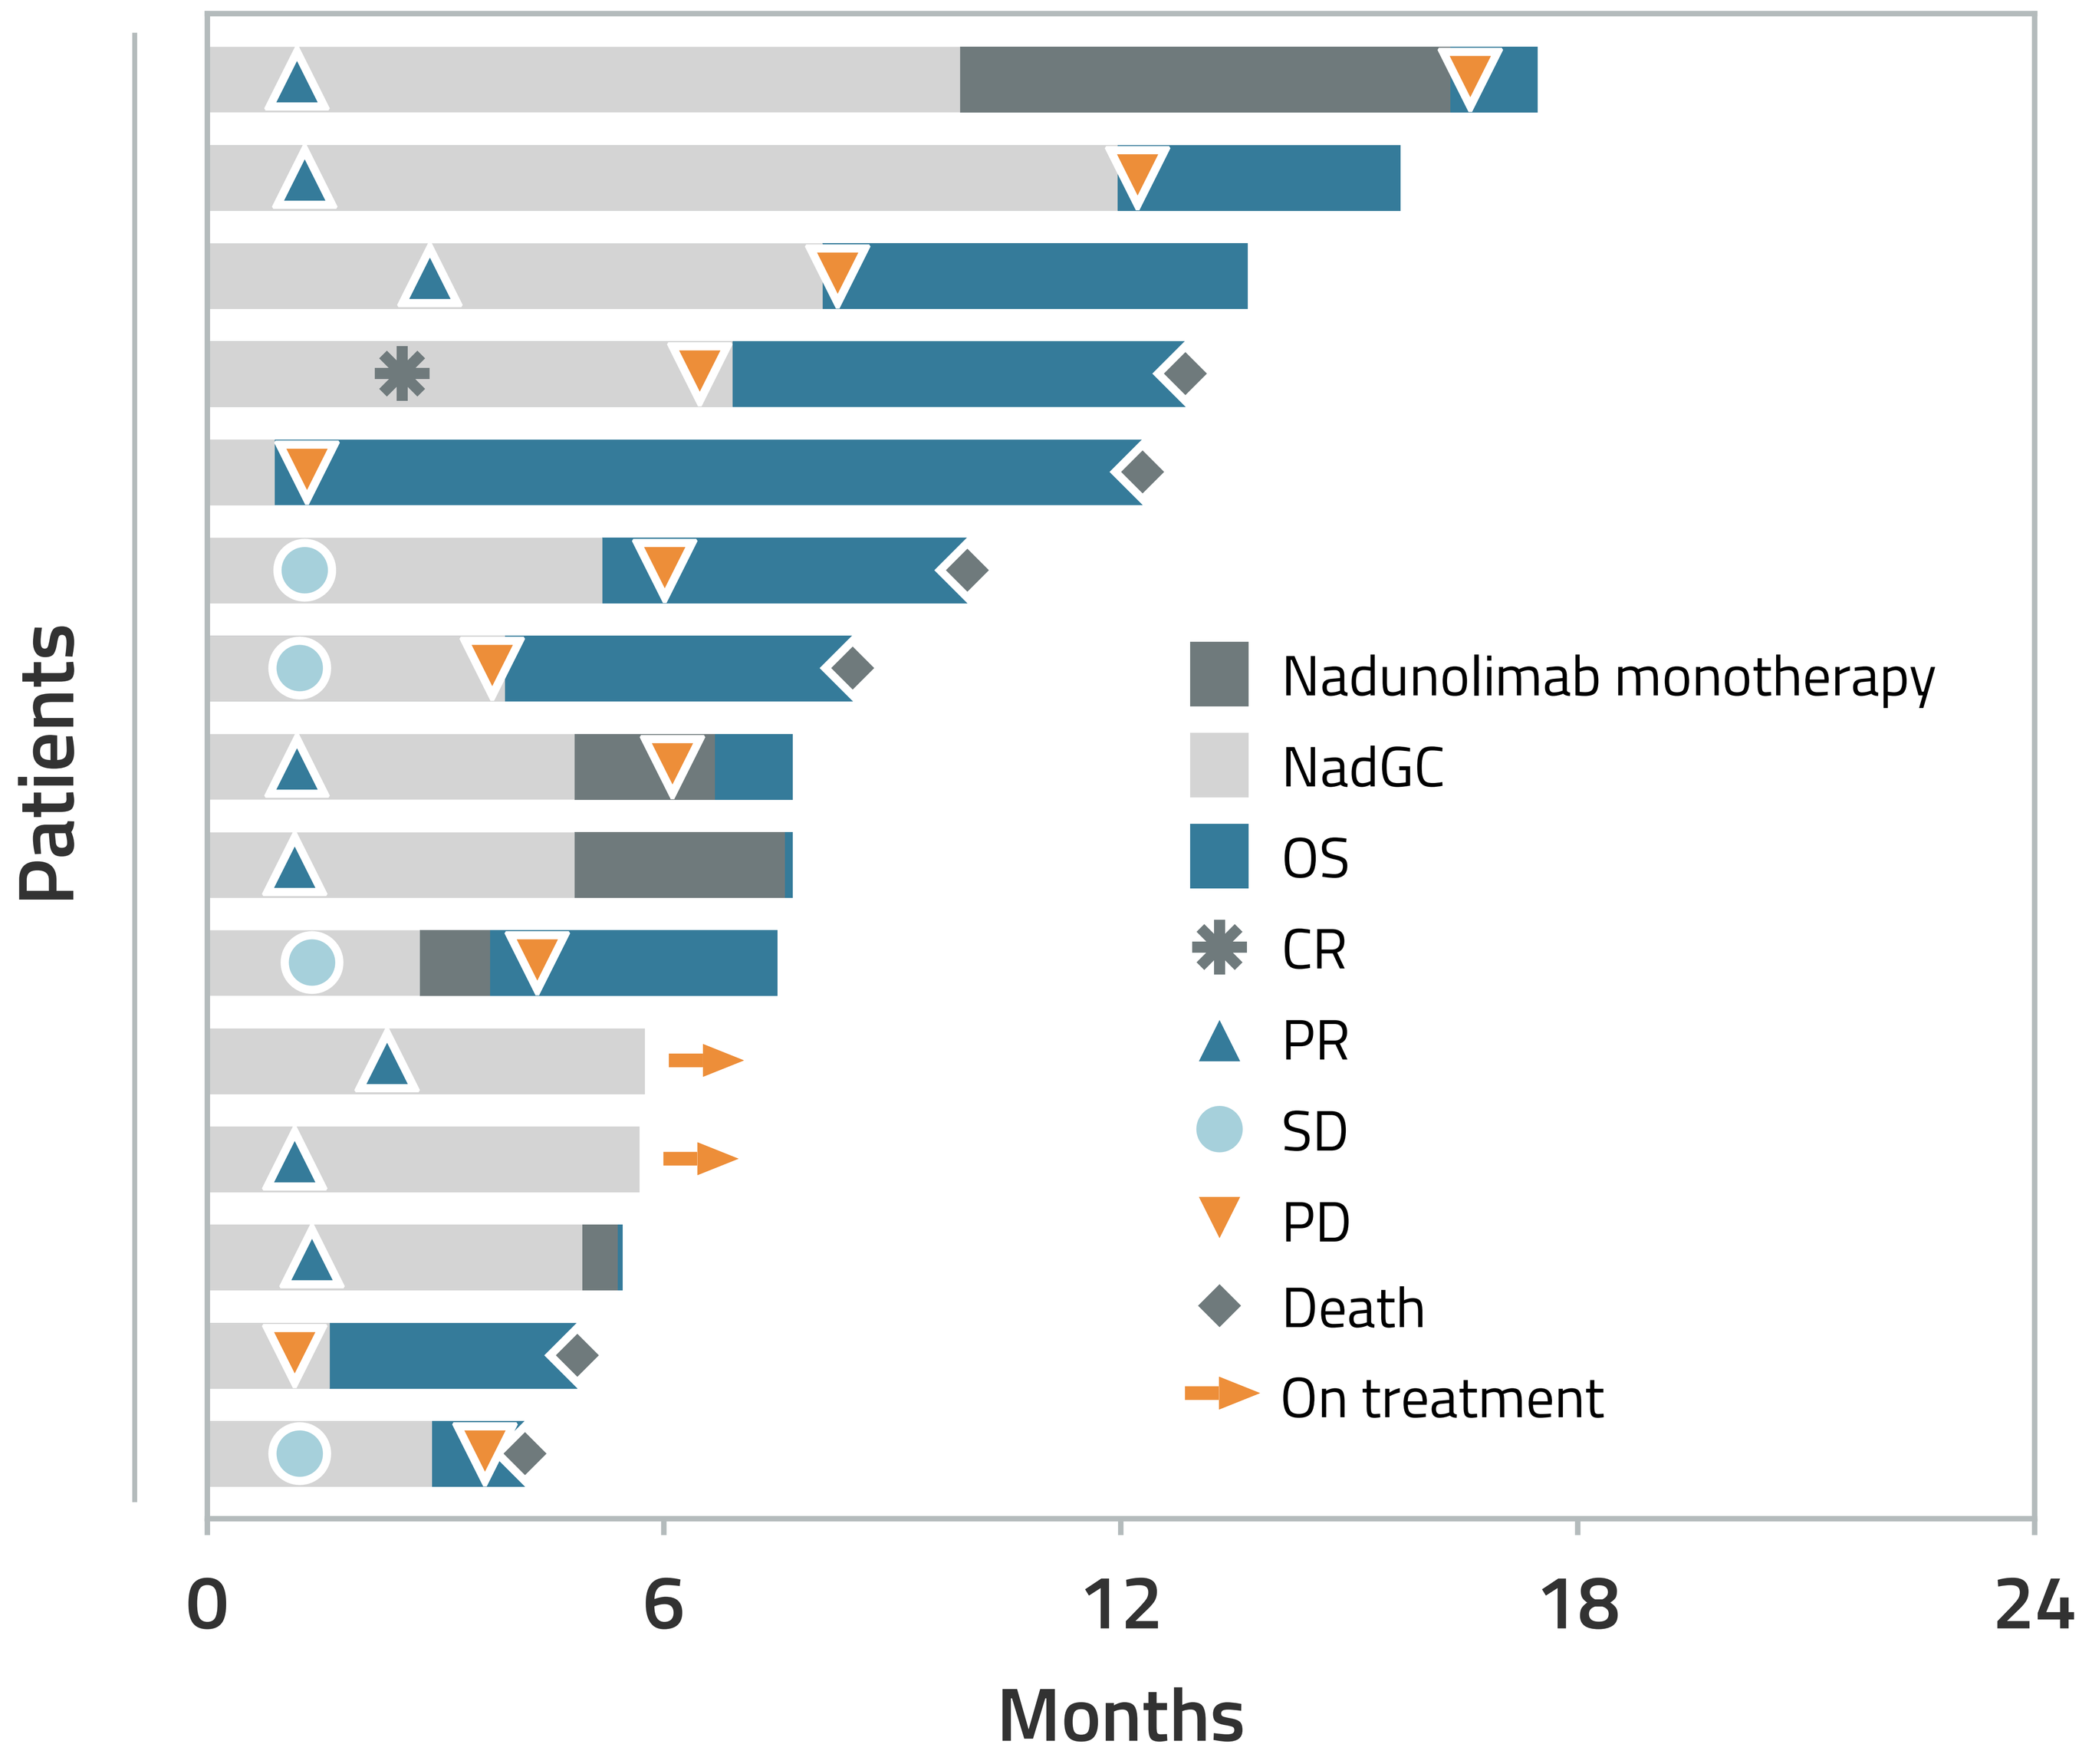
Patients
Nadunolimab monotherapy
NadGC
OS
CR
PR
SD
PD
Death
On treatment
0 6 12 18 24
Months

Figure 5: *Treatment course for each individual patient (n=15)*

Conclusions

- **Nadunolimab plus standard gemcitabine and carboplatin (NadGC) is well tolerated, and preliminary data shows promising antitumor activity in metastatic TNBC compared to historical data[7]:**
 - **ORR: 60%**
 - **Median OS: 12.3 months**
 - **Median PFS: 6.6 months**
- **The safety profile of NadGC was similar to previous reports for gemcitabine and carboplatin only**
- **The randomized phase II part of TRIFOUR is currently enrolling patients at the 2.5 mg/kg nadunolimab dose**

References

[1] Bruchard et al; Nat Med (2013)
[2] Chung et al; NPJ Breast Cancer (2022)
[3] Tjomsland et al; Neoplasia (2011)
[4] Liu et al; Cancer Res (2018)
[5] Zhang et al; Cancer Res (2018)
[6] Van Cutsem et al; Cancer Res (2023)
[7] O'Shaughnessy et al; J Clin Oncol (2014)

Acknowledgements

We would like to thank the patients and their families for participating in the study, and all study staff at the clinical sites.