4148

Impact on chemotherapy induced peripheral neuropathy of nadunolimab, a first in class monoclonal antibody against IL1RAP, in patients with pancreatic cancer, with supportive mouse model data

Eric Van Cutsem[1]; Sebastian Ochsenreither[2,3,4], Joelle Collignon[5], Rikke Eefsen[6], Audrius Ivanauskas[7], Svetlana Shatunova[8], Irina Vetter[8], Elin Jaensson Gyllenbäck[9], Petter Skoog[9], Nedjad Losic[9], Dominique Tersago[9], Hana Starobova[8]

[1]UZ Leuven Gasthuisberg, Leuven, Belgium; [2]Charité Comprehensive Cancer Center, Berlin, Germany; [3]Charité, Department of Hematology, Oncology and Tumor Immunology, Berlin, Germany; [4]German Cancer Consortium (DKTK), Berlin, Germany; [5]CHU de Liège, Liège, Belgium; [6]Herlev og Gentofte Hospital, Herlev, Denmark, [7]The Hospital of Lithuanian University of Health Sciences, Kaunas, Lithuania; [8]University of Queensland, Institute for Molecular Bioscience; [9]Cantargia AB, Lund, Sweden

Introduction

Chemotherapy induced peripheral neuropathy (CIPN) is one of the major nonhematological adverse events arising from chemotherapies. CIPN is a neuroinflammatory condition, with nerve cell damage, immune cell activation and interleukin-1 (IL-1) release being significant drivers of CIPN following tubulin targeting drugs such as paclitaxel and vincristine (References 1-4).

Nadunolimab is a fully humanized monoclonal IgG1 antibody targeting IL1 receptor accessory protein (IL1RAP). Through binding IL1RAP, nadunolimab can induce antibody-dependent cellular cytotoxicity (ADCC) of IL1RAP-expressing tumor and immune cells and inhibit tumor-promoting and inflammatory signals mediated by both IL-1α and IL-1β.

CANFOUR (NCT03267316) was a phase I/IIa study evaluating nadunolimab in combination with nab-paclitaxel and gemcitabine (GN) in previously untreated, unresectable, locally advanced or metastatic PDAC. Preliminary results from the trial demonstrate promising efficacy metrics, including median overall survival (OS) of 13.2 months and median progression-free survival (iPFS) of 7.2 months, with a favorable safety profile. Notably, a low incidence of grade ≥ 3 peripheral neuropathy was reported, with only 1 event in 73 patients,

Study Design

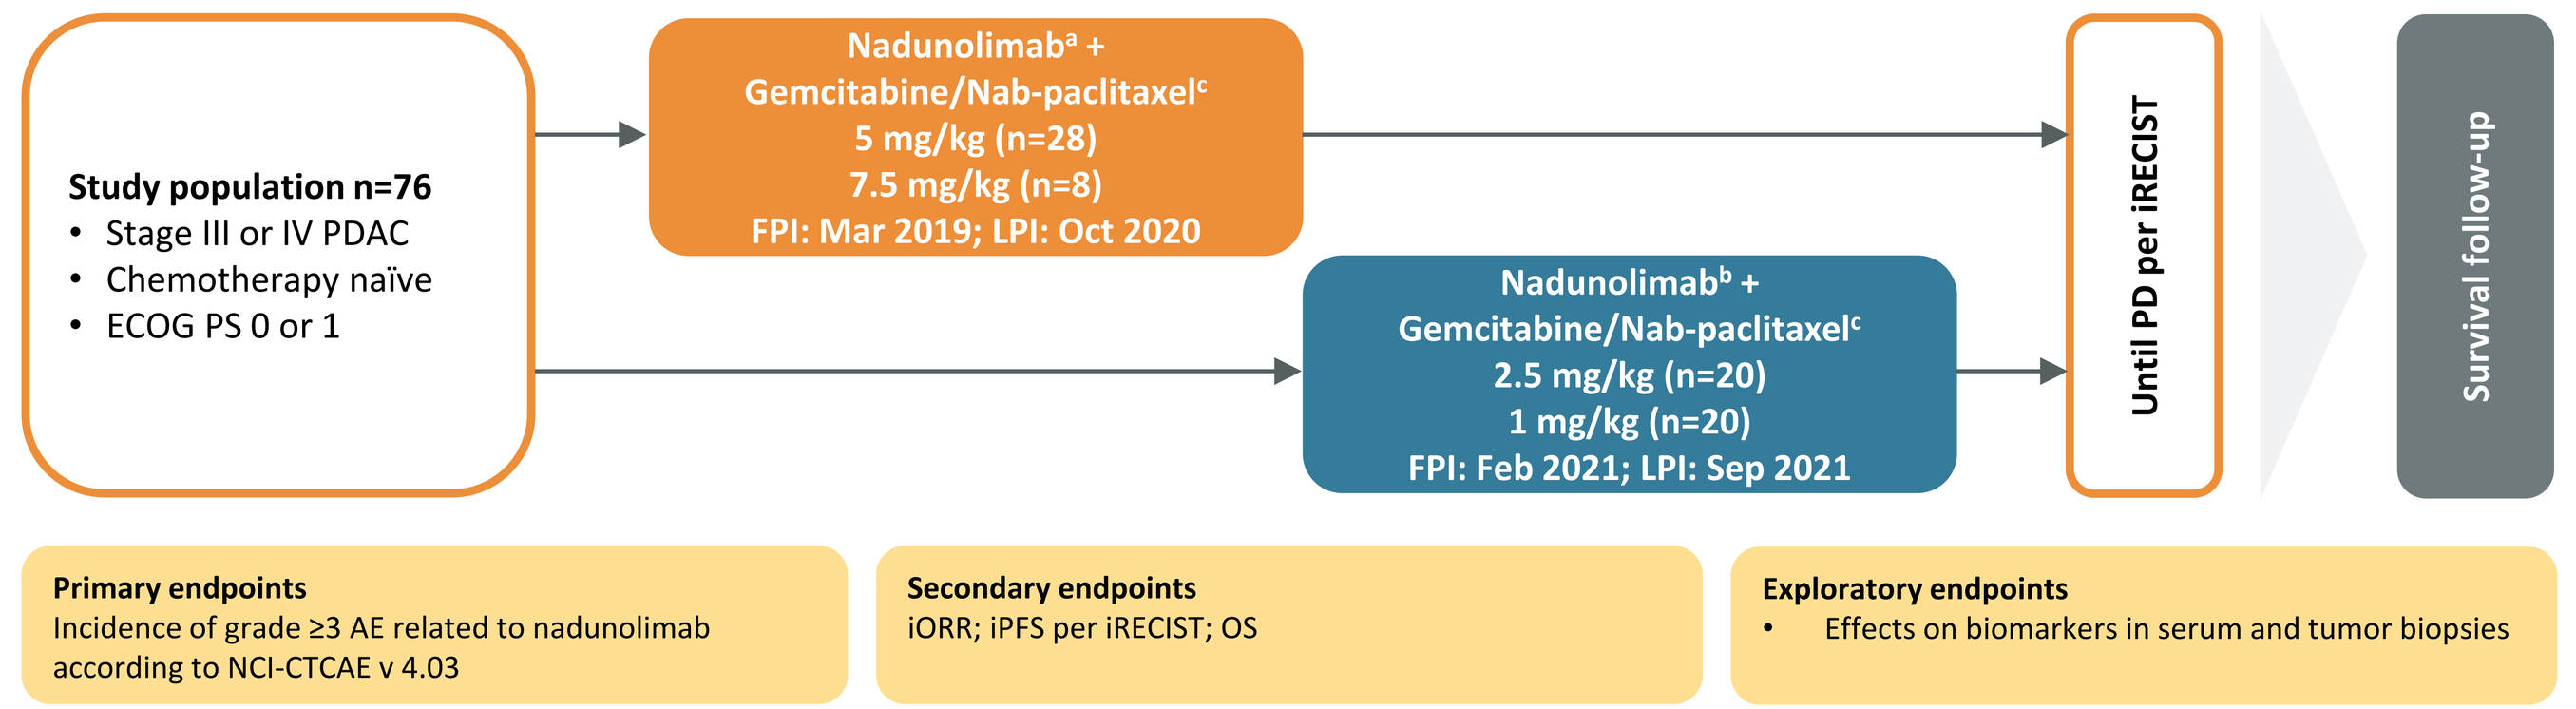
Study population n=76
Stage III or IV PDAC
Chemotherapy naïve
ECOG PS 0 or 1
Nadunolimab[a] + Gemcitabine/Nab-paclitaxel[c]
5 mg/kg (n=28)
7.5 mg/kg (n=8)
FPI: Mar 2019; LPI: Oct 2020
Nadunolimab[b] + Gemcitabine/Nab-paclitaxel[c]
2.5 mg/kg (n=20)
1 mg/kg (n=20)
FPI: Feb 2021; LPI: Sep 2021
Until PD per iRECIST
Survival follow-up
Primary endpoints
Incidence of grade ≥3 AE related to nadunolimab according to NCI-CTCAE v 4.03
Secondary endpoints
iORR; iPFS per iRECIST; OS
Exploratory endpoints
Effects on biomarkers in serum and tumor biopsies

[a]Nadunolimab given Q1W for first 6 wks followed by Q2W; priming dose (0.5 mg/kg) given on Day -7 to mitigate infusion-related reactions.
[b]Nadunolimab given on Day 1 and 15 in cycles of 28 days and on Day 8 in Cycle 1 only; no priming dose.
[c]Gemcitabine (1000 mg/m^2)/Nab-paclitaxel (125 mg/m^2) given in cycles of 28 days on Day 1, 8 and 15 of each cycle.

Figure 1: Summary of the study design for the PDAC cohorts in part IIa of the CANFOUR study.

Efficacy

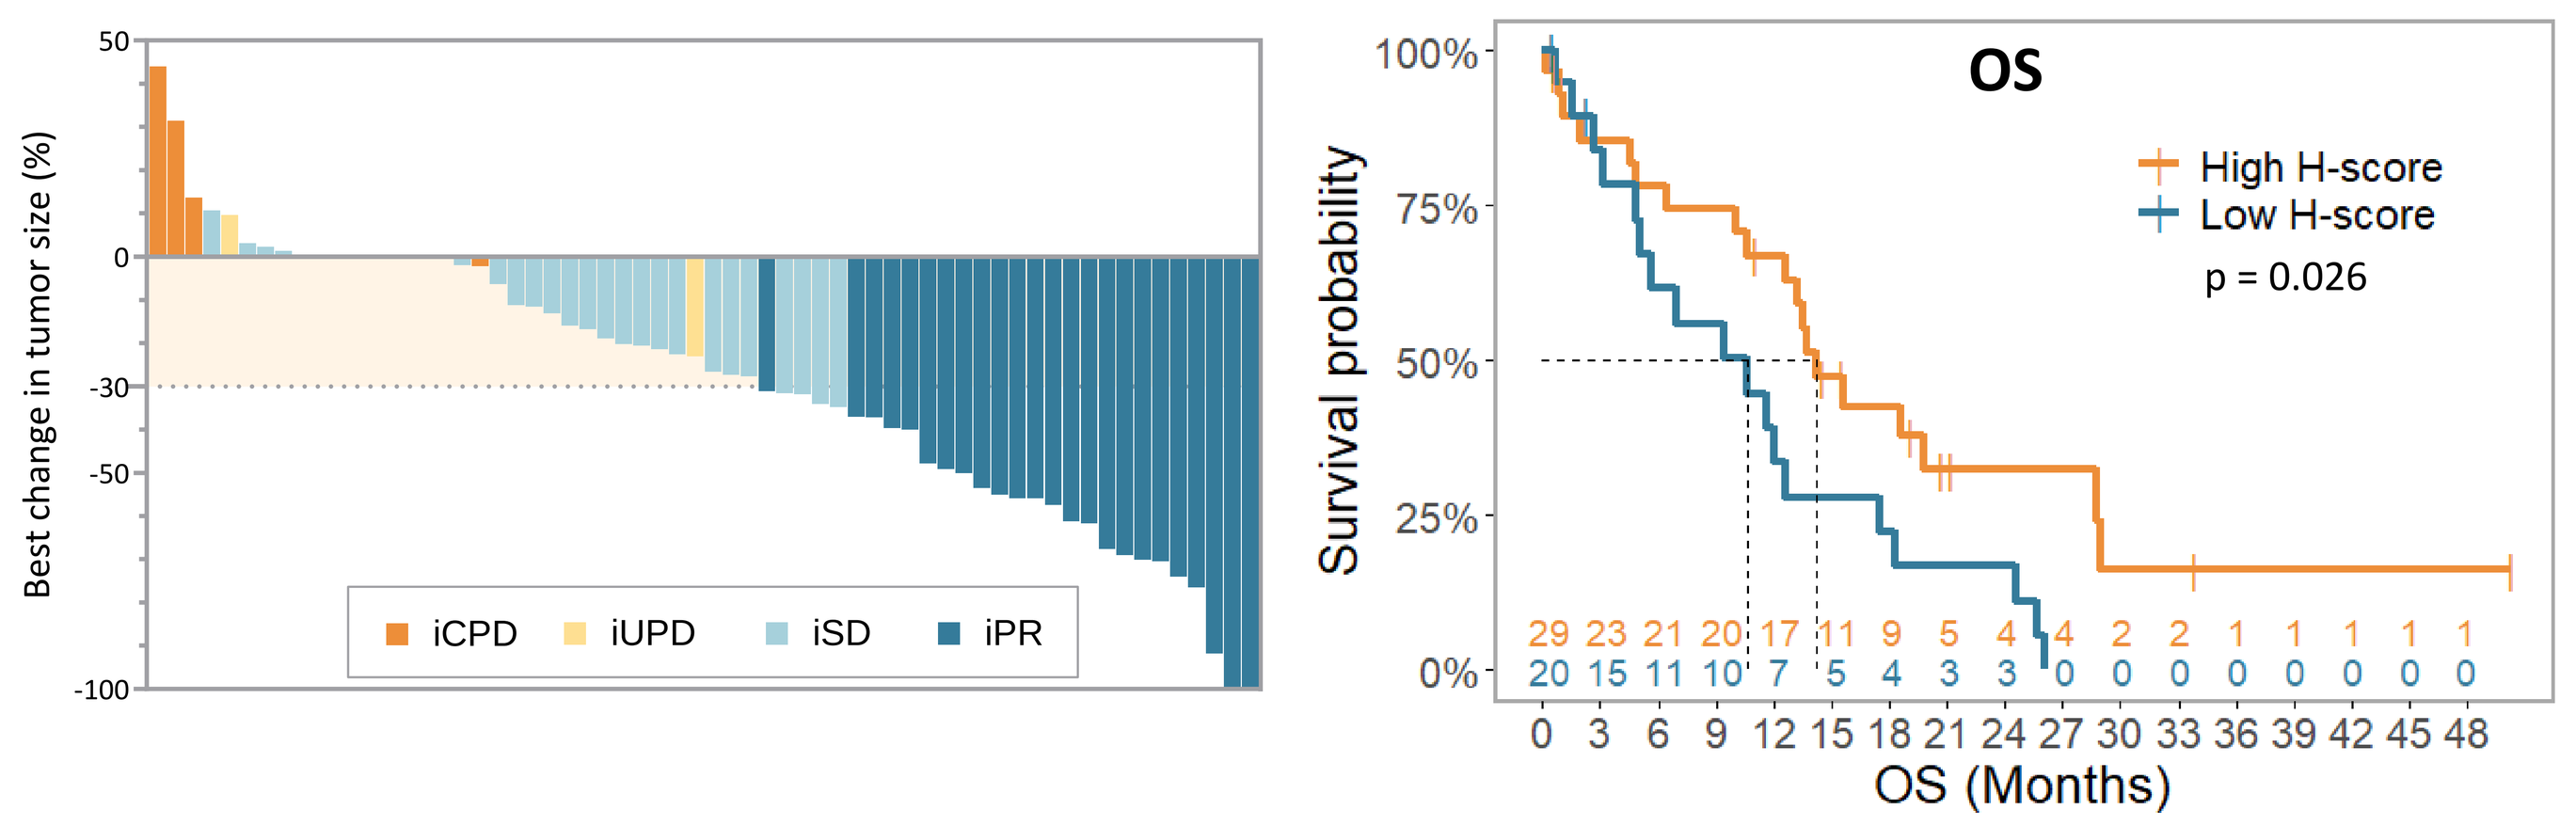
Best change in tumor size (%)
iCPD
iUPD
iSD
iPR
OS
High H-score
Low H-score
p = 0.026
Survival probability
OS (Months)

Figure 2: Tumor response evaluated according to iRECIST criteria.
Waterfall of all evaluable patients (left): Twenty-four (33%) patients had iPR as best overall response, 28 (38%) patients had iSD, 5 (7%) patients had iUPD and 5 (7%) patients had iCPD. Right: 49 patients with available tumor biopsies at screening were grouped based on IL1RAP expression on tumor cells. Patients with high target expression of IL1RAP showed significantly longer OS.

Table 1: Efficacy parameters in patients treated with nadunolimab/GN in the CANFOUR study.

Efficacy parameter (95% CI)	mITT (n=73)	Biopsy subgroup (n=49)	IL1RAP High (n=29)	IL1RAP Low (n=20)
OS; median, months	13.2 (10.6-15.5)	12.6 (9.5-17.5)	14.2 (10.0-28.7)	10.6 (4.8-12.6)
iPFS; median, months	7.2 (5.2-8.5)	7.2 (3.9-8.9)	7.4 (3.7-11.2)	5.8 (2.7-7.4)
1-year survival	58% (45%-69%)	56% (40%-69%)	67% (46%-81%)	39% (18%-60%)
iORR	33% (22%-45%)	41% (27%-56%)	48% (29%-67%)	30% (12%-54%)
iDoR; median, months	7.3 (5.5-11.0)	6.9 (5.5-10.0)	9.5 (3.7-11.8)	5.6 (3.9-NE*)

*NE; not estimable

Efficacy population: modified intention to treat (mITT): n=73. Three patients only received a priming dose of nadunolimab and discontinued before receiving any chemotherapy

Background and Aim

Treatment-Emergent Adverse Events

Table 2: Treatment-emergent adverse events (TEAEs, worst grade by patient)

	Grade 3-4 (n=76)	All grade (n=76)
Hematological TEAE; n (%)		
Neutropenia	50 (66%)	58 (76%)
Leukopenia/WBC decreased	18 (24%)	22 (29%)
Thrombocytopenia	10 (13%)	30 (40%)
Anemia	10 (13%)	39 (51%)
Febrile neutropenia	10 (13%)	10 (13%)
Non-hematological TEAE; n (%)		
GGT increased	13 (17%)	17 (22%)
Hypertension	7 (9%)	10 (13%)
ALT increased	5 (7%)	15 (20%)
Fatigue	6 (8%)	41 (54%)
AST increased	4 (5%)	13 (17%)
Vomiting	4 (5%)	28 (37%)
Neuropathy*	**1 (1%)**	**31 (42%)**

*Definition of peripheral neuropathy used: all AEs that coded to high level group term (HLGTs) of Peripheral neuropathies plus the preferred term (PT) Paraesthesia (HLGT Neurological disorders NEC) were included

TEAEs of grade 3-4 were reported in 89% of the patients. Neutropenia was the most frequently observed TEAE, and was managed by treatment with G-CSF. 5 mg/kg was considered as Maximum Tolerated Dose.

The observation of only one ≥G3 neuropathy in the CANFOUR study suggest a novel therapeutic approach that not only addresses tumor progression but also offers a potential therapeutic targeting of peripheral neuropathy.

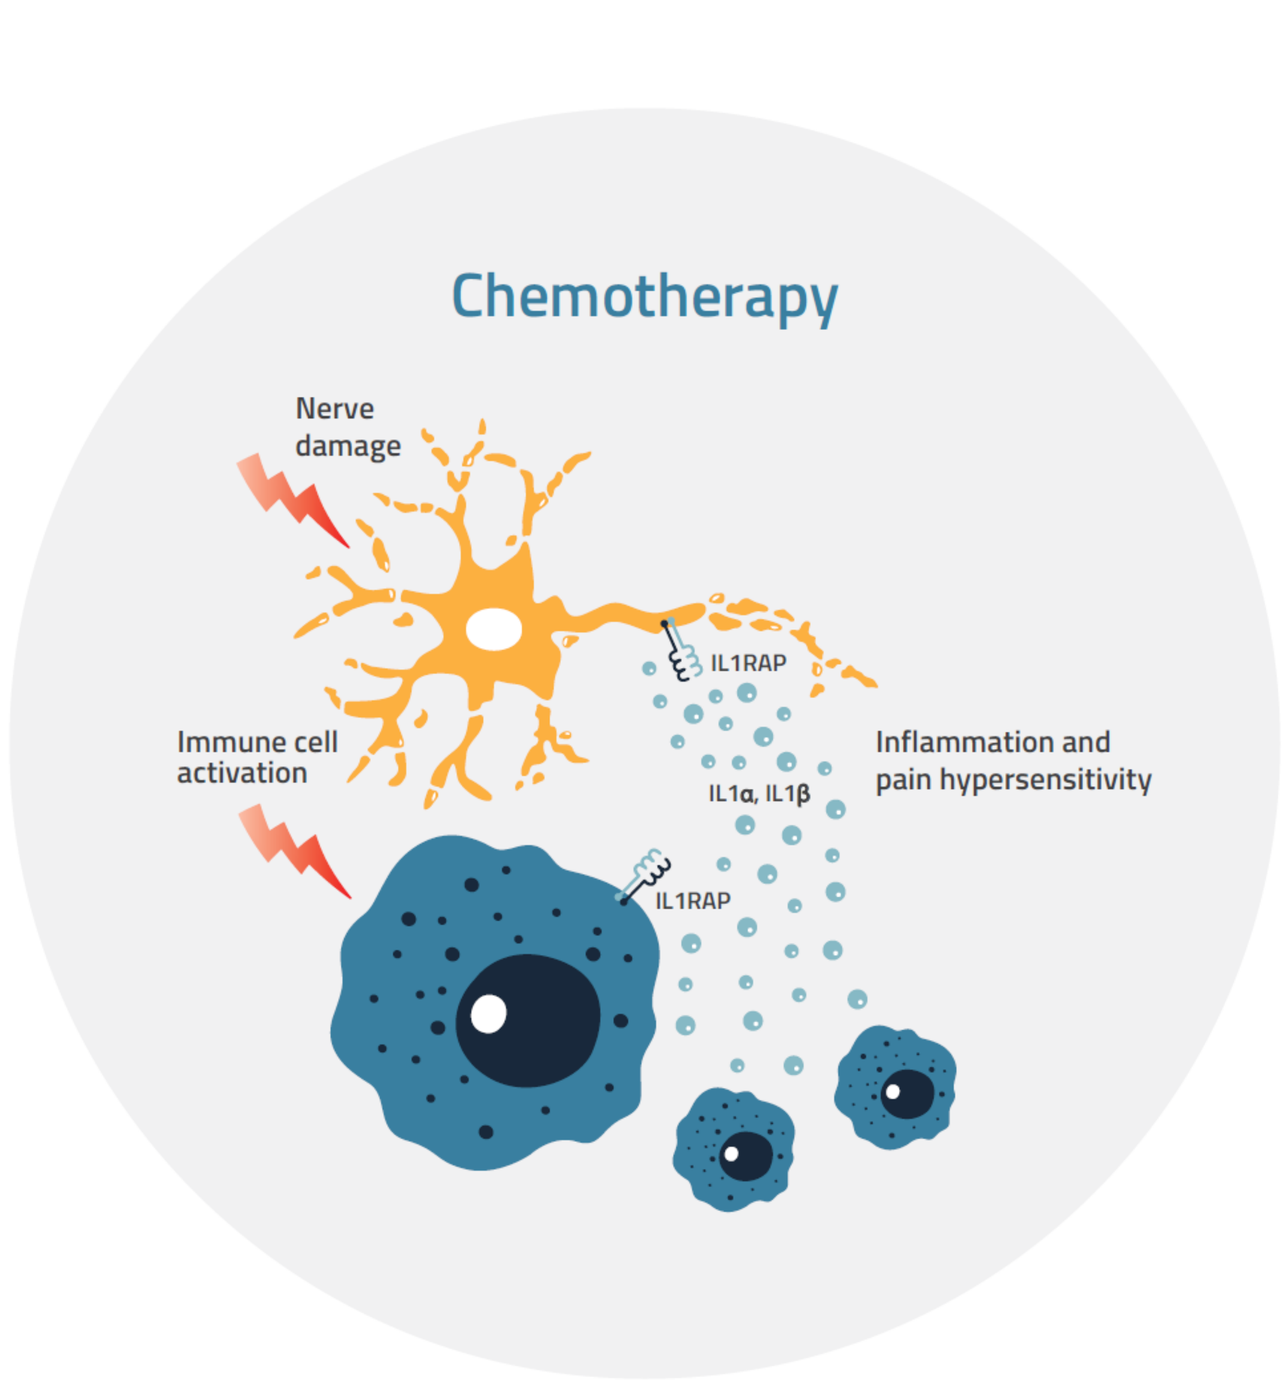
Chemotherapy
Nerve damage
Immune cell activation
IL1RAP
IL1α, IL1β
Inflammation and pain hypersensitivity

The low incidence of neuropathy after treatment with nadunolimab is in line with published data indicating that IL-1 mediated mechanisms play a key role in CIPN. IL-1α and IL-1β have been shown to be upregulated in response to chemotherapy and contributes to the neuroinflammation leading to neuropathic pain.

The aim of further analyses was to investigate the impact of blocking IL1RAP, and thereby inhibiting IL-1α and IL-1β signalling, on chemotherapy induced peripheral neuropathy.

Figure 3: Chemotherapy can induce nerve cell damage and immune cell activation, leading to neuropathies.

Patient Characteristics

Table 3: Patient demographics and disease characteristics at study start

	mITT (n=73)	1 mg/kg (n=20)	2.5 mg/kg or above (n=53)
Age; years; Median (Range)	63 (43-89)	63 (43-78)	62 (43-89)
Sex; F/M; n (%)	30 (41%) / 43 (59%)	7 (35%) / 13 (65%)	23 (43%) / 30 (57%)
ECOG PS; 0/1; n (%)	32 (44%) / 41 (56%)	7 (35%) / 13 (65%)	26 (49%) / 27 (51%)
BMI; Median (Range)	24.4 (16.5-36.1)	23.1 (17.9-31.4)	24.4 (16.5-36.1)
Prior therapies; n (%)			
Adjuvant/neoadjuvant chemotherapy	7 (9%)	2 (10%)	5 (9%)
Radical surgery	9 (12%)	2 (10%)	7 (13%)
Tumor location at study entry; n (%)			
Pancreas	67 (92%)	19 (95%)	48 (91%)
Liver	47 (64%)	10 (50%)	37 (70%)
Medical history; n (%)			
Diabetes*	21 (29%)	6 (30%)	15 (28%)
Peripheral Neuropathies**	4 (5%)	2 (10%)	2 (4%)

** Diabetes Mellitus and Type 2 Diabetes Mellitus in PT*
*** Peripheral Neuropathies NEC in High Level Term (HLT) and Paraesthesia in PT*

Results

Clinical data

Table 4: Dose groups and distribution of neuropathies

Parameter	1mg/kg (n=20pts)	2.5 to 7.5mg/kg (n=53 pts)
Grade 1	30% (6/20 pts)	15% (8/53 pts)
Grade 2+	30% (6/20 pts)	21% (11/53 pts)
any-grade CIPN	60%	36% *
Time to onset (median)	112 days	Not estimable**
Nab-paclitaxel doses/weeks on treatment (median)	15 doses/18.2 weeks	14 doses /18.1 weeks

*36% vs 60% (Chi-square test p=0.06)
**significantly longer time to onset; median not estimable vs 112 days (hazard ratio=0.48, log-rank p=0.04).

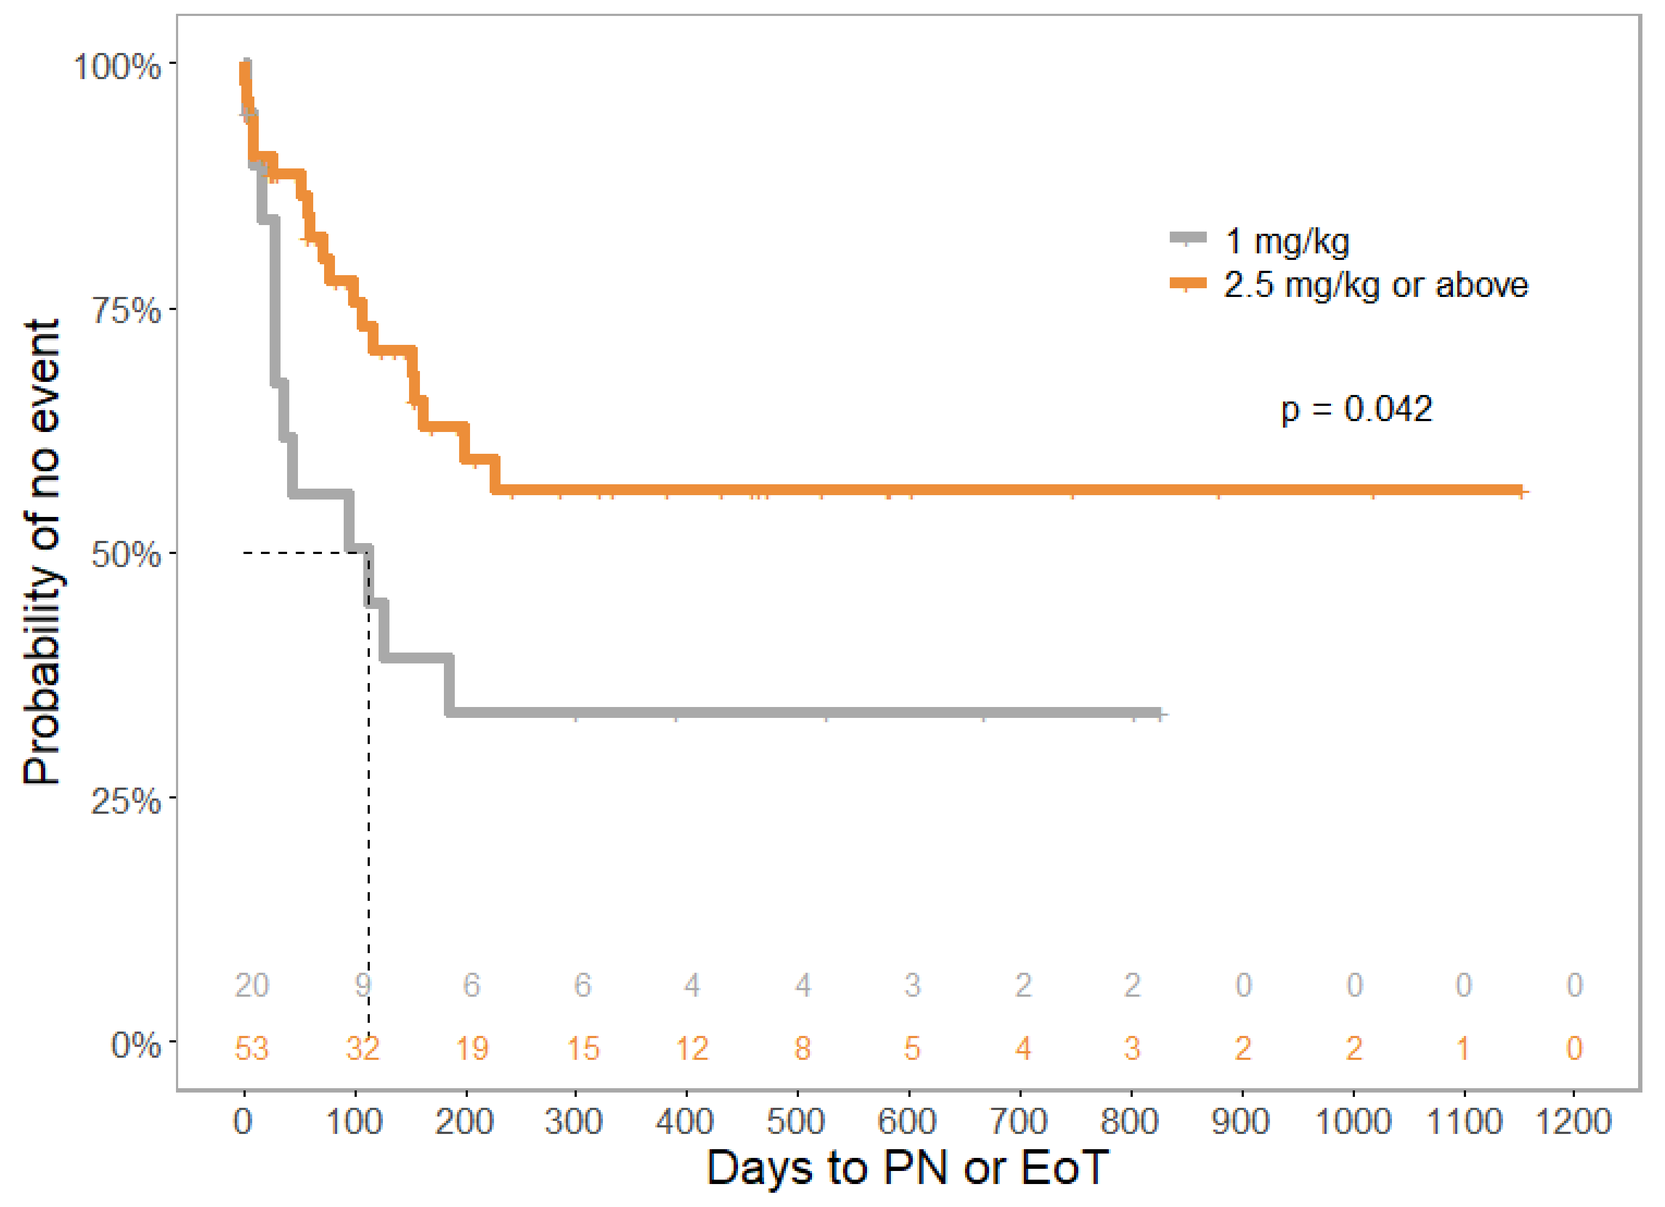
1 mg/kg
2.5 mg/kg or above
p = 0.042
Probability of no event
Days to PN or EoT

Figure 4: Reduced number of peripheral neuropathies after nadunolimab/GN treatment in the CANFOUR trial.

Dose groups 2.5-7.5 mg/kg were pooled and compared to the 1 mg/kg dose group. The higher dose groups showed a lower incidence of any-grade peripheral neuropathy. Chemotherapy doses given were comparable between the dose groups.

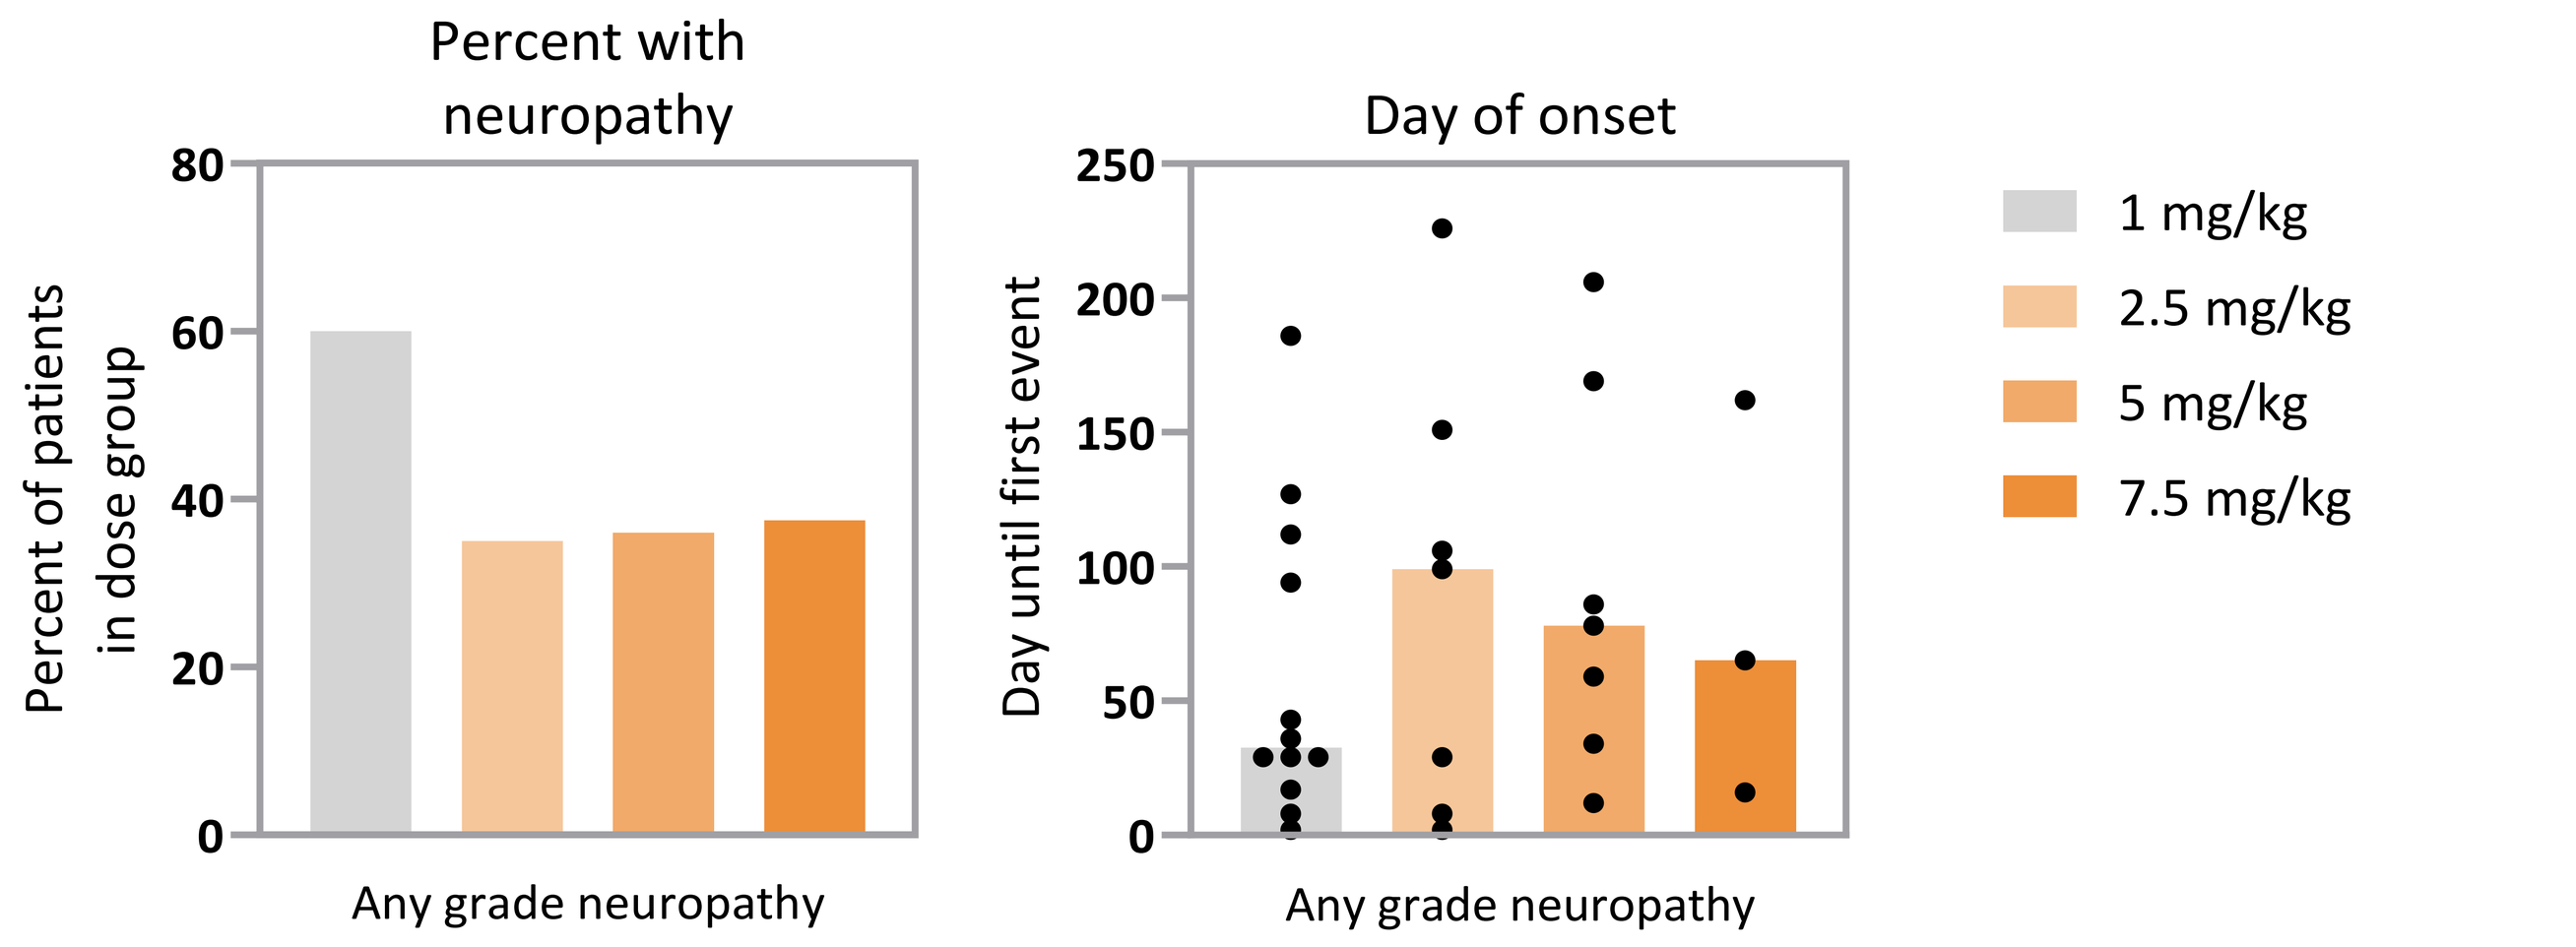
Percent with neuropathy
Percent of patients in dose group
Any grade neuropathy
Day of onset
Day until first event
Any grade neuropathy
1 mg/kg
2.5 mg/kg
5 mg/kg
7.5 mg/kg

Figure 5: Patients treated with higher doses of nadunolimab in combination with GN show lower incidence and later onset of peripheral neuropathies. *Percentage of patients in each dose group who had a neuropathy of any grade (left). For patients with a reported neuropathy of any grade, the day of onset of first event for patients in the different dose groups is shown to the right.*

Preclinical data: nab-paclitaxel

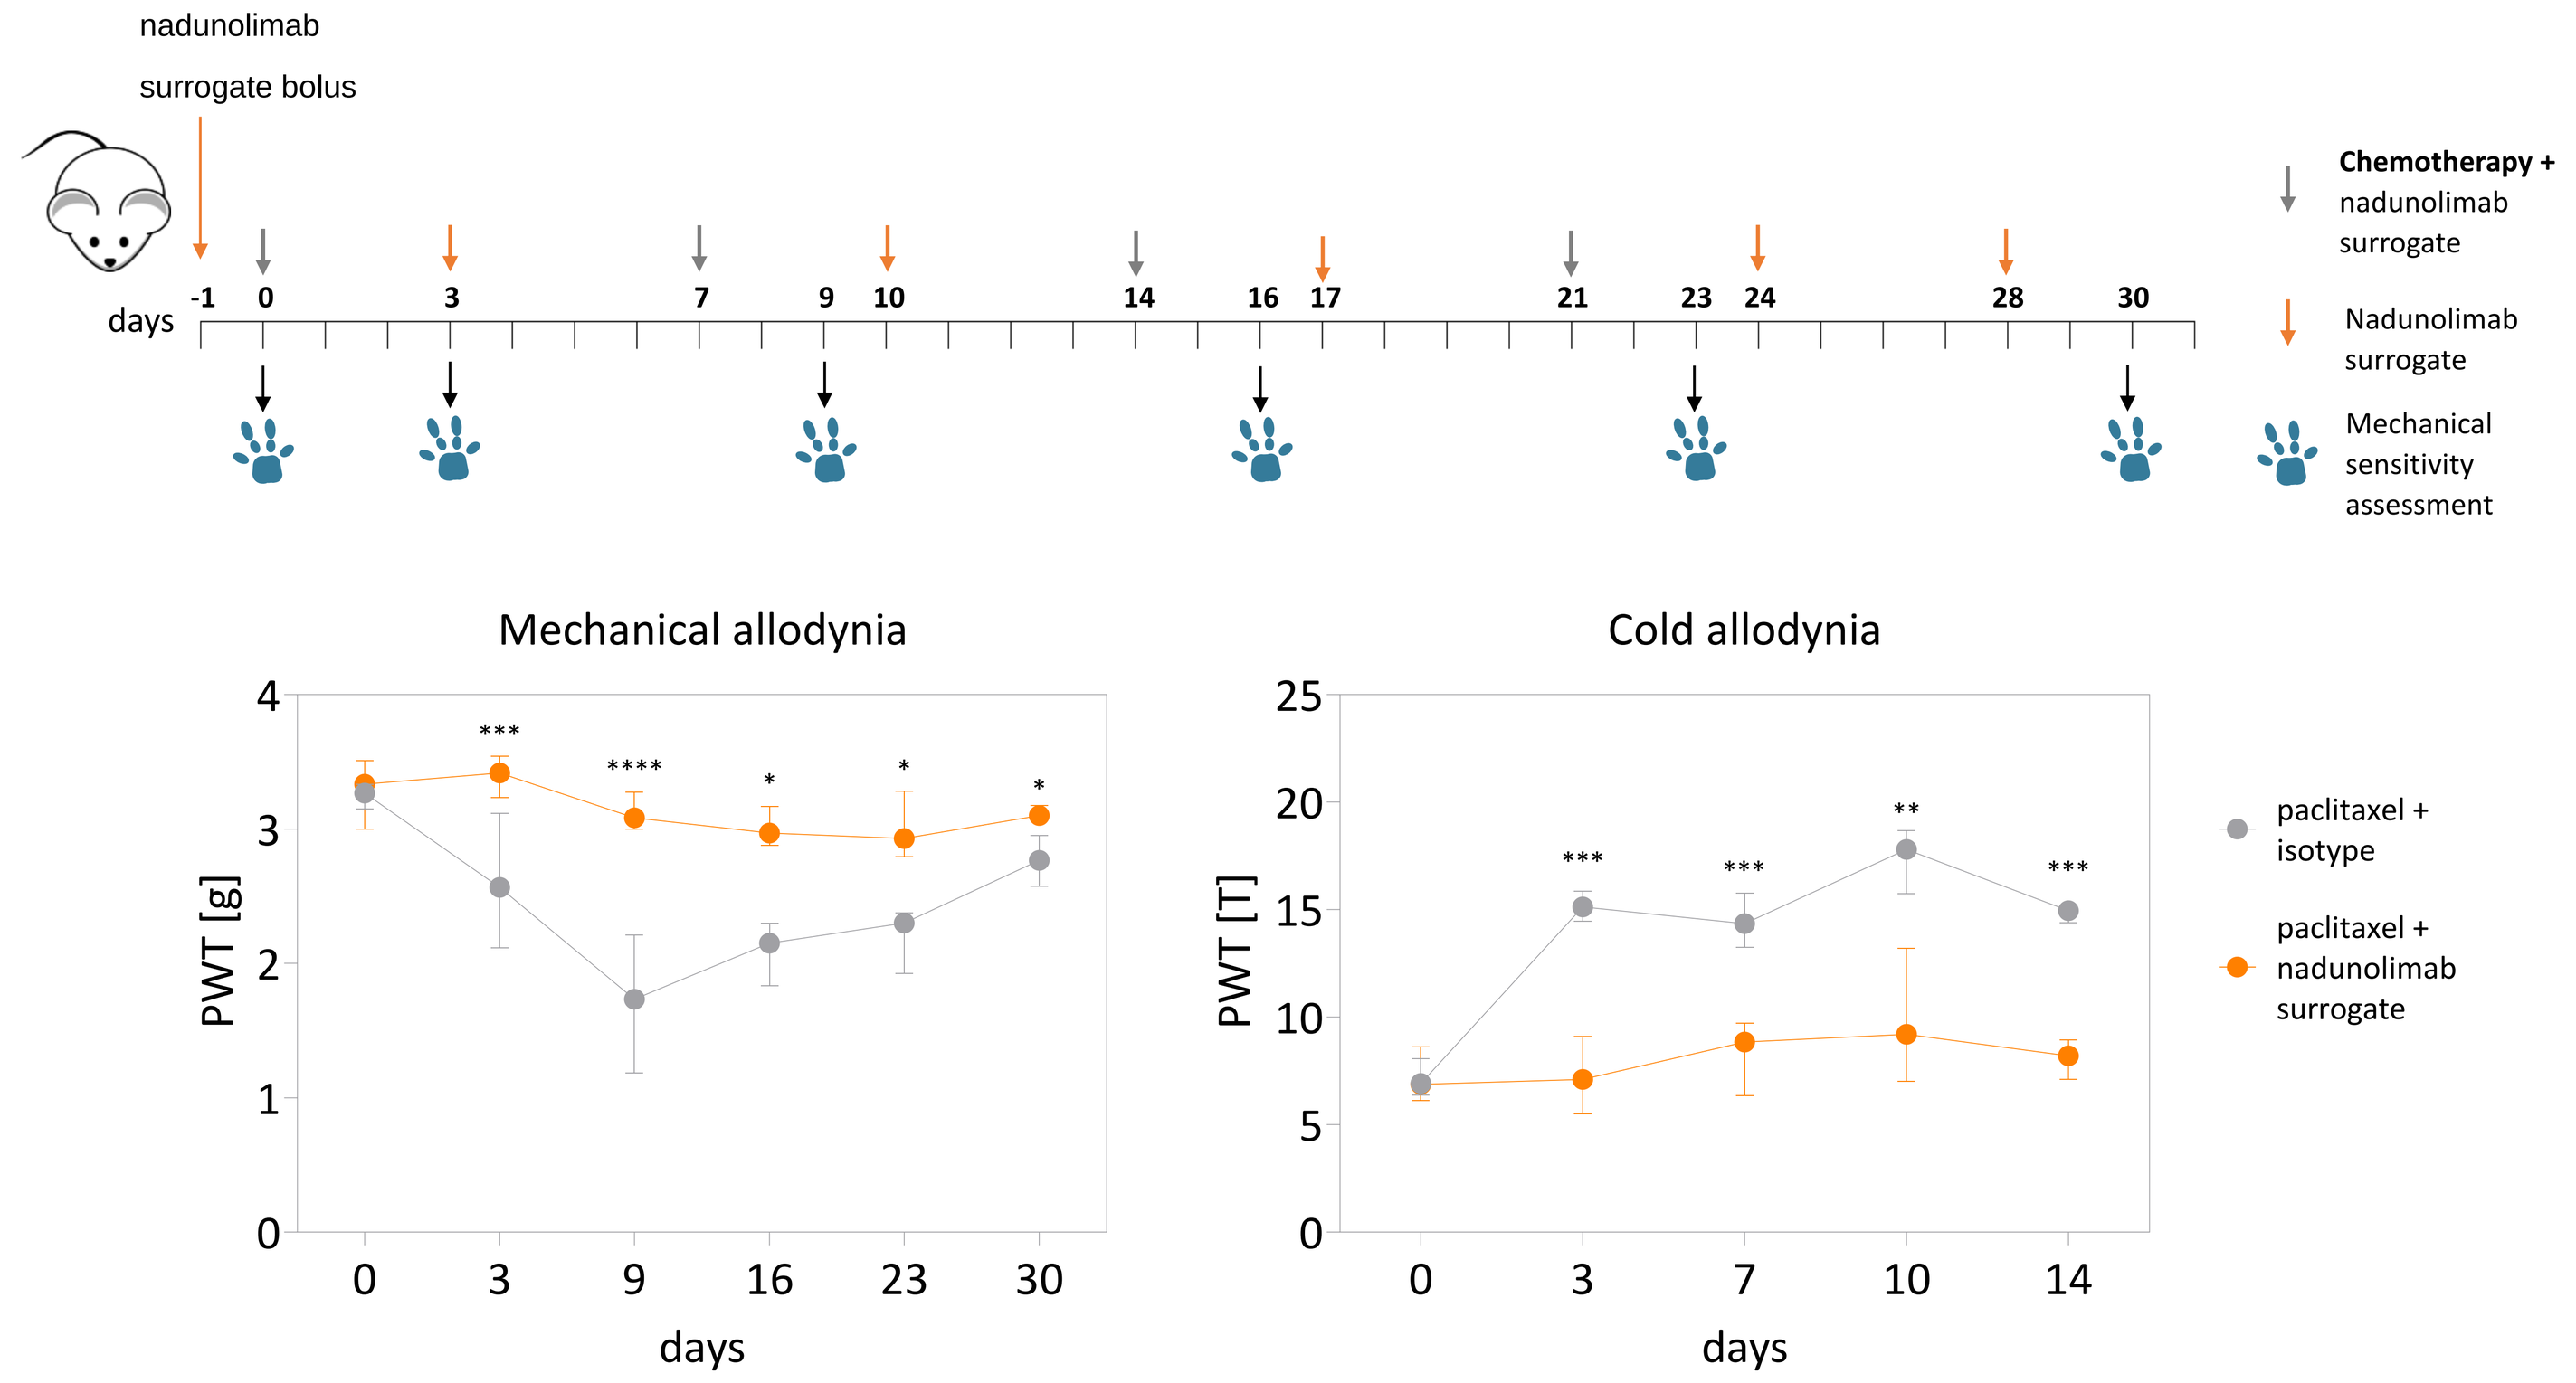
nadunolimab surrogate bolus
days
Chemotherapy + nadunolimab surrogate
Nadunolimab surrogate
Mechanical sensitivity assessment
Mechanical allodynia
PWT [g]
days
Cold allodynia
PWT [T]
days
paclitaxel + isotype
paclitaxel + nadunolimab surrogate

Figure 6: A surrogate antibody to nadunolimab prevents nab-paclitaxel induced mechanical and cold allodynia in mice. *Nab-paclitaxel was administered to mice i.p 4mg/kg once weekly and the aIL1RAP nadunolimab surrogate was administered i.p 10mg/kg twice weekly. Mechanical allodynia was measured by mechanical paw withdrawal thresholds (PWTs) using an electronic von Frey apparatus, TopCat Metrology (left). Cold allodynia was measured by MouseMet Thermal, TopCat Metrology (right). Graphs show median and interquartile range. Multiple Mann-Whitney test, *p<0.05; **p<0.01; ***p<0.005; ****p<0.001.*

Preclinical data: vincristine

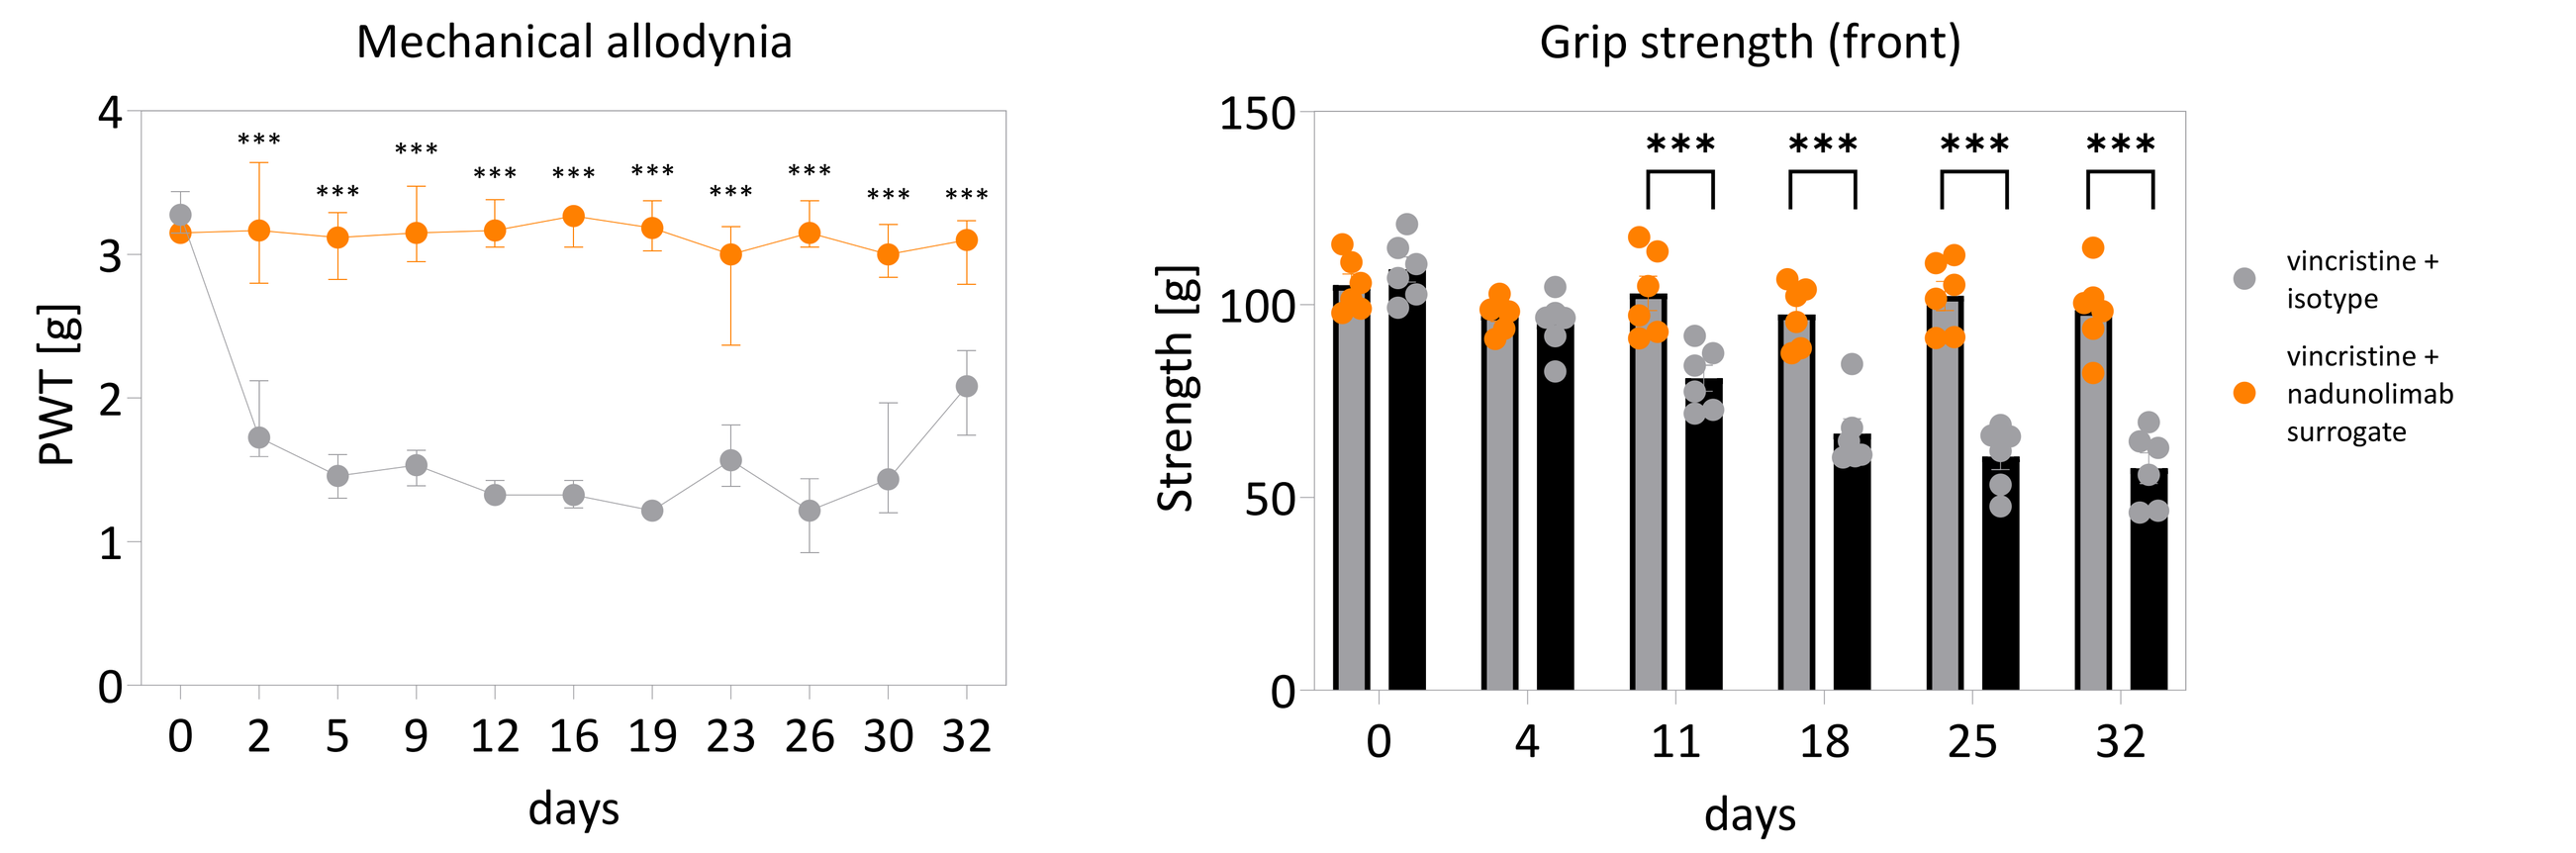
Mechanical allodynia
PWT [g]
days
Grip strength (front)
Strength [g]
days
vincristine + isotype
vincristine + nadunolimab surrogate

Figure 7: A surrogate antibody to nadunolimab prevents vincristine induced mechanical allodynia and grip strength loss in mice. *Vincristine was administered to mice i.p., at 0.25 mg/kg once a week for 4 weeks, and the aIL1RAP nadunolimab surrogate was administered i.p 10mg/kg twice weekly as described in Figure 6. Mechanical allodynia was measured as in Fig 7 (left). Grip strength was measured using the grip strength meter, Stoelting (right). Graph shows each data point and median. Multiple Mann-Whitney test, *p<0.05; **p<0.01; ***p<0.005; ****p<0.001.*

Conclusions

- Nadunolimab in combination with gemcitabine and nab-paclitaxel (GN) shows promising efficacy in first line PDAC; with a median iPFS of 7.2 months and median OS of 13.2 months.
- Only one grade 3 peripheral neuropathy was observed.
- Mouse model data with two types of chemotherapy show protection against neuropathy after treatment with a murine nadunolimab surrogate antibody.
- The reduction of neuropathy in CANFOUR appears driven by nadunolimab as patients in the 2.5-7.5 mg/kg dose groups had a lower incidence of any-grade neuropathy and a significantly longer time to onset, as compared to the 1 mg/kg dose group.
- **Data suggest that targeting IL1RAP by nadunolimab may combine antitumor activity with potent reduction of chemotherapy induced neuropathies.**

References:
1. Starobova et al; JEM 218(5), (2021)
2. Brandolini et al; Int. J. Mol. Sci. 20(12) (2019)
3. Klein et al; Toxics 9(10), (2021)
4. Domoto et al; Cells 10(8), (2021)

Acknowledgements: On behalf of the study team, the authors thank the patients and their families for participating in the study. This study was sponsored by Cantargia AB.

THE UNIVERSITY OF QUEENSLAND AUSTRALIA
CREATE CHANGE

cantargia